2-Drug Regimen Comparable to 3-Drug Regimens up to 18 Months in a Real-World Setting

Gerald Pierone[1], Kathy L. Schulman[2], Jennifer Fusco[2], Vani Vannappagari[3], Michael Aboud[4], Jean van Wyk[4], Leigh Ragone[3], Gregory Fusco[2]

[1]Whole Family Health Center, Vero Beach, FL, USA; [2]Epividian, Durham, NC, USA; [3]ViiV Healthcare, Research Triangle Park, NC, USA; [4]ViiV Healthcare, London, United Kingdom

Contact Information:
Jennifer S. Fusco
4819 Emperor Blvd, Suite 400, Durham, NC 27703
p: (224) 632-1863
@: jennifer.fusco@epividian.com

Poster #0491

Background

- Three antiretrovirals from two classes have long been the standard of care for people living with HIV (PLWH)[1-4]
- Newer, more powerful antiretrovirals have introduced the potential for effective therapy with fewer agents[5,6]
- Dolutegravir/rilpivirine (DTG/RPV) was the first single-tablet, once-daily regimen containing only two antiretrovirals to be approved (late 2017)[7]

OBJECTIVE

To compare the effectiveness and durability of DTG/RPV to standard three-drug regimens (3-DR) in a real-world setting

Methods

Study population

- Data source: OPERA database of electronic health records from 105,643 PLWH (148 cities in U.S. and Puerto Rico) as of 12 July 2019
- Inclusion Criteria:
 - » HIV- 1 positive, HIV-2 negative, ≥13 years of age
 - » Initiated a 2-DR (DTG/RPV) or 3-DR (DTG, EVG, RAL, DRV, RPV, or ATV + 2 NRTIs, boosted or unboosted) between 1 Jan 2018 and 31 Dec 2018
 - » Last viral load <50 copies/mL on or before initiation of regimen of interest
 - » No exposure to DTG/RPV prior to initiation
- Baseline: Date of initiation of 2-DR or 3-DR of interest
- Study outcomes:
 - » Virologic failure: 2 VL ≥ 200 copies/mL or 1 VL ≥ 200 copies/mL + regimen discontinuation
 - » Sustained suppression: Last VL <50 copies/mL and <200 copies/mL
 - » Treatment discontinuation: Modification or discontinuation of regimen of interest
- Follow-up until:
 - » Regimen discontinuation
 - » Death
 - » Study end (30 June 2019)

Analyses

- Description of patient characteristics and outcomes
 - » Categorical variables: Pearson's chi-square or Fisher exact tests
 - » Continuous variables: Wilcoxon rank-sum
- Time to discontinuation and virologic failure
 - » Kaplan-Meier methods
 - » Multivariable Cox Proportional Hazards models

Results

Table 1. Baseline demographic and clinical characteristics

Characteristic n (%)	DTG/RPV (n=545)	3-DR (n=5,524)	p-value
Age ≥50 years	298 (54.7%)	2,258 (40.9%)	<.0001
Female sex	97 (17.8%)	1,078 (19.5%)	0.5199
Black race	177 (32.5%)	2,445 (44.3%)	<.0001
Hispanic ethnicity	163 (29.9%)	1,139 (20.6%)	<.0001
Care in Southern US	350 (64.2%)	3,146 (57.0%)	<.0001
Hx of AIDS	146 (26.8%)	1,565 (28.3%)	0.4453
CD4 Count >500 cells/ µL	424 (77.8%)	3,959 (71.7%)	0.0178
Hx of Syphilis	164 (30.1%)	1,895 (34.3%)	0.0475
Any Comorbidity	475 (87.2%)	4,416 (79.9%)	<.0001

Figure 1. Distribution of core agents in the 3-DR group*

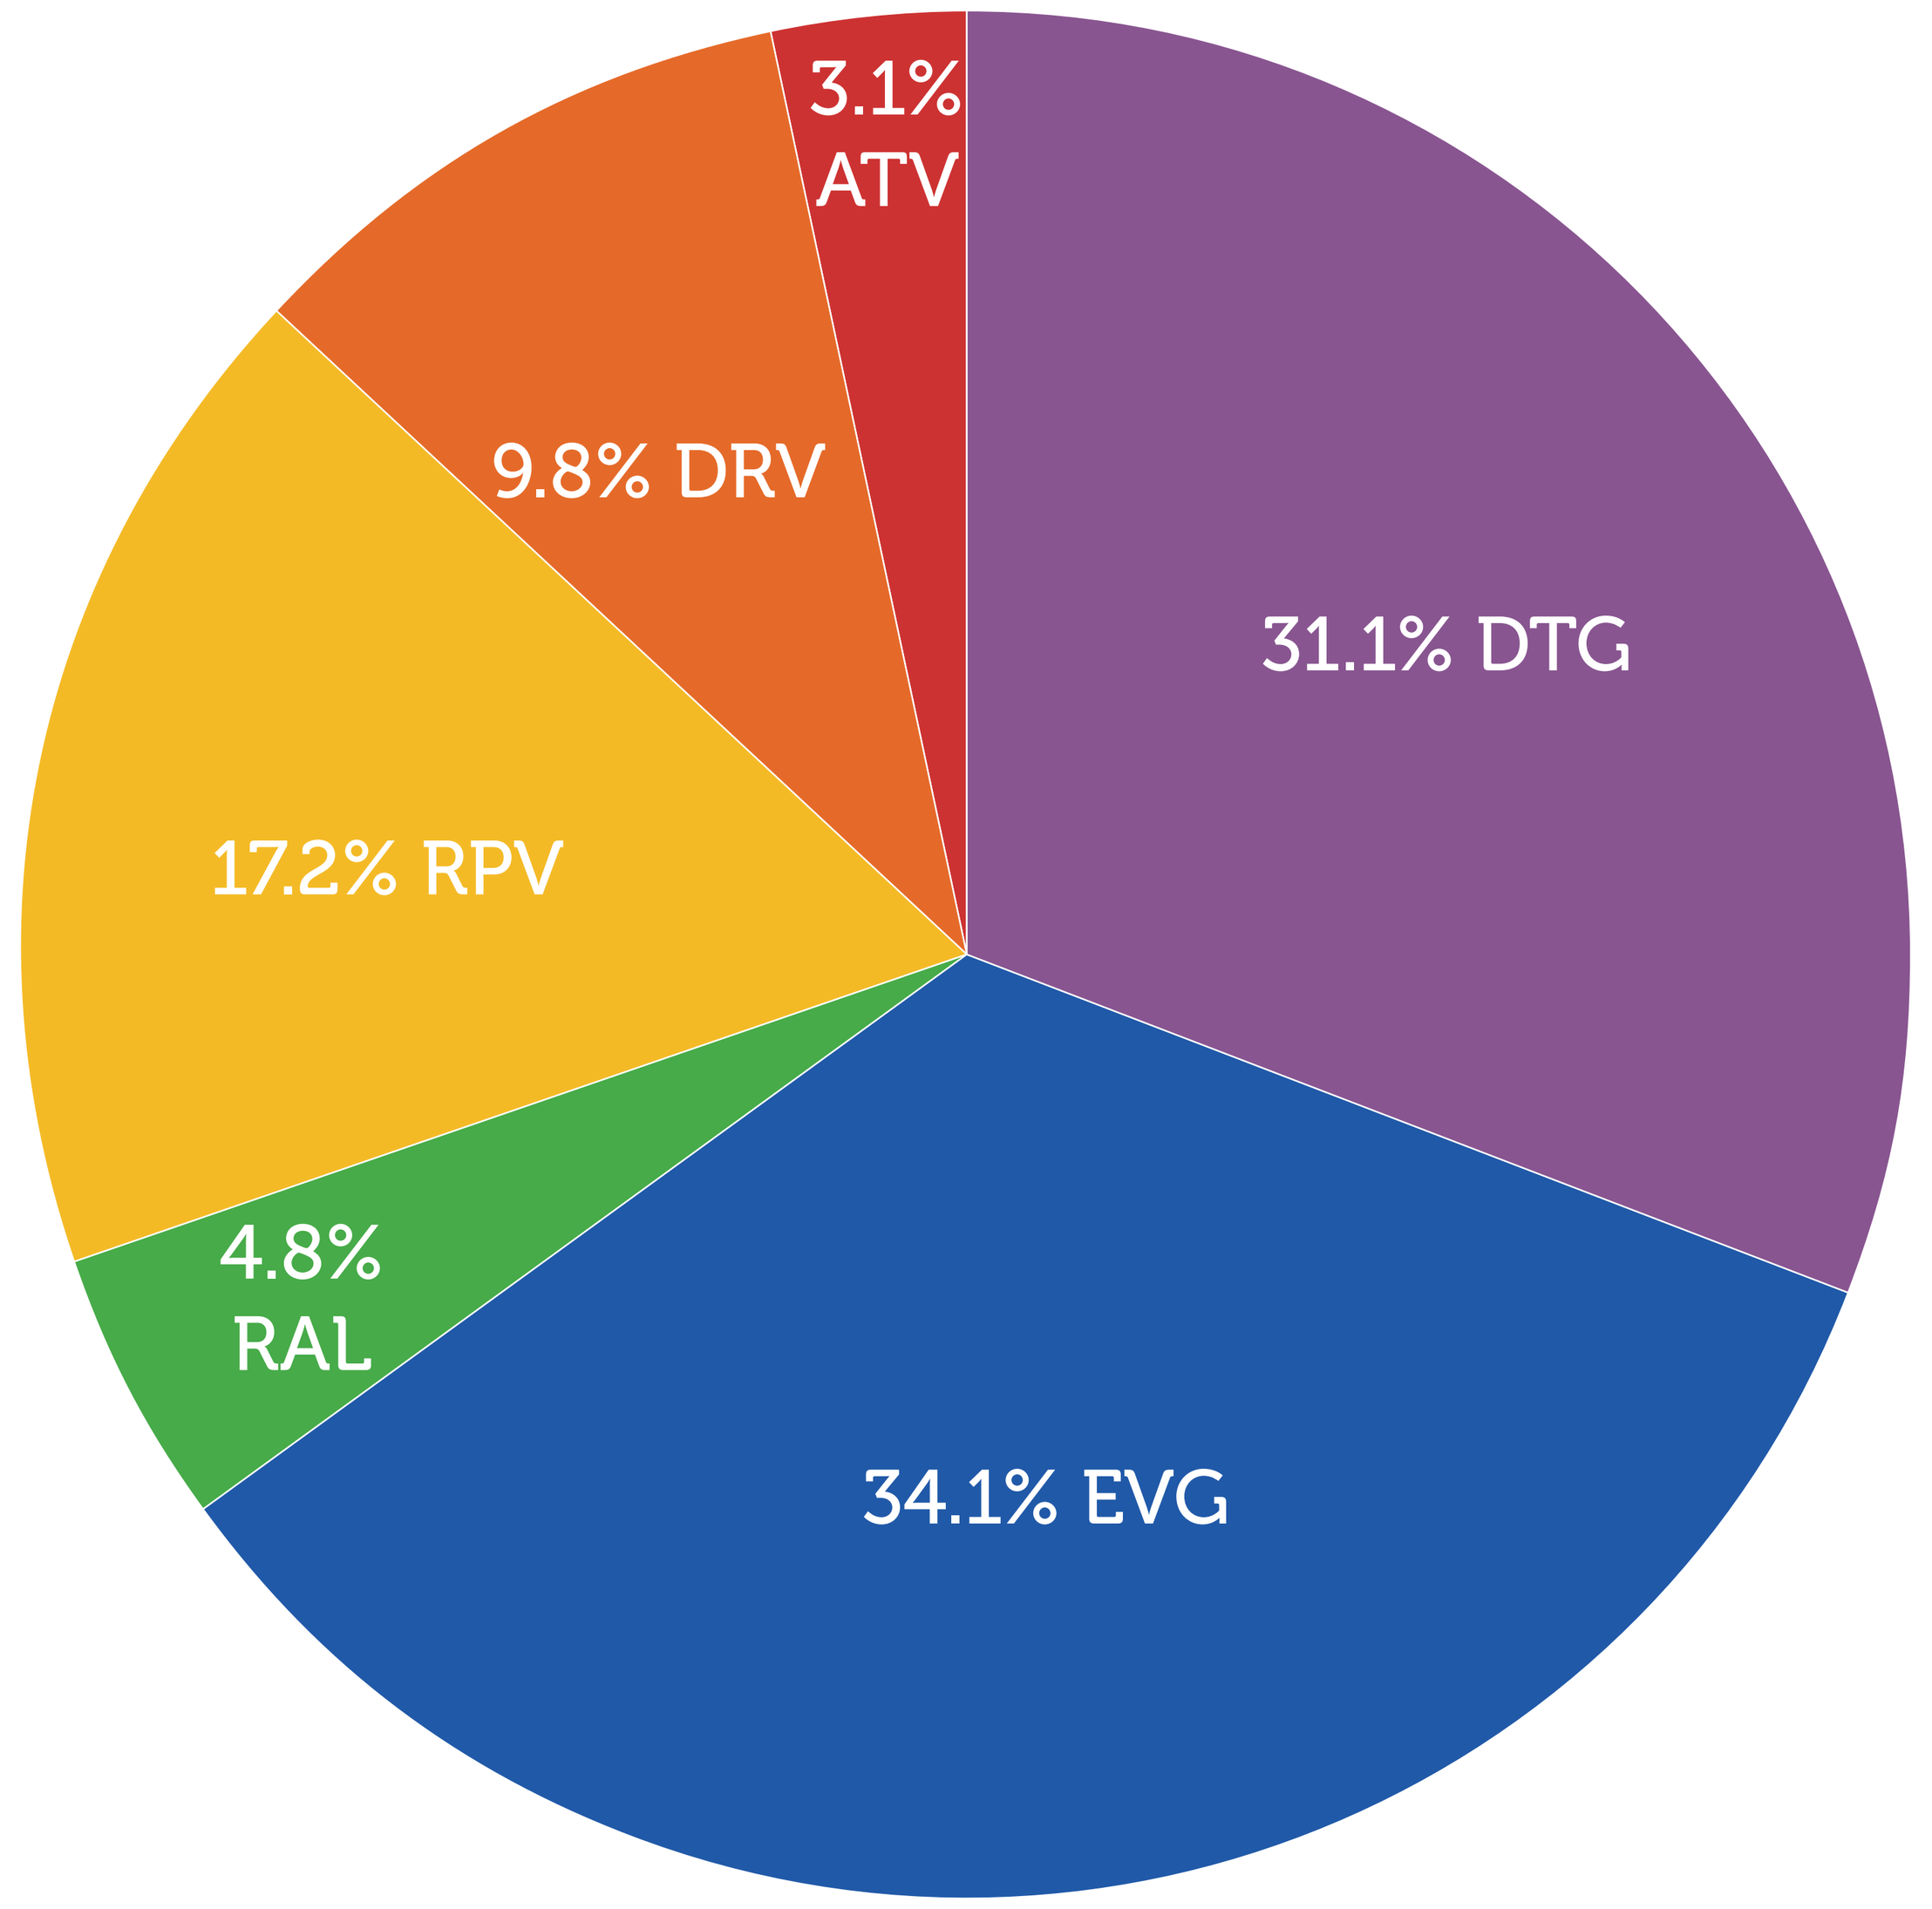
3.1% ATV
31.1% DTG
9.8% DRV
17.2% RPV
4.8% RAL
34.1% EVG

*DTG = Dolutegravir, EVG = Elvitegravir, RAL = raltegravir, RIL = rilpivirine, DRV = darunavir, ATV = Atazanavir

Table 2. Durability and virologic suppression with 2-DR versus 3-DR

Outcome (n, % or median, IQR)	DTG/RPV (n=545)	3-DR (n=5,524)	p-value
Durability			
Months on regimen	11.0 (8.2–14.2)	10.6 (6.8-14.6)	0.1432
Discontinuations	82 (15.0%)	1,544 (28.0%)	<.0001
Suppression among those tested			
Last VL < 50 copies/mL	457 (94.0%)	3,984 (89.0%)	0.0006
Last VL < 200 copies/mL	475 (97.7%)	4,275 (95.5%)	0.0221

Figure 2. Unadjusted cumulative probability of discontinuation of 2-DR versus 3-DR

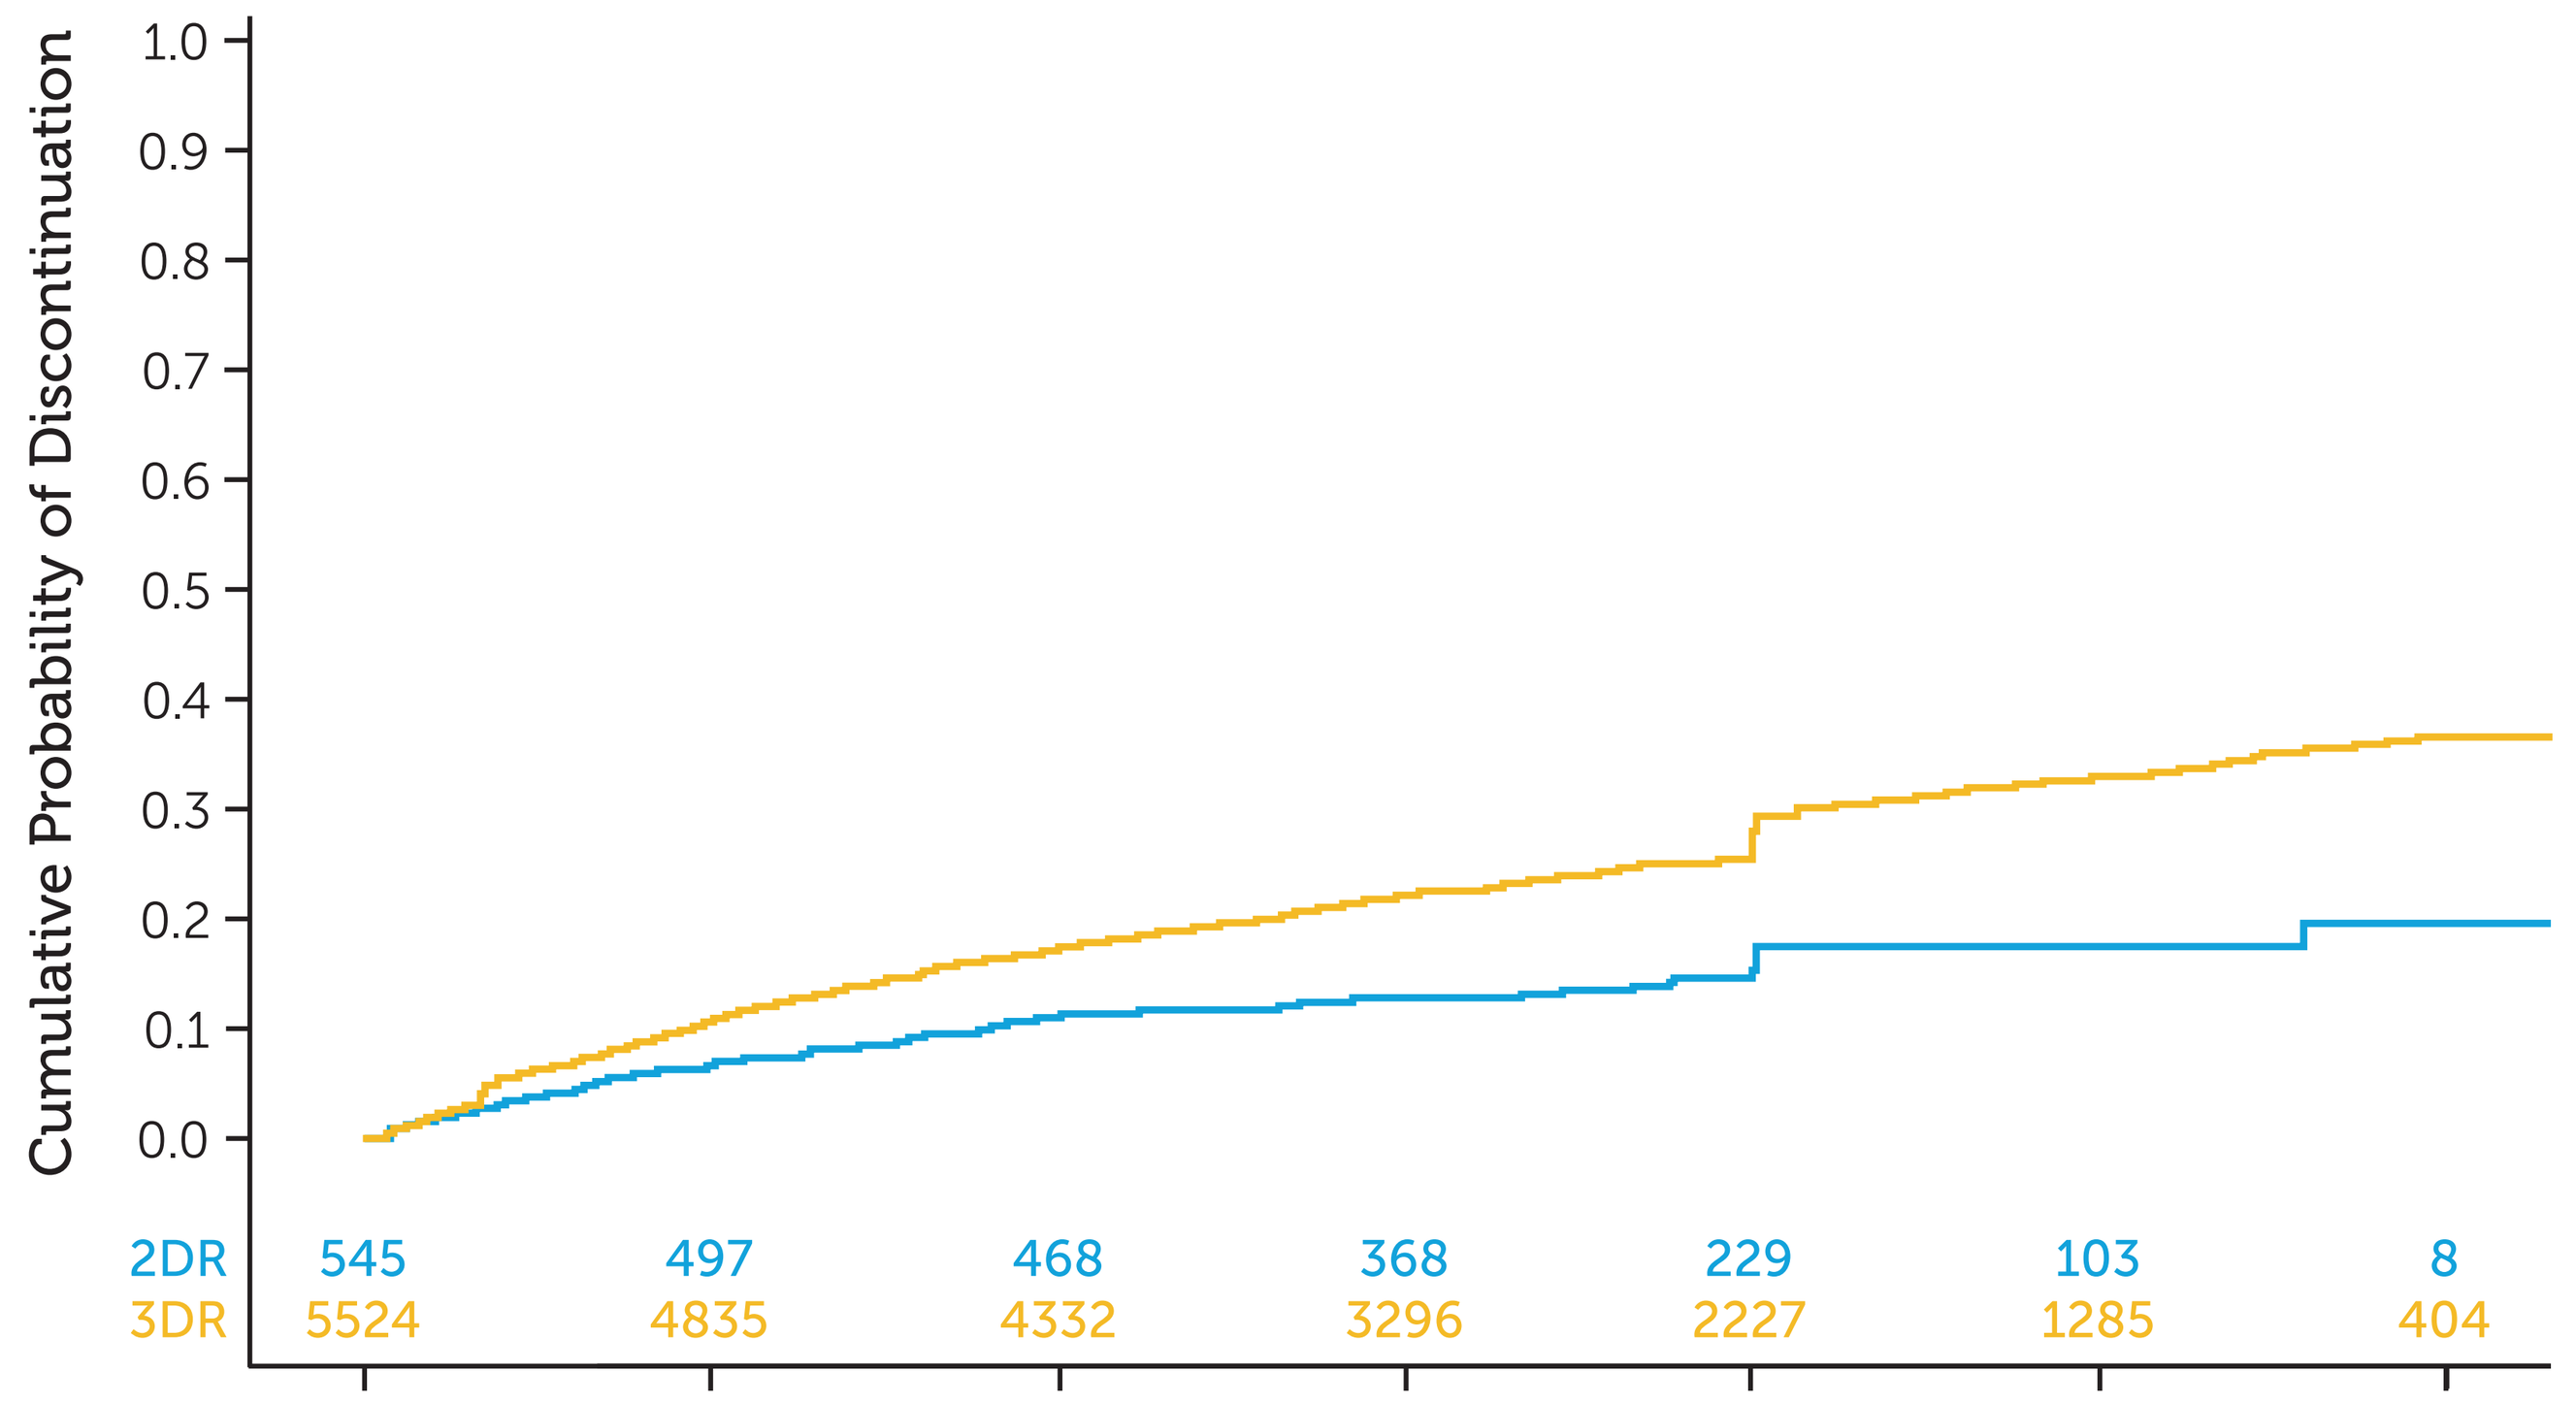
Cumulative Probability of Discontinuation
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
2DR 545 497 468 368 229 103 8
3DR 5524 4835 4332 3296 2227 1285 404

Figure 3. Unadjusted cumulative probability of virologic failure of 2-DR versus 3-DR

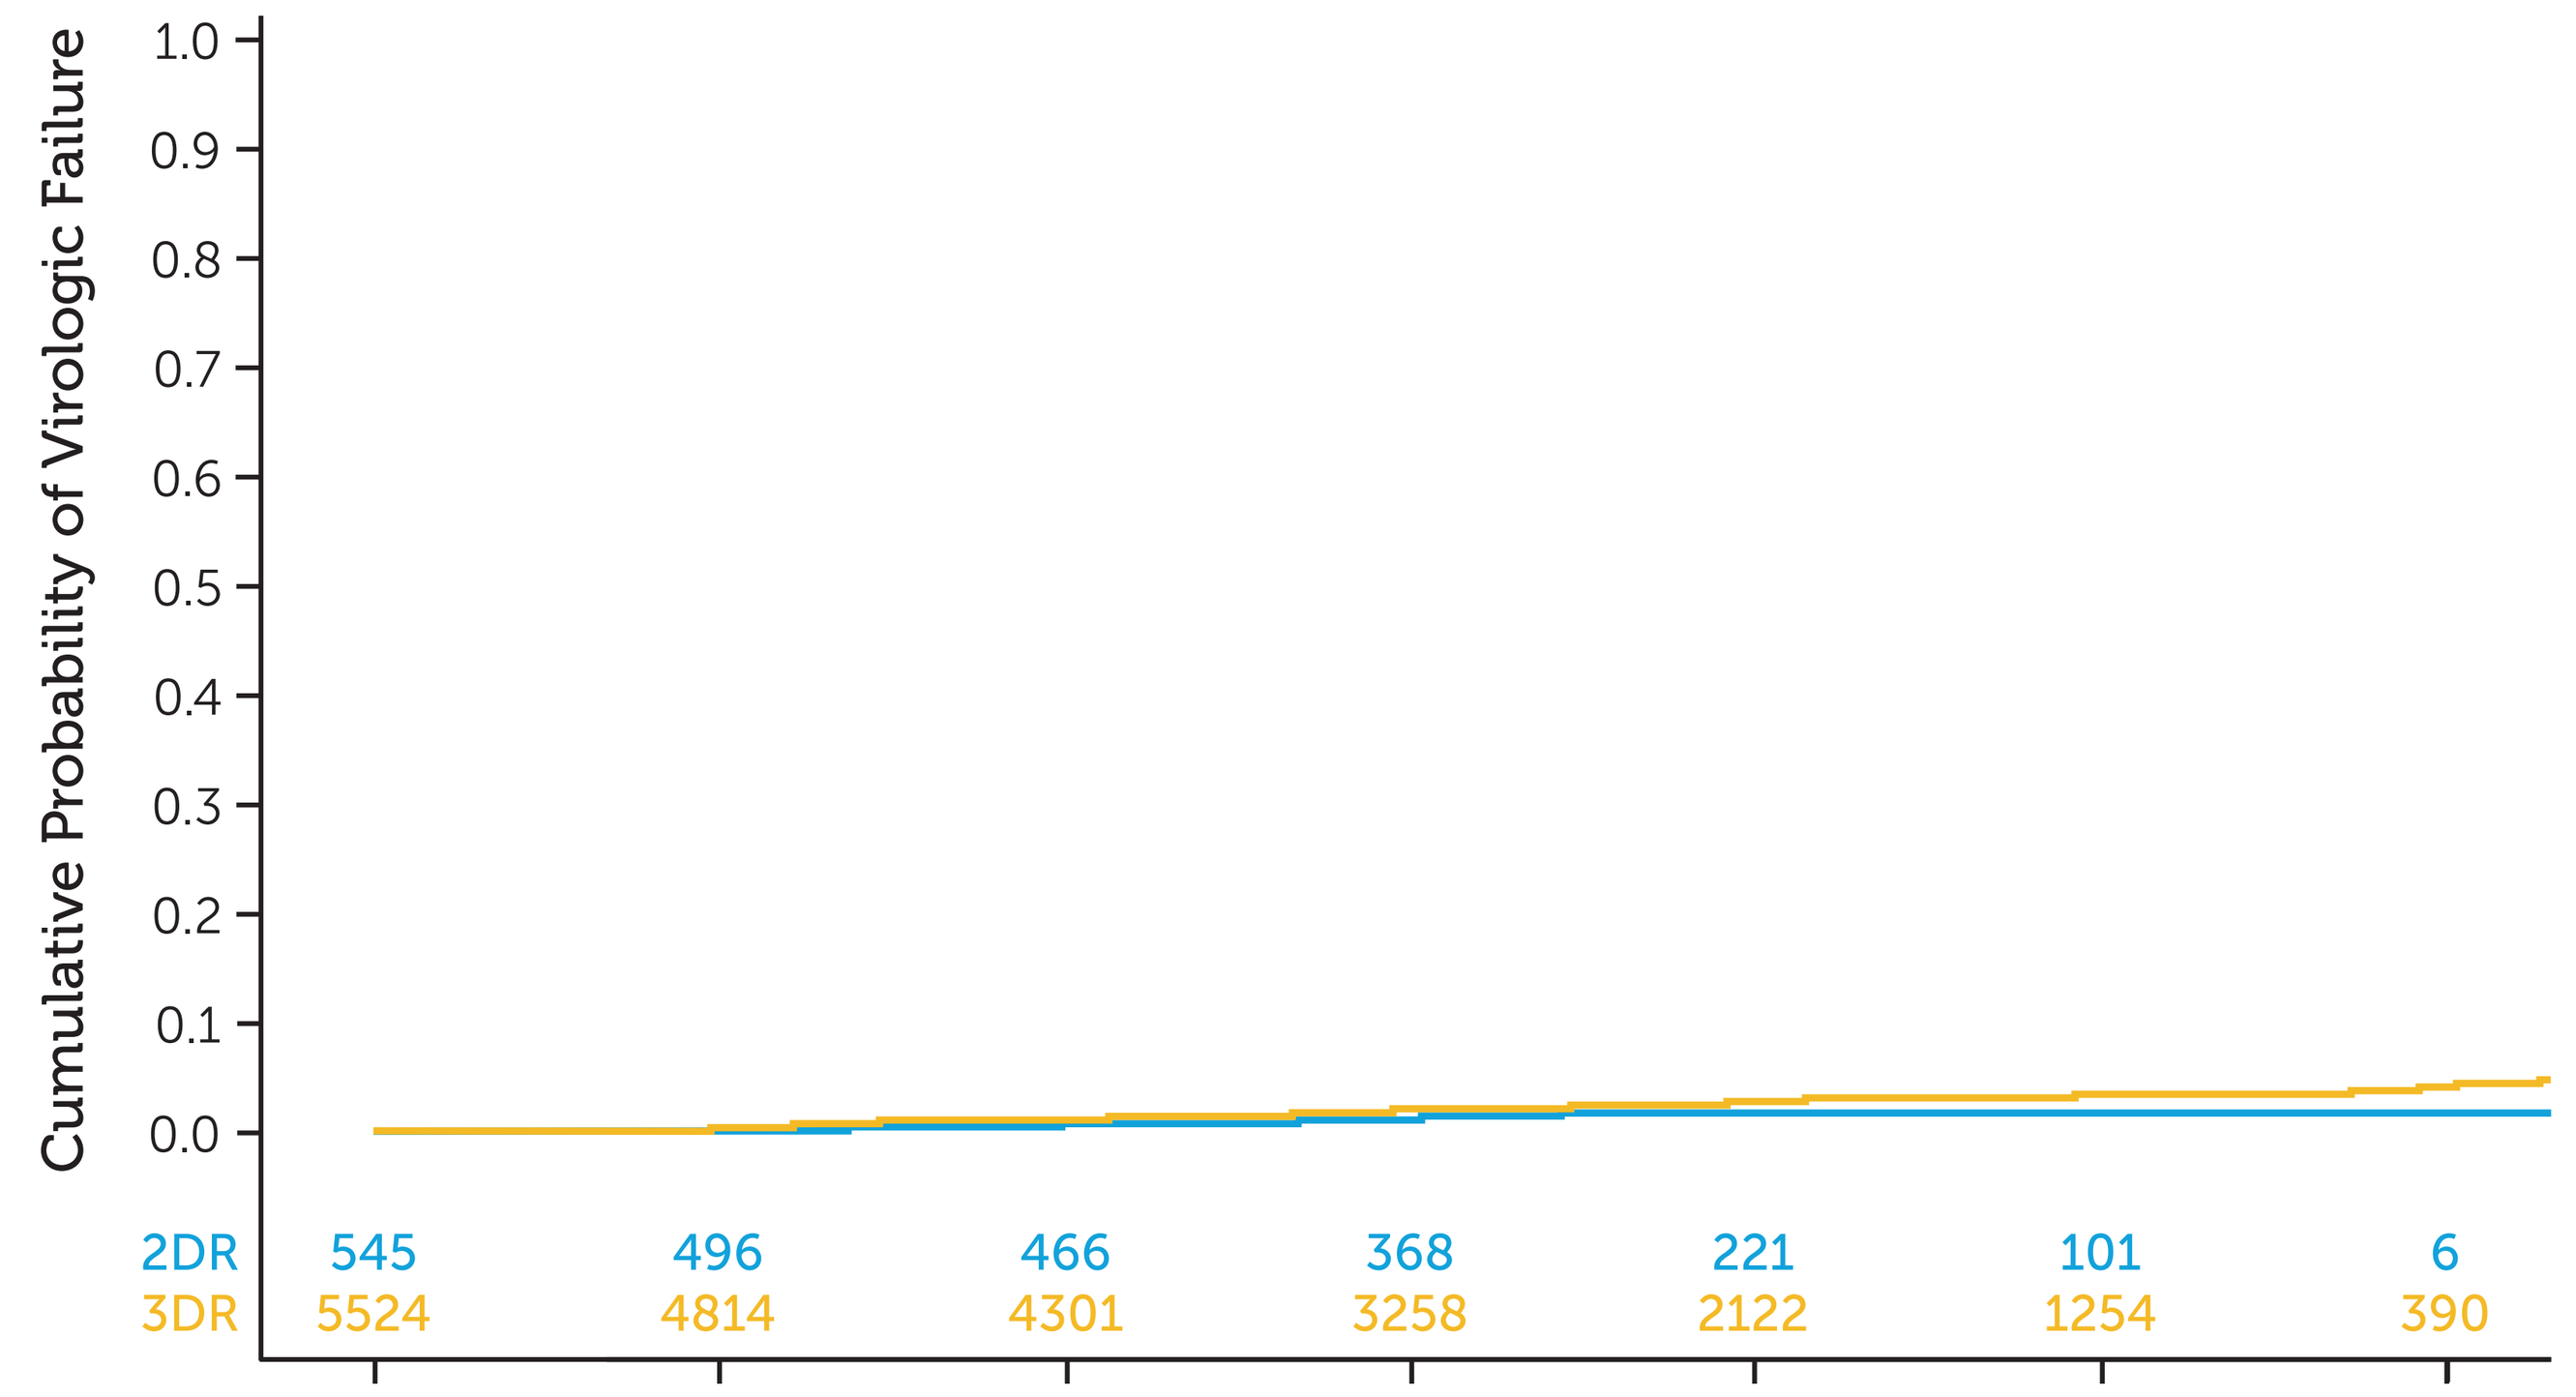
Cumulative Probability of Virologic Failure
1.0
0.9
0.8
0.7
0.6
0.5
0.4
0.3
0.2
0.1
0.0
2DR 545 496 466 368 221 101 6
3DR 5524 4814 4301 3258 2122 1254 390

Table 3. Virologic Failure with 2-DR versus 3-DR

Virologic Failure among those tested	DTG/RPV (n=545)	3-DR (n=5,524)	p-value
Virologic Failures, n (%)	7 (1.4%)	124 (2.8%)	0.0823
Incidence Rate (95% CI)*	1.45 (0.69, 3.03)	2.63 (2.21, 3.14)	0.1422
Unadjusted HR (95% CI)†	1.0	1.38 (0.43, 4.43)	0.1279
Adjusted HR (95% CI)‡	1.0	1.32 (0.61, 2.90)	0.4813

*per 100 person-years
†HR=Hazard Ratio
‡HR adjusted for age, sex, race, ethnicity, region, CD4 cell count, history of comorbidities

Discussion

- DTG/RPV users differed from 3-DR users notably (Table 1)
 - » DTG/RPV users were older, more likely to be Hispanic, to live in the southern US, and have comorbidities
 - » 3-DR users were younger, more likely to be African American, and have a history of syphilis (an indicator of a complex lifestyle)
- DTG/RPV users experienced fewer discontinuations and were more likely to be suppressed at study end compared to 3-DR users (Table 2, Figure 2)
- Virologic failure was uncommon and did not differ between DTG/RPV and 3-DR users (Table 3, Figure 3)
- Strengths: Large, diverse population of PLWH in the US
- Limitations: No reasons for discontinuation or resistance data for those who failed

KEY FINDINGS

Among ART-experienced, virologically suppressed PLWH initiating DTG/RPV or standard 3-DR, there was no observed difference in their risk of virological failure in a real-world setting over the first 18 months of approval

References

1. Merrill DP, Moonis M, Chou TC, Hirsch MS. Lamivudine or stavudine in two- and three-drug combinations against human immunodeficiency virus type 1 replication in vitro. J. Infect. Dis. 1996;173:355-364.
2. Hammer SM, Squires KE, Hughes MD, et. al. A controlled trial of two nucleoside analogues plus indinavir in persons with human immunodeficiency virus infection and CD4 cell counts of 200 per cubic millimeter or less. N. Engl. J. Med. 1997;337:725-733.
3. Squires KE, Gulick R, Tebas P, Santana J, Mulanovich, et. al. A comparison of stavudine plus lamivudine versus zidovudine plus lamivudine in combination with indinivir in antiretroviral naïve individuals with HIV infection: selection of thymidine analog regimen therapy (START I). AIDS. 2000;14(11):1591-1600.
4. Gulick RM, Mellors JW, Havlir D, Eron JJ, Gonzalez C, McMahon D, Jonas L, Meibohm A, et. al. Simultaneous vs Sequential Initiation of Therapy with Indinivir, Zidovudine, and Lamivudine for HIV-1 Infection. JAMA. 1998;280:35-41.
5. Pierone G, Henegar C, Fusco J, Vannappagari V, Aboud M, Ragone L, Fusco G. Virologic response to 2-drug ART regimens among treatment-experienced HIV+ patients. Conference on Retroviruses and Opportunistic Infections. Boston, MA. March 03-07, 2018.
6. Pierone G, Schulman KL, Fusco J, Vannappagari V, Aboud M, Ragone L, Fusco G. Characteristics of early adopters of a two-drug regimen (dolutegravir/rilpivirine) for treatment of human immunodeficiency virus type 1 in the United States. AMCP Managed Care and Specialty Pharmacy Annual Meeting. San Diego, CA. March 25-28, 2019.
7. Cahn P, Sierra Madero J, Arribas J, et al; and GEMINI Study Team. Dolutegravir plus lamivudine versus dolutegravir plus tenofovir disoproxil fumarate and emtricitabine in antiretroviral-naive adults with HIV-1 infection (GEMINI-1 and GEMINI-2): week 48 results from two multicentre, double-blind, randomised, non-inferiority, phase 3 trials. Lancet. 2019;393(10167):143-155.

Acknowledgements

This research would not be possible without the generosity of people living with HIV and their OPERA caregivers. Additionally, we are grateful for the following individuals: Amelito Torres (SAS programming), Jeff Briney (QA), Bernie Stooks (Database Arch & Mgmt), Judy Johnson (Med Terminology Classification), Rodney Mood (Site Support) and Antonin Fusco (Poster Preparation).

Support

This research was sponsored by ViiV Healthcare, Research Triangle Park, NC, USA.

Conference on Retroviruses and Opportunistic Infections (CROI 2020), Boston, MA, 8-11 March 2020