Poster #337

Switching from TDF to TAF: Missed Opportunities for Statin Use in HIV

Patrick Mallon[1], Laurence Brunet[2], Jennifer Fusco[2], Girish Prajapati[3], Andrew Beyer[3], Gregory Fusco[2], Michael Wohlfeiler[4]

[1]School of Medicine, University College Dublin, Dublin, Ireland; [2]Epividian, Durham, NC, USA; [3]Merck & Co., Inc., Kenilworth, NJ, USA; [4]AIDS Healthcare Foundation, Miami, FL USA

OPERA® The Longitudinal Cohort

Contact Information:
Laurence Brunet
4505 Emperor Blvd, Suite 220, Durham, NC 27703
p: 919-827-0010 e: laurence.brunet@epividian.com

Background

- People living with HIV (PLWH) have twice the risk for atherosclerotic cardiovascular disease (ASCVD) as the general population
- Increases in total and LDL cholesterol have been observed in PLWH switching from tenofovir disoproxil fumarate (TDF) to tenofovir alafenamide (TAF)[1]
- Current guidelines recommend initiating statin therapy in individuals with an elevated ASCVD risk (ASCVD ≥7.5%)[2]

OBJECTIVE

To assess missed opportunities for statin therapy in PLWH switching from TDF- to TAF-containing antiretroviral therapy

Methods

Study population

- OPERA cohort
 - Prospectively captured, routine clinical data from electronic health records (84 clinics, 17 states, 1 US territory)
 - ~7% of PLWH in care in the US
- Inclusion criteria
 - HIV positive, ≥18 years of age
 - Switched from TDF to TAF between 5Nov2015 and 31Mar2018
 - ≥1 lipid measure on TDF ≤6 months prior to switch
 - ≥1 lipid measure ≥7 days after switch to TAF
 - No PEP, PrEP or clinical trial participation
- Observation period from TDF-to-TAF switch until the first censoring event:
 - TAF discontinuation
 - ≥12 months without contact
 - Death
 - Study end (30Jun2018)

Lipid lowering agents

- Statins
 - Prescription of atorvastatin, pitavastatin, pravastatin, rosuvastatin, or simvastatin
 - With or without other lipid lowering agents
- Non-statins
 - Prescription of cholestyramine resin, ethyl eicosapentaenoic acid, ezetimibe, fenofibrate, gemfibrozil, niacin, Omega-3 acid ethyl esters, Omega-3 fatty acids
 - Without statins

ASCVD risk score

- Derived from the Pooled Cohort Equations calculator[2]
- Predicts the risk of developing a first ASCVD event (i.e. nonfatal myocardial infarction, coronary heart disease death, or fatal or nonfatal stroke) over 10 years among people free from ASCVD
- Based on sex, age, race, total cholesterol, HDL, systolic blood pressure, hypertension treatment, diabetes and smoking status
- Imputed using limit value if age <40 or >79, systolic BP <90 or >200 mmHg, HDL <20 or >100 mg/dL, total cholesterol <130 or >320 mg/dL

Analyses

- Proportion of PLWH prescribed statins pre- and post-switch
- Stratified by ASCVD risk

Results

Figure 1. Demographic and clinical characteristics of PLWH switching from TDF to TAF (N=6,451)

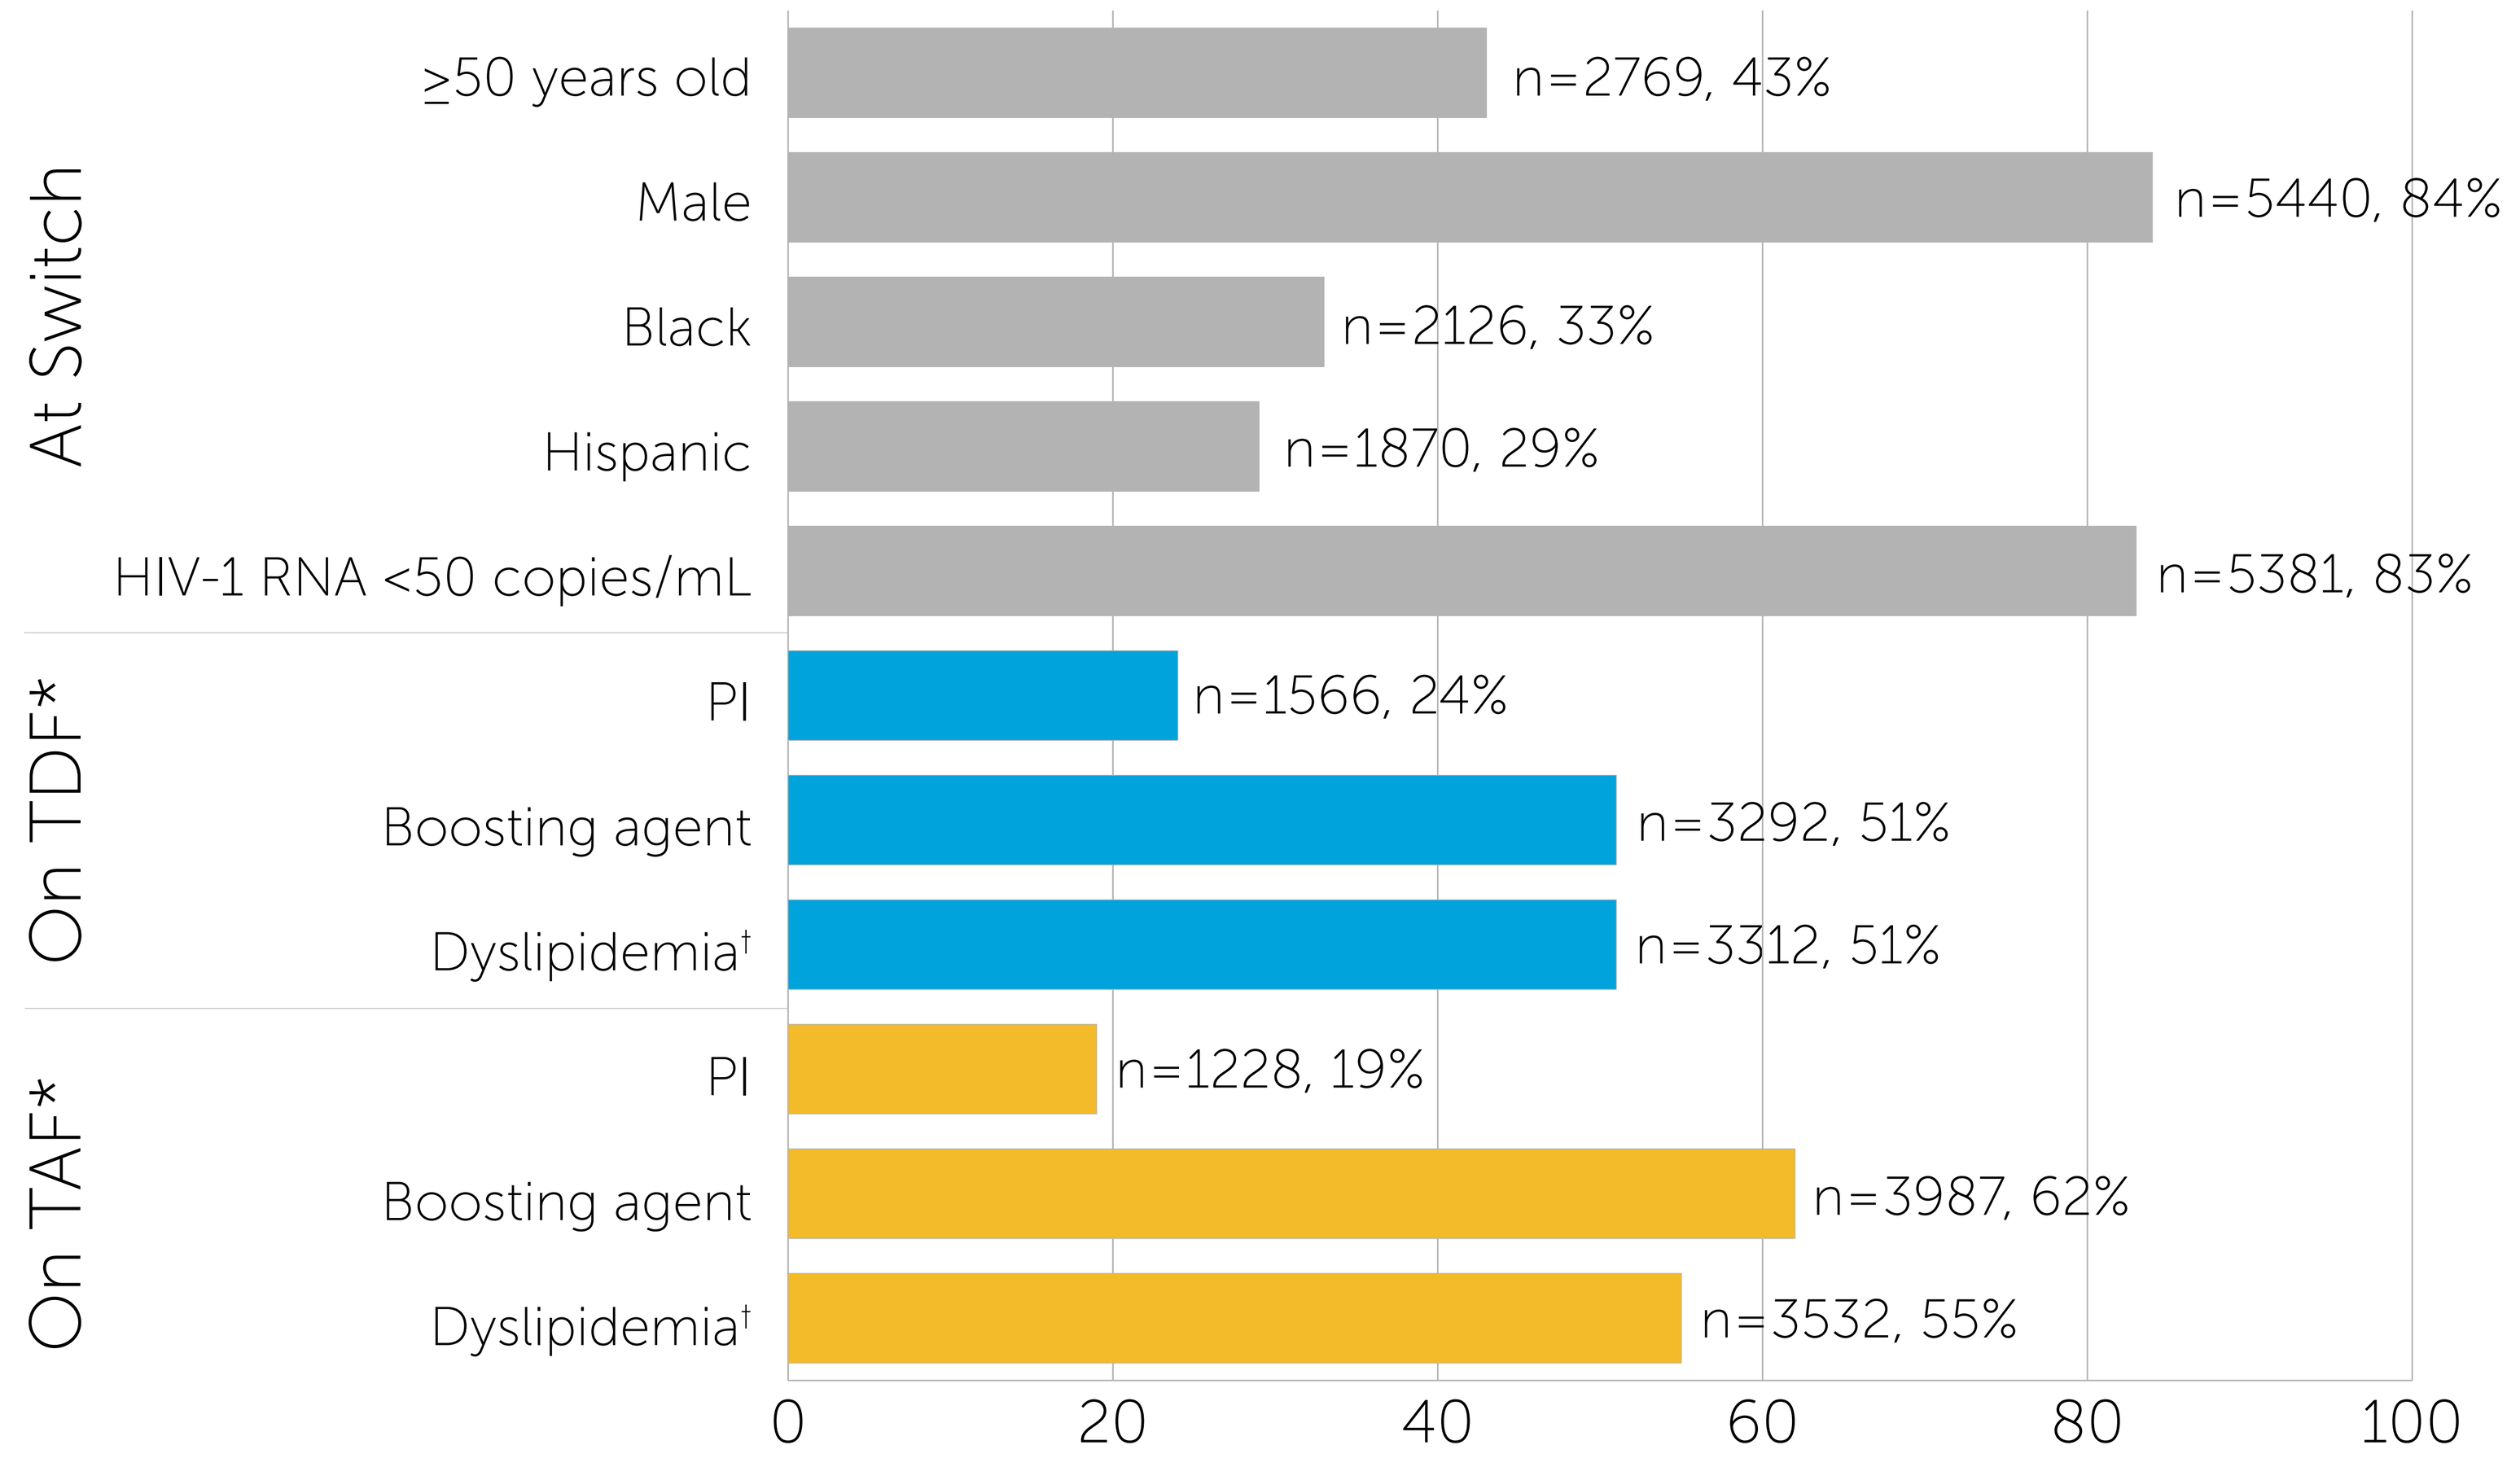

At Switch
≥50 years old
n=2769, 43%
Male
n=5440, 84%
Black
n=2126, 33%
Hispanic
n=1870, 29%
HIV-1 RNA <50 copies/mL
n=5381, 83%
On TDF*
PI
n=1566, 24%
Boosting agent
n=3292, 51%
Dyslipidemia†
n=3312, 51%
On TAF*
PI
n=1228, 19%
Boosting agent
n=3987, 62%
Dyslipidemia†
n=3532, 55%
0
20
40
60
80
100

* At the time of the lipid panel on TDF or on TAF
† NCEP ATP III dyslipidemia definition: total cholesterol ≥240 mg/dl, or LDL ≥130 mg/dl, or HDL <40 mg/dl, or triglycerides≥ 200 mg/dl

Figure 2. Lipid-lowering agent use before and after switch (N=6,451)

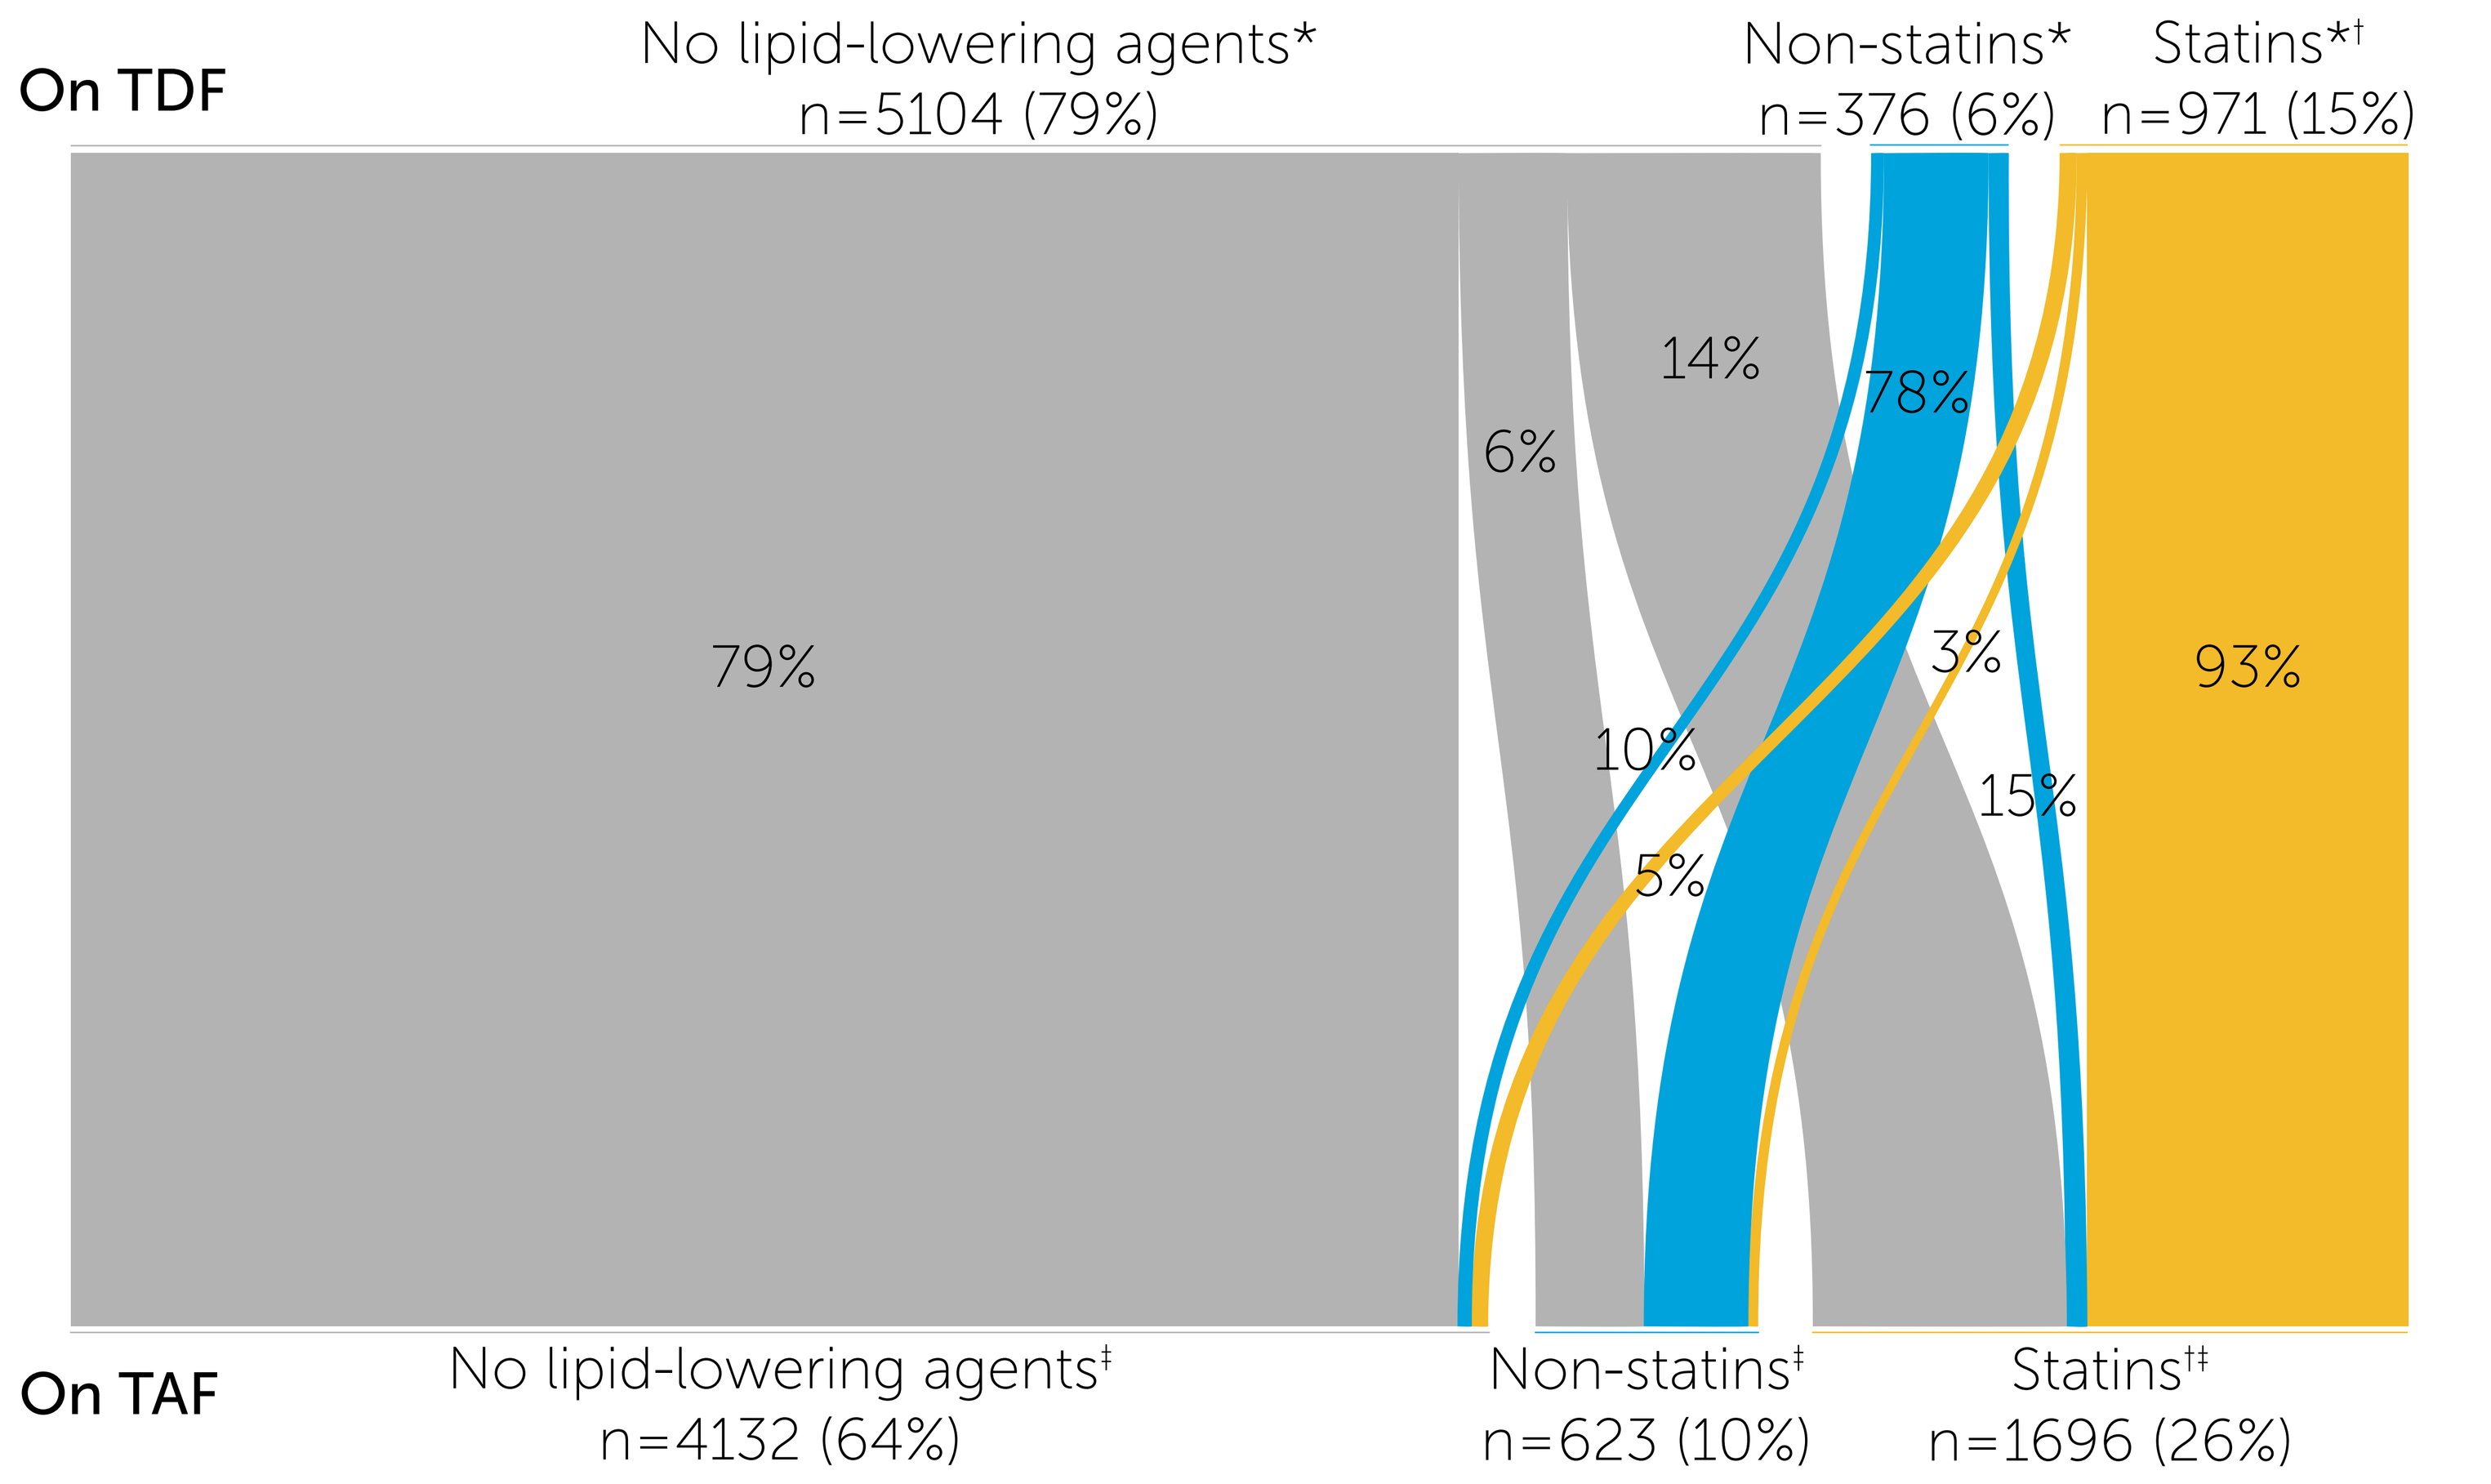

On TDF
No lipid-lowering agents*
n=5104 (79%)
Non-statins*
n=376 (6%)
Statins*†
n=971 (15%)
79%
6%
14%
78%
10%
5%
3%
15%
93%
On TAF
No lipid-lowering agents‡
n=4132 (64%)
Non-statins‡
n=623 (10%)
Statins†‡
n=1696 (26%)

* Lipid lowering agent use assessed at time of lipid panel on TDF
† Statins used with or without other lipid lowering agents
‡ First lipid lowering agent use on/after first lipid panel on TAF

Figure 3. Risk of ASCVD in PLWH switching from TDF to TAF with an ASCVD† risk score

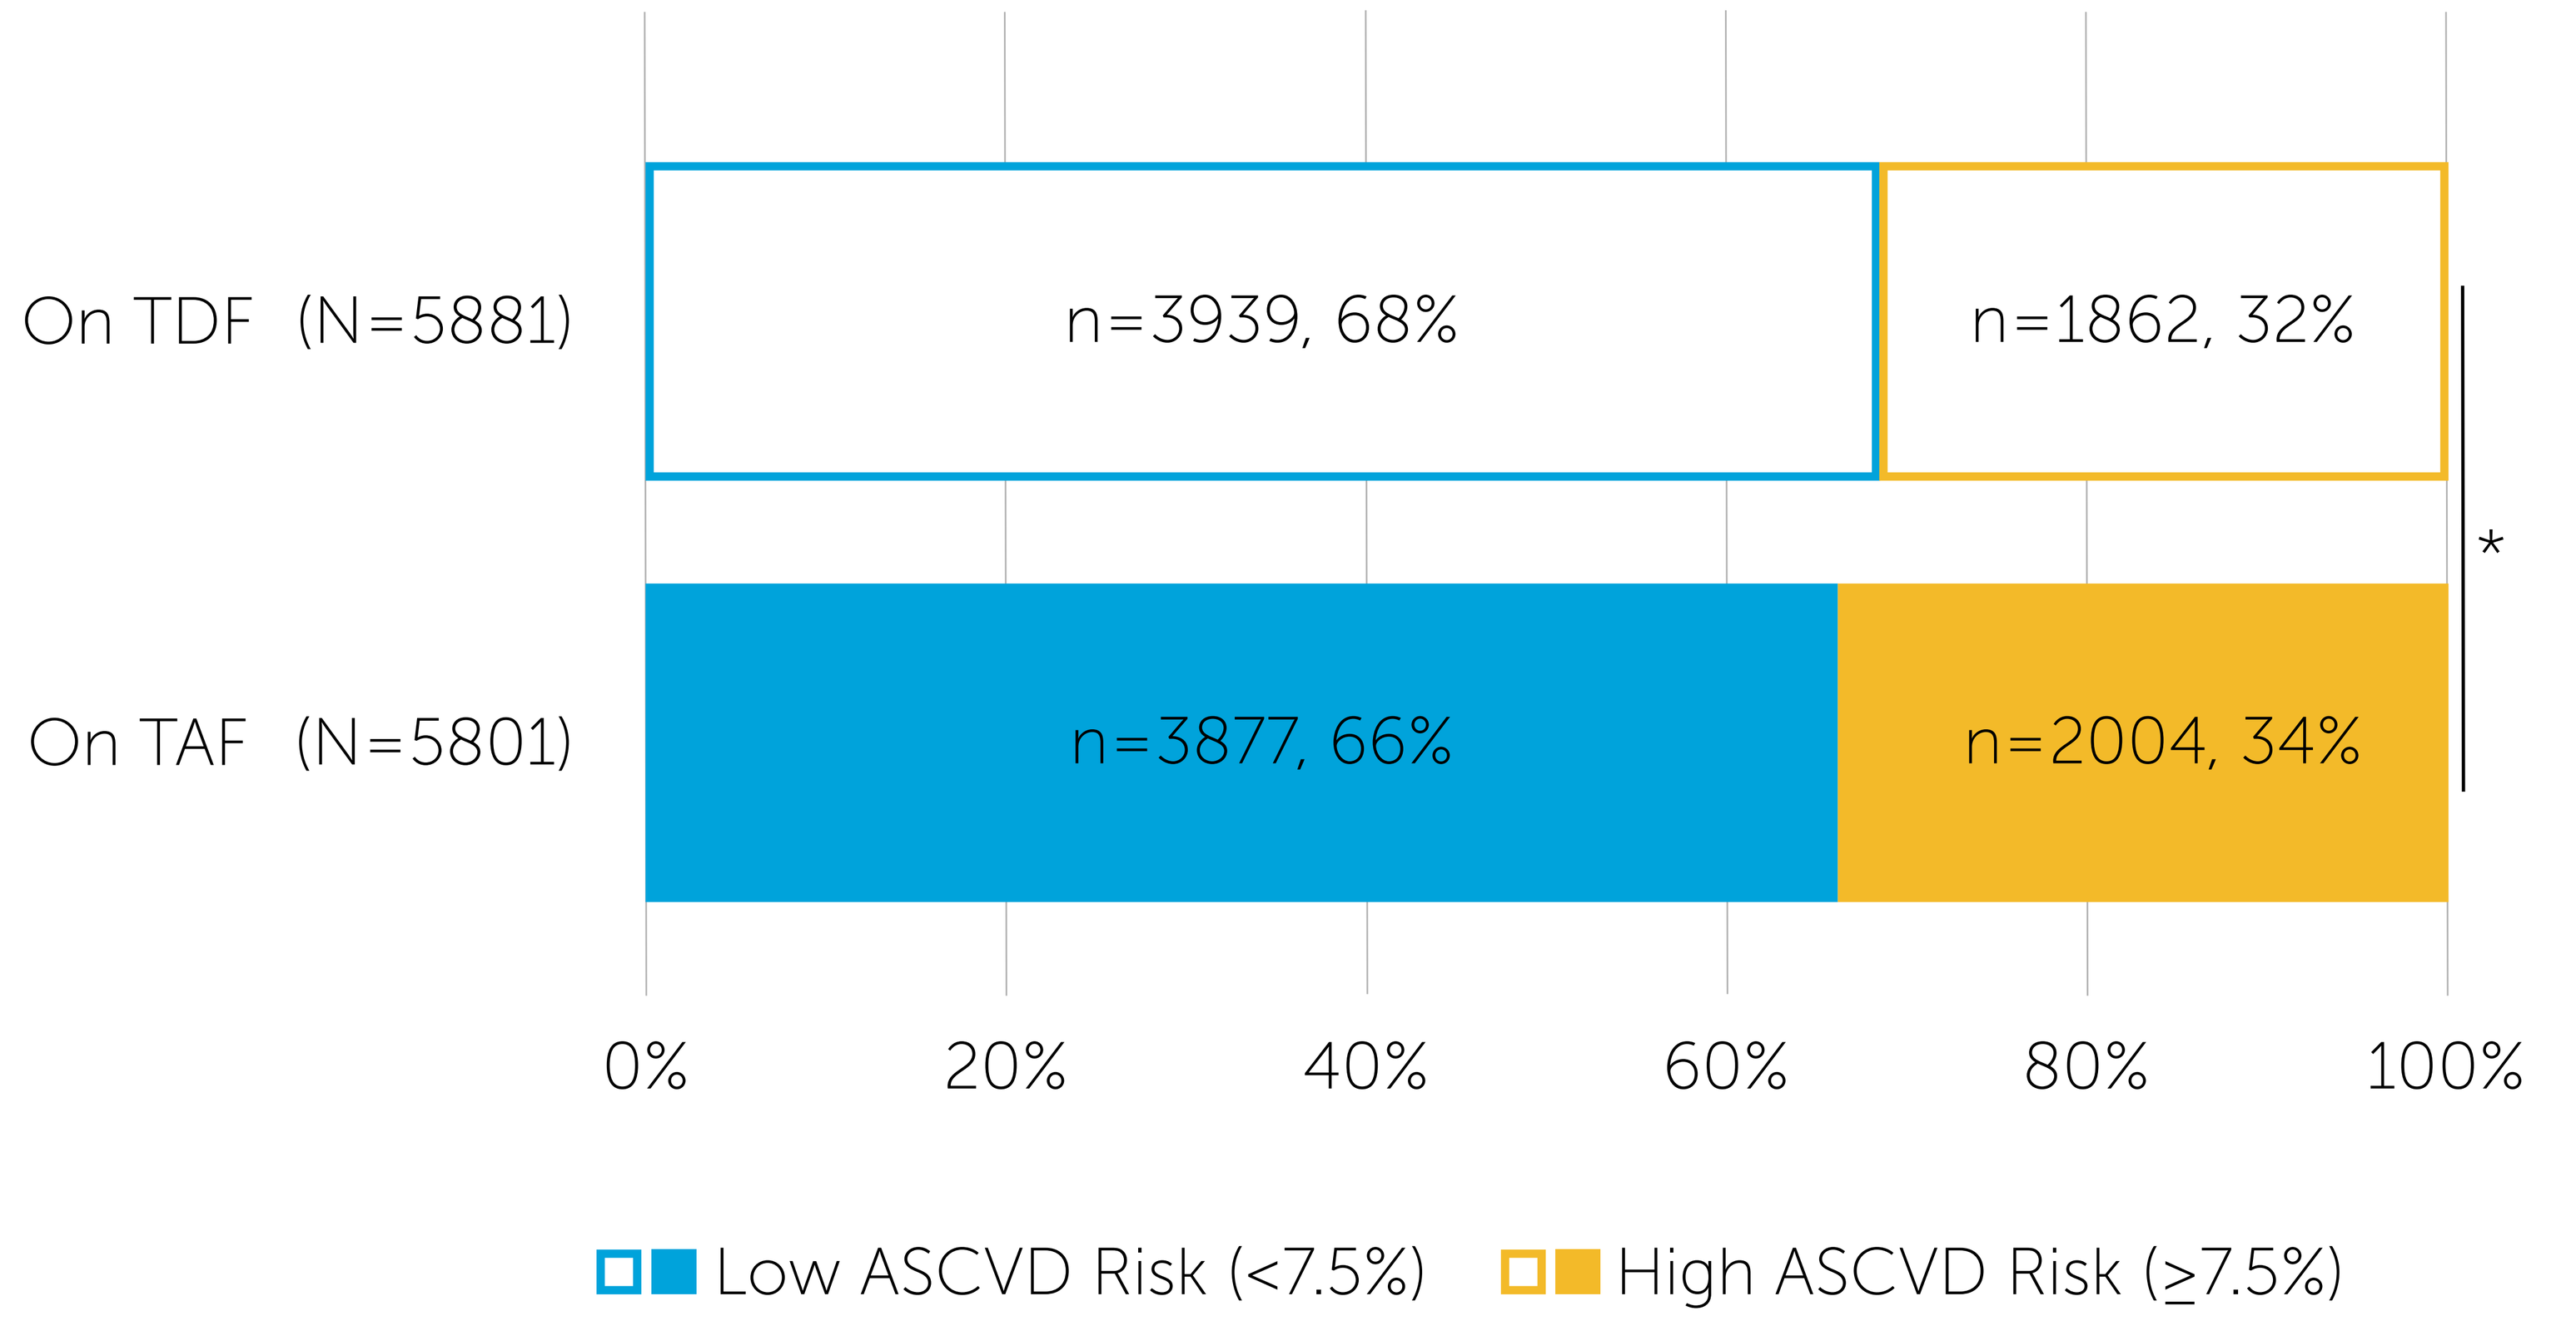

On TDF (N=5881)
n=3939, 68%
n=1862, 32%
On TAF (N=5801)
n=3877, 66%
n=2004, 34%
*
0%
20%
40%
60%
80%
100%
Low ASCVD Risk (<7.5%)
High ASCVD Risk (≥7.5%)

*p-value = 0.02
†All factors included in the ASCVD score calculation were measured within 6 months before the lipid panel

Figure 4. Statin use by ASCVD* risk in PLWH switching from TDF to TAF

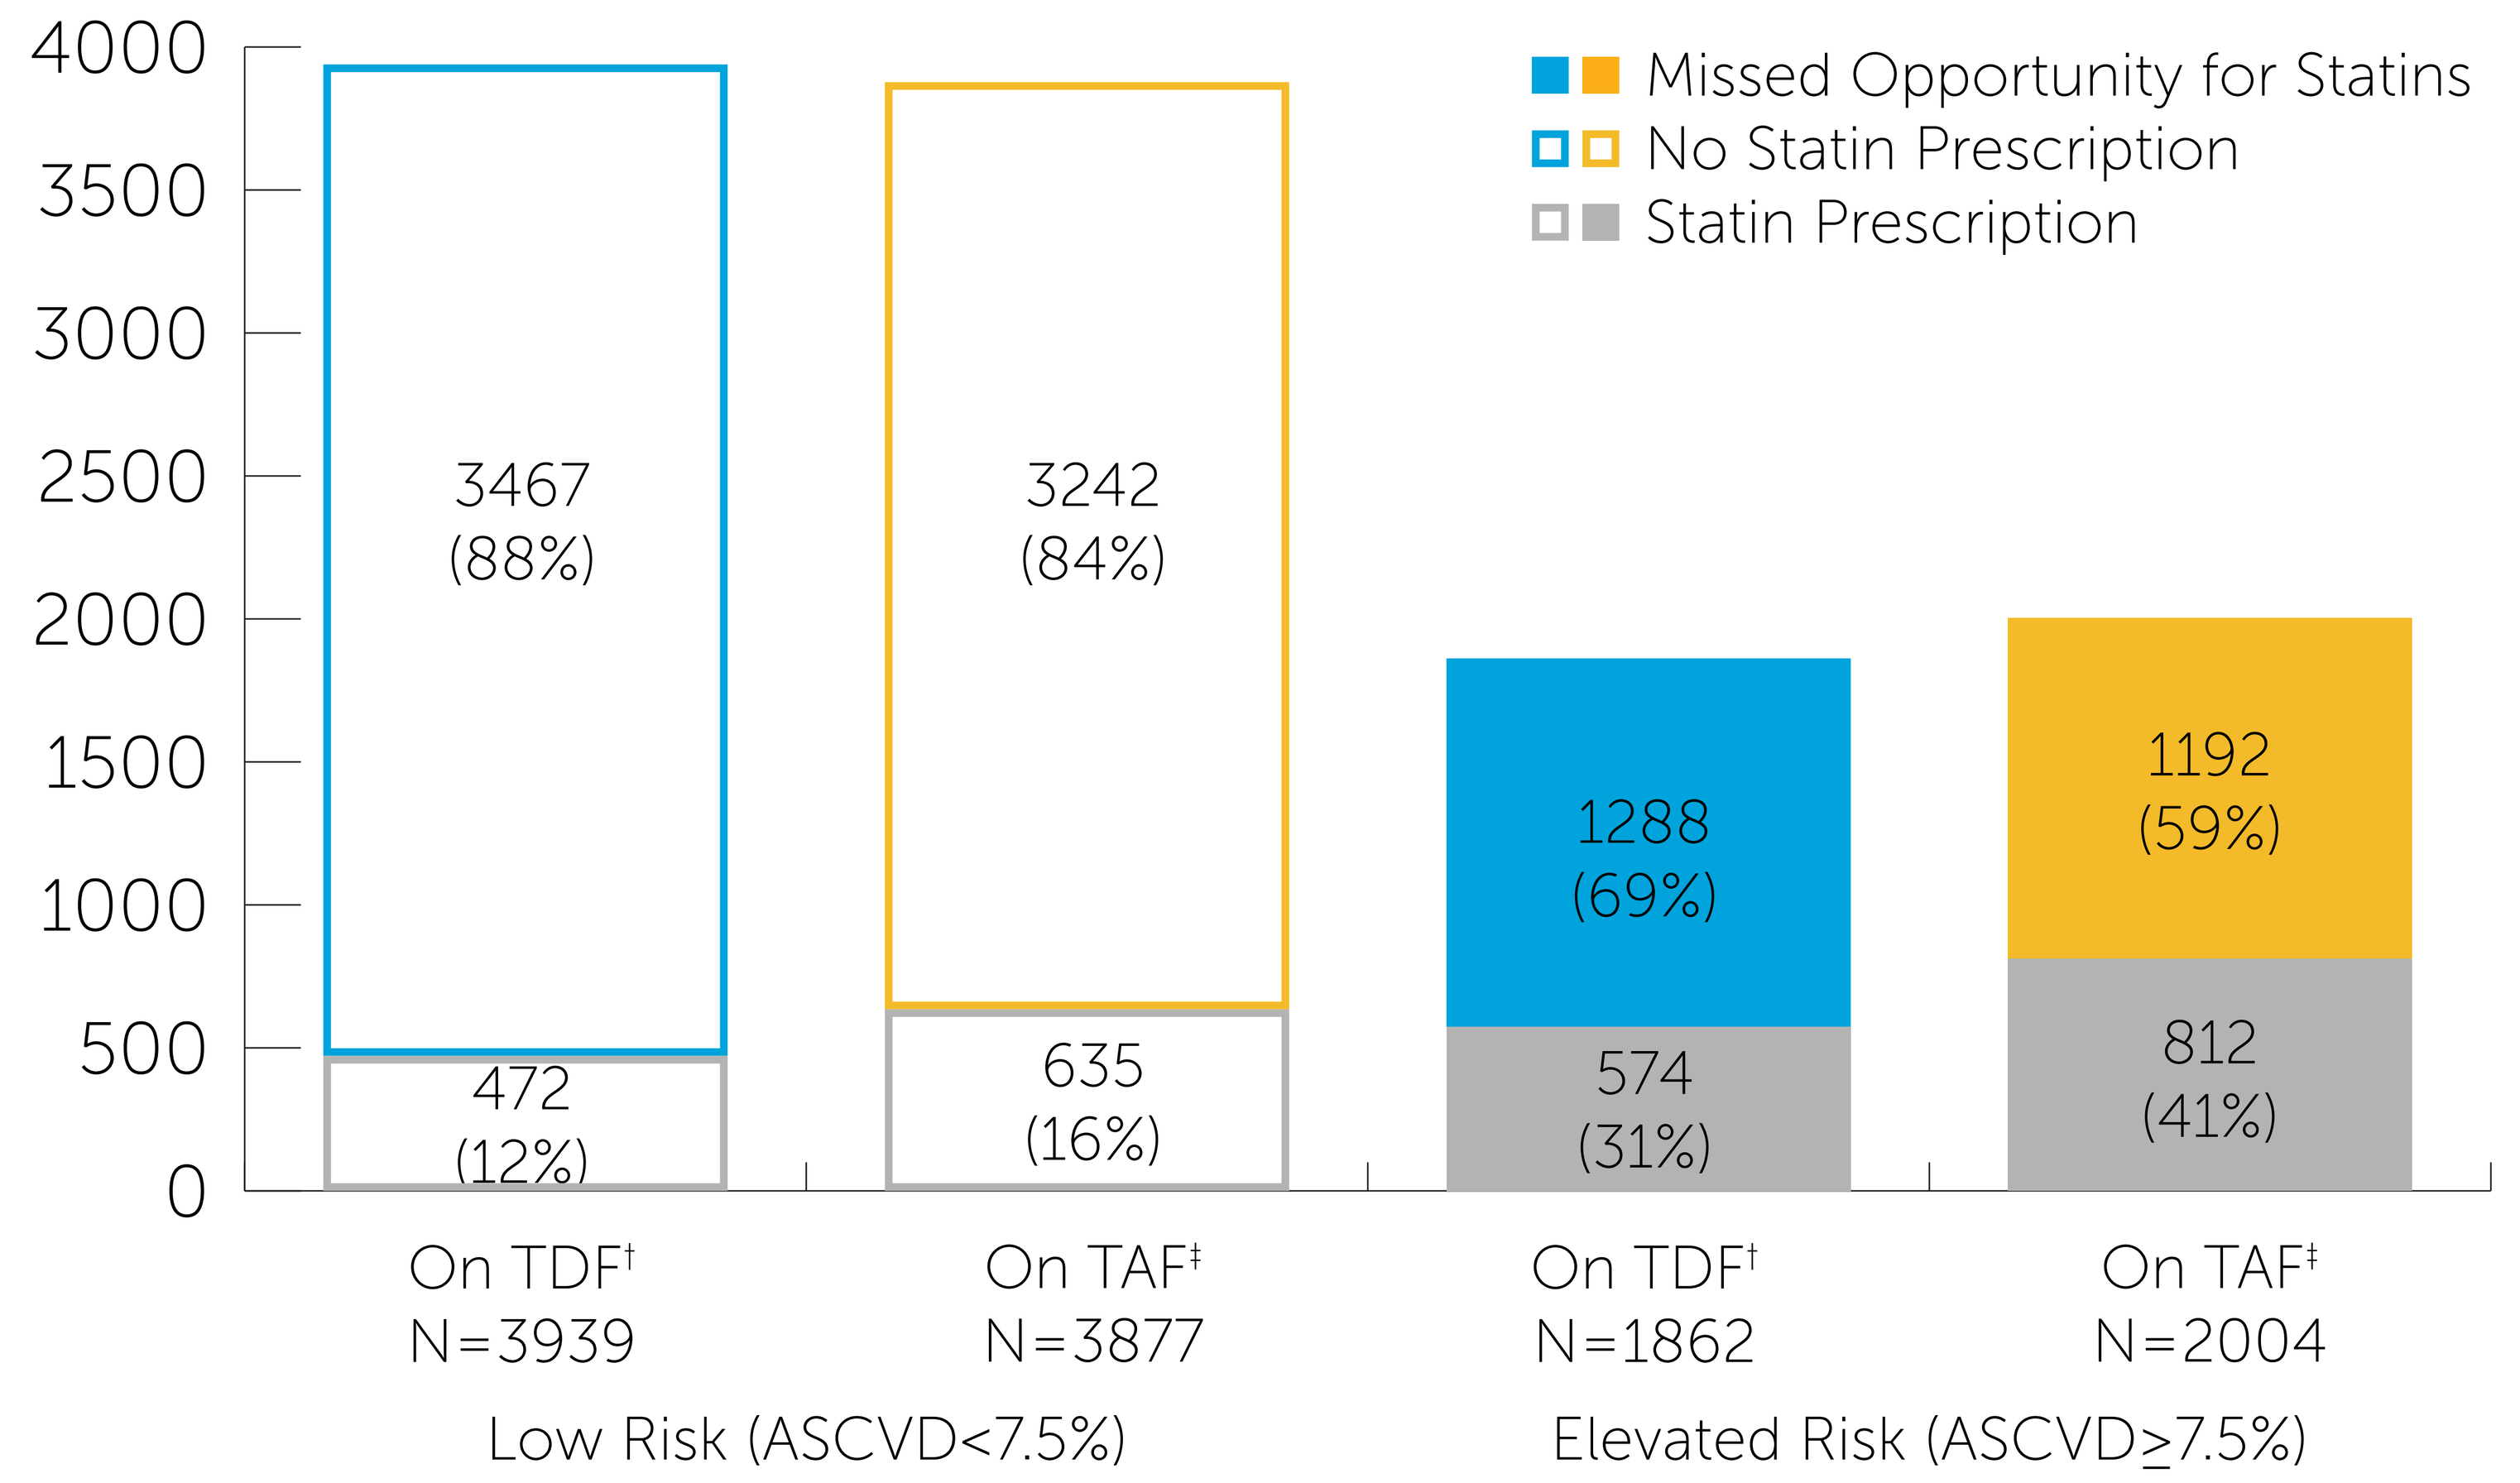

4000
3500
3000
2500
2000
1500
1000
500
0
Missed Opportunity for Statins
No Statin Prescription
Statin Prescription
3467 (88%)
472 (12%)
3242 (84%)
635 (16%)
1288 (69%)
574 (31%)
1192 (59%)
812 (41%)
On TDF† N=3939
On TAF‡ N=3877
On TDF† N=1862
On TAF‡ N=2004
Low Risk (ASCVD<7.5%)
Elevated Risk (ASCVD≥7.5%)

*All factors included in the ASCVD score calculation were measured within 6 months before the lipid panel
†Statin prescription at or after the last lipid panel on TDF
‡Statin prescription at or after the first lipid panel on TAF

Discussion

- Changes in regimens represent an opportunity for healthcare providers to assess health markers and address clinical concerns
- A third of PLWH had an elevated ASCVD risk at the time of switch from TDF to TAF
- Failure to initiate statins in PLWH with an ASCVD ≥7.5% at switch represents a missed opportunity for statin initiation
 - 69% had no statin prescription before switch
 - 59% had no statin prescription after switch
- Considerable missed opportunity given that the Pooled Cohort Equations calculator may underestimate risk among PLWH due to non-traditional risk factors driving ASCVD[3]
- Comparable results among PLWH who did not require ASCVD score imputation (data not shown)

KEY FINDINGS

- Only an additional ~10% of PLWH with elevated ASCVD risk initiated statins after switching from TDF to TAF
- Considerable missed opportunities to reduce risk of cardiovascular disease in this at-risk population

References

1. Mallon P, Brunet L, Fusco J, et al. Changes in Lipids After a Direct Switch from TDF to TAF. CROI Conference on Retroviruses and Opportunistic Infections. Seattle, WA, 2019.
2. Stone NJ, Robinson JG, Lichtenstein AH, et al. 2013 ACC/AHA Guideline on the Treatment of Blood Cholesterol to Reduce Atherosclerotic Cardiovascular Risk in Adults: A Report of the American College of Cardiology/American Heart Association Task Force on Practice Guidelines. Journal of the American College of Cardiology 2014; 63(25, Part B): 2889-934.
3. Zanni MV, Fitch KV, Feldpausch M, et al. 2013 American College of Cardiology/American Heart Association and 2004 Adult Treatment Panel III cholesterol guidelines applied to HIV-infected patients with/without subclinical high-risk coronary plaque. AIDS (London, England) 2014; 28(14): 2061-70.

Acknowledgements

This research would not be possible without the generosity of people living with HIV and their OPERA caregivers. Additionally, we are grateful for the following individuals: Robin Beckerman (SAS programming), Jeff Briney (QA), Bernie Stooks (Database Arch & Mgmt), Judy Johnson (Med Terminology Classification), Rodney Mood (Site Support), & Antonin Fusco (Poster Design).

Support

This research was funded by Merck Sharp & Dohme Corp., a subsidiary of Merck & Co., Inc., Kenilworth, NJ, USA

MERCK

Epividian®

https://bit.ly/2MhxR8f

IDWeek 2019, Washington, DC, 2-6 October 2019