POSTER #555

Characteristics of HIV+ Patients Prescribed Raltegravir QD in the United States

Philip Lackey[1], Kathy L. Schulman[2], Jennifer Fusco[2], Jean Marie Arduino[3], Girish Prajapati[3], Gregory Fusco[2]

[1]Atrium Health, Charlotte, NC; [2]Epividian, Inc., Durham, NC; [3]Merck & Co., Inc., Kenilworth, NJ, USA

OPERA® The Longitudinal Cohort

Contact Information:
4505 Emperor Blvd., Suite 220 Durham, NC 27703
P: 224-632-1863 F: 919-313-4505
Email: jennifer.fusco@epividian.com

BACKGROUND

- Raltegravir (RAL) 400 mg twice-daily (RAL BID) has been an integral part of antiretroviral therapy (ART) in both ART naïve and experienced HIV-1 infected patients for the last decade
- RAL BID has demonstrated long-term efficacy in both ART naïve and experienced HIV-1 infected patients with favorable safety/tolerability profile and low risk of drug-drug interactions[1,2,3]
- RAL 1200 mg (2 x 600 mg tablets), a once-daily formulation (RAL QD), demonstrated similar efficacy (durable up to 96 weeks) and safety profile as RAL BID with convenience of once-daily dosing and was approved in 2017[4]

OBJECTIVE:

To characterize the early utilization of RAL QD in the United States

METHODS

STUDY POPULATION

- Observational clinical cohort analysis utilizing prospectively collected electronic medical record (EMR) data obtained from the Observational Pharmaco-Epidemiology Research and Analysis (OPERA) database, following more than 80,000 patients from 84 clinics across 17 states throughout the U.S.
- Inclusion criteria: HIV-1 positive patients ≥18 years of age prescribed RAL QD between July 1, 2017 and June 30, 2018
- Exclusion criteria: Patients co-infected with HIV-2 or who previously initiated RAL QD as part of a clinical trial
- Data have been updated to reflect new users of RAL QD through June 30, 2018

ANALYSIS

- Overall and stratified analyses were conducted
- Patient demographics were described as of the date of RAL QD initiation
- Clinical and treatment characteristics were described using a baseline period of 12 months
- Patients were stratified based on baseline treatment status (treatment-naïve, treatment experienced-RAL BID, treatment-experienced-other ART regimens)

RESULTS

- A total of 322 patients were prescribed RAL QD during the study period. RAL QD was most often prescribed in combination with emtricitabine/tenofovir alafenamide (44.1%) followed by emtricitabine/tenofovir disoproxil fumarate (11.5%), abacavir/lamivudine (7.1%) and darunavir/cobicistat (5.6%)
- Sixteen patients (5%) were ART naïve, 105 (33%) switched from non-RAL based regimens, and 201 patients (62%) switched from RAL BID
- Among patients switching from RAL BID to RAL QD, most (77.6%) had no other regimen changes other than switching to RAL QD

- Patient demographic characteristics at the time of RAL QD initiation are shown in Figure 1, stratified by baseline treatment status. The majority of patients were ≥50 years of age (57.8%), male (77.6%), and not Hispanic (82.0%). A total of 44.1% were African American.

Figure 1. Demographic Characteristics of Raltegravir QD Initiators, by Baseline Treatment Status*

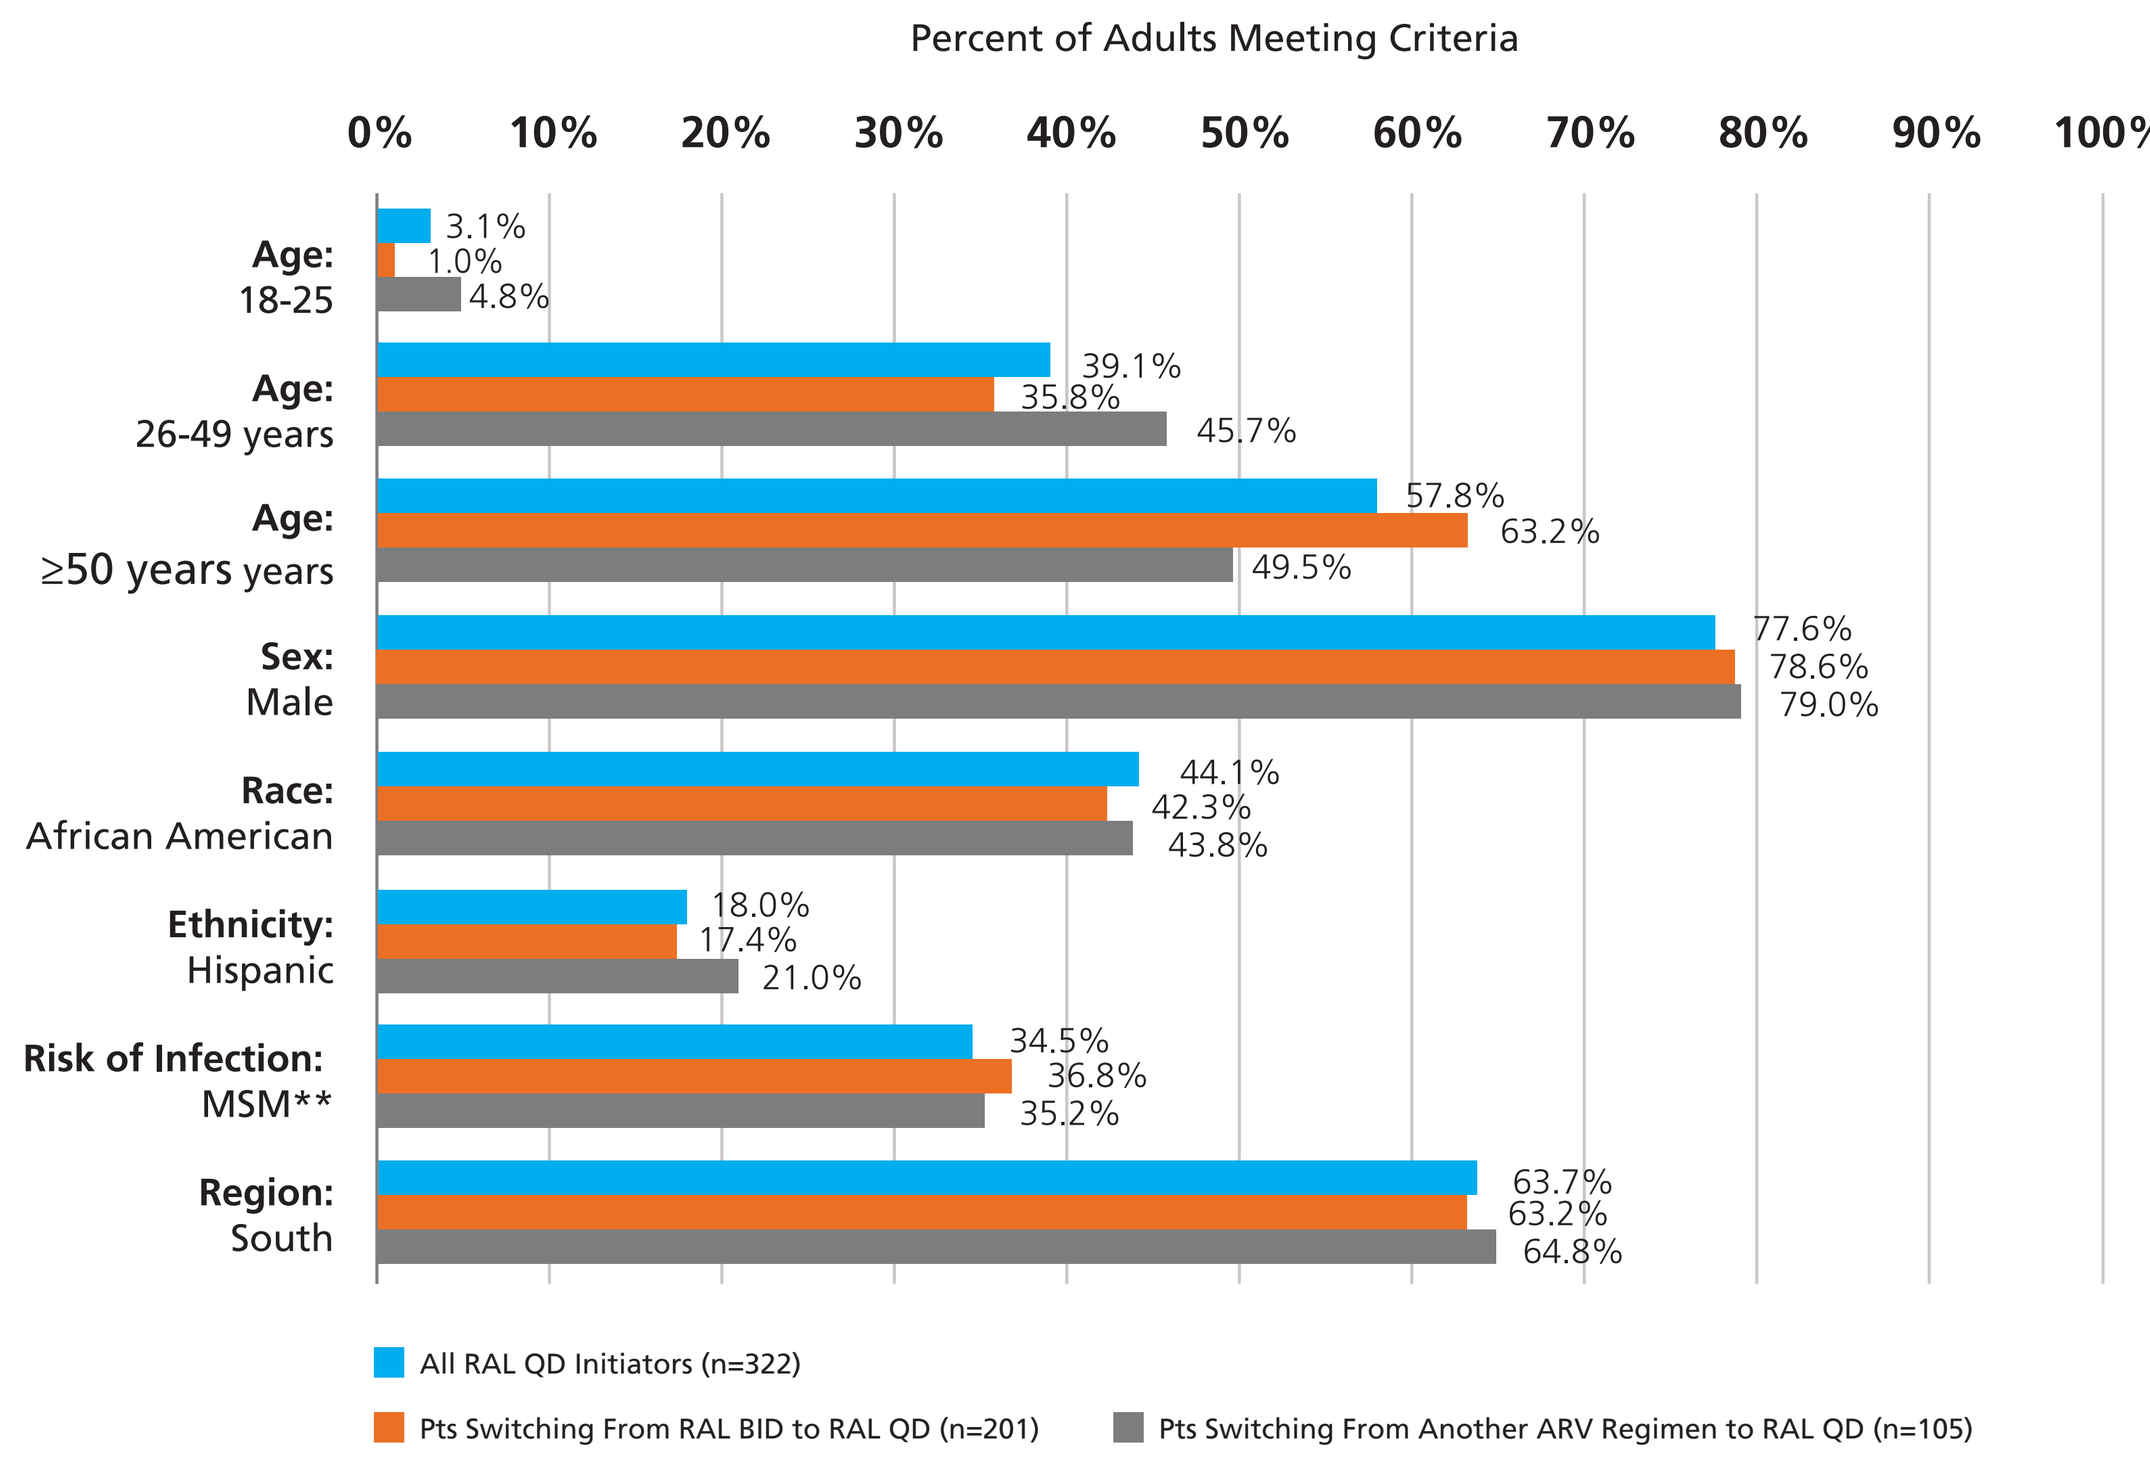

Percent of Adults Meeting Criteria
0% 10% 20% 30% 40% 50% 60% 70% 80% 90% 100%
Age: 18-25 3.1% 1.0% 4.8%
Age: 26-49 years 39.1% 35.8% 45.7%
Age: ≥50 years years 57.8% 63.2% 49.5%
Sex: Male 77.6% 78.6% 79.0%
Race: African American 44.1% 42.3% 43.8%
Ethnicity: Hispanic 18.0% 17.4% 21.0%
Risk of Infection: MSM** 34.5% 36.8% 35.2%
Region: South 63.7% 63.2% 64.8%
All RAL QD Initiators (n=322)
Pts Switching From RAL BID to RAL QD (n=201)
Pts Switching From Another ARV Regimen to RAL QD (n=105)

*ART Naive Patients (n=16) not shown separately §Classification based solely on the regimen immediately prior to initiation of RAL QD. Patients classified as other ARV (non-RAL) may have been prescribed RAL BID at a previous point in time. **Men who have sex with men

- Figure 2 depicts the virologic status among ART experienced patients at the time of RAL QD initiation, overall and stratified by baseline treatment status. Table 1 presents other key, HIV specific, clinical characteristics.

Figure 2: Baseline Virologic Status of ART Experienced Patients Initiating Raltegravir QD

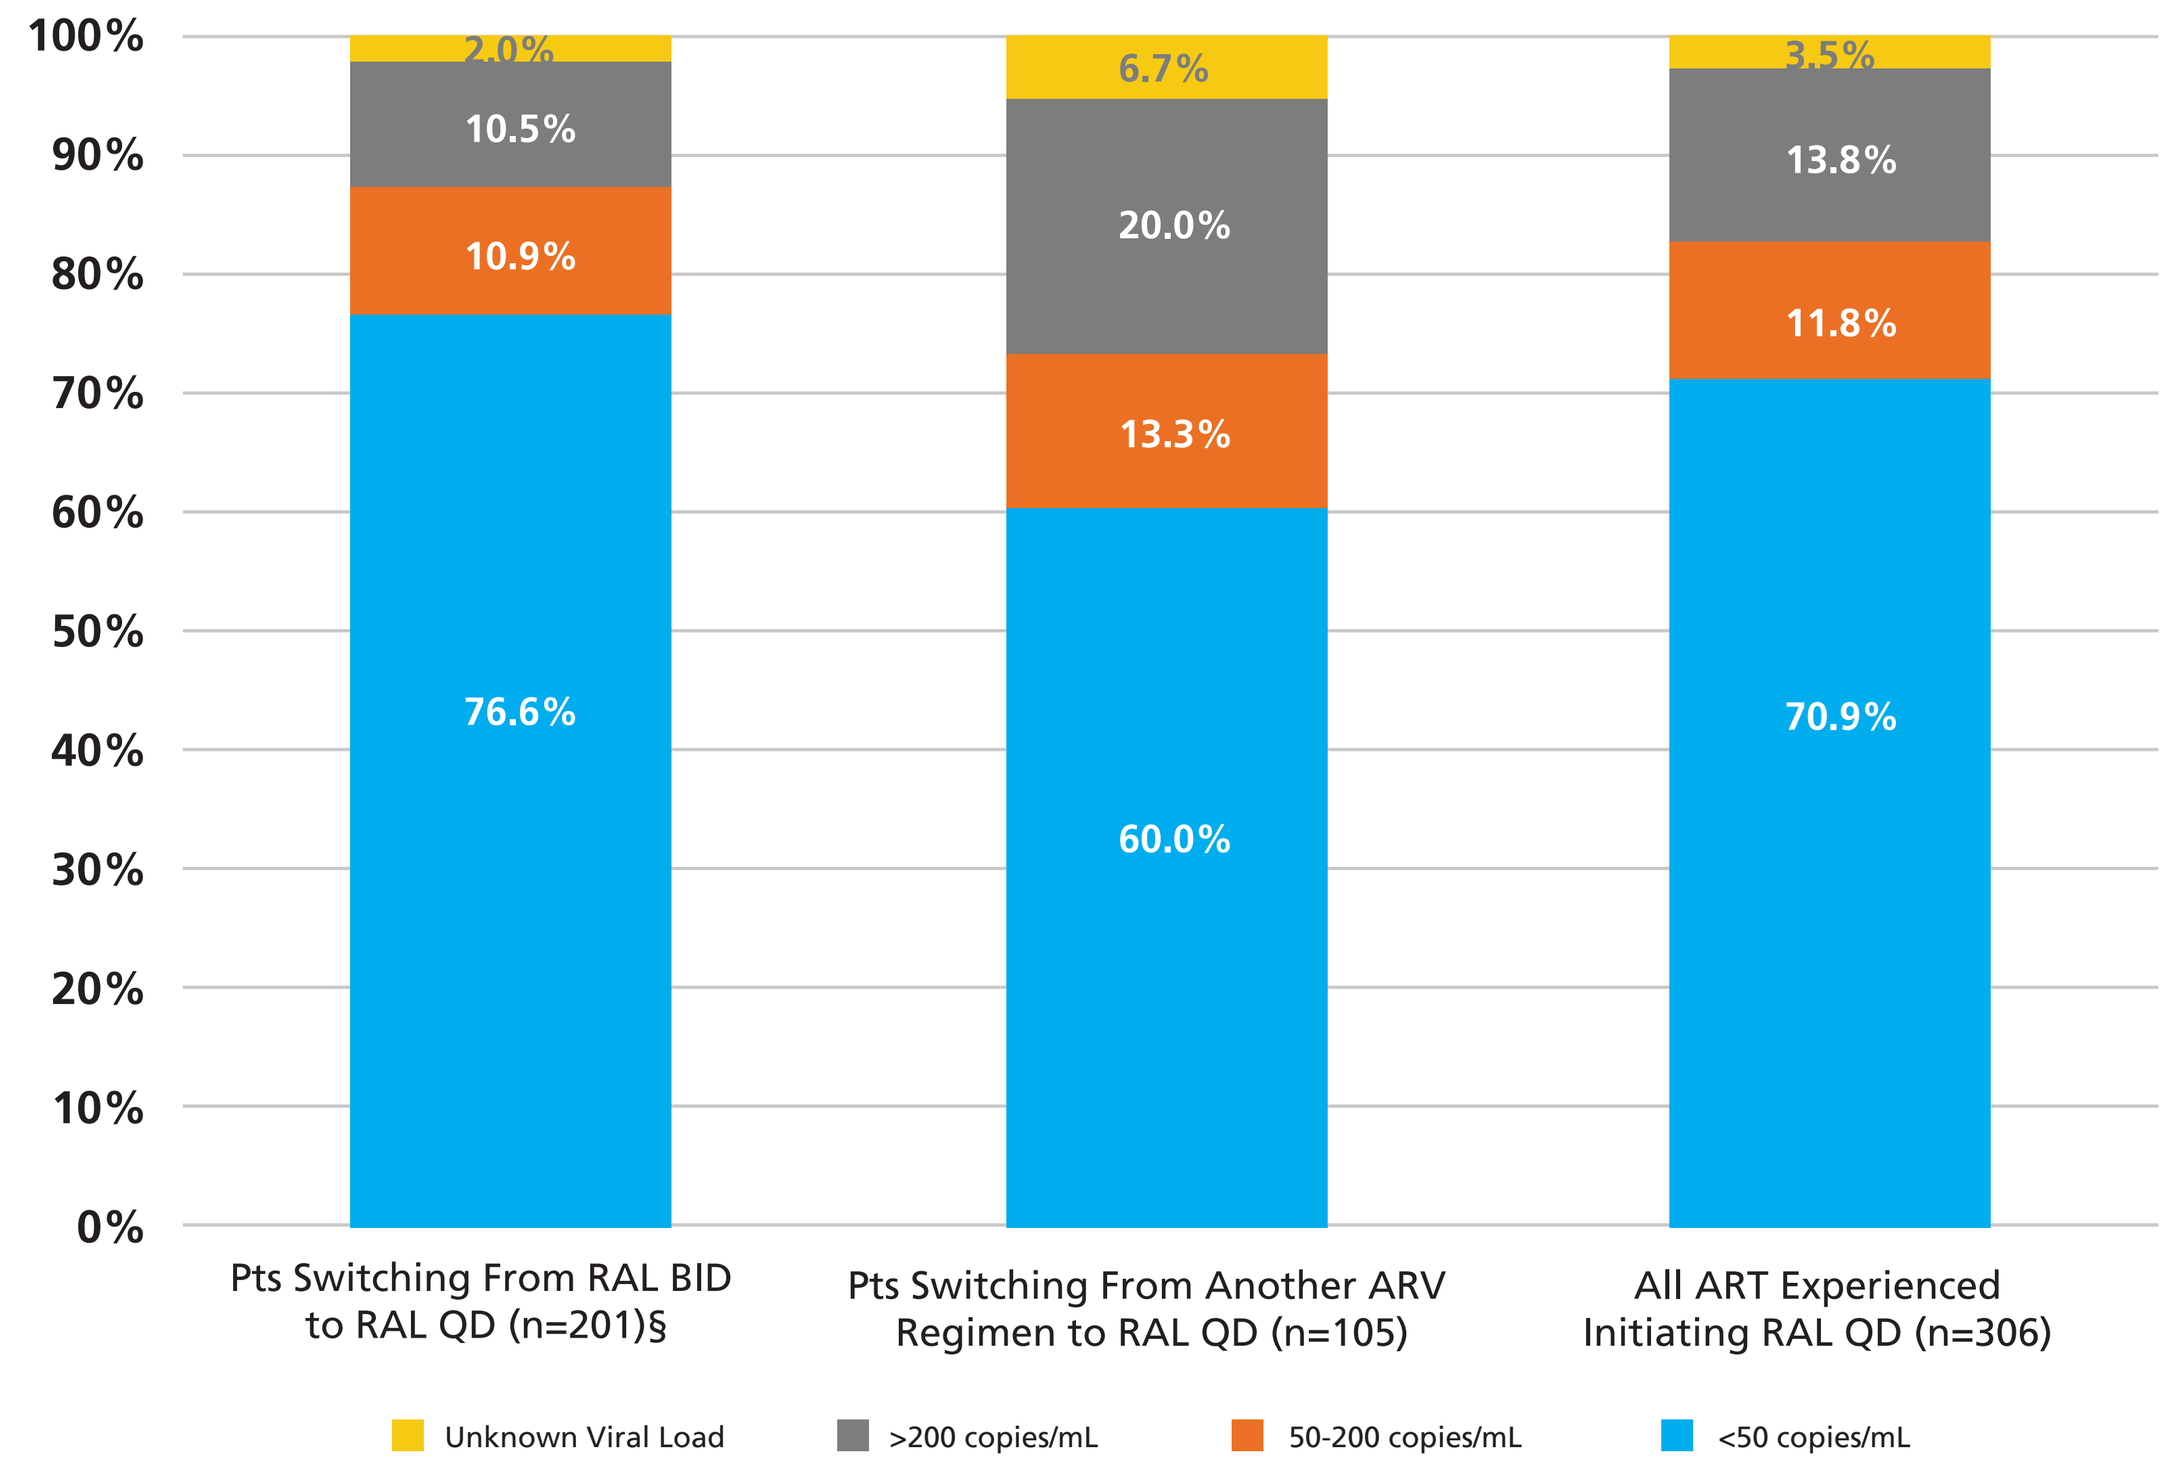

100% 90% 80% 70% 60% 50% 40% 30% 20% 10% 0%
2.0% 10.5% 10.9% 76.6%
6.7% 20.0% 13.3% 60.0%
3.5% 13.8% 11.8% 70.9%
Pts Switching From RAL BID to RAL QD (n=201)§
Pts Switching From Another ARV Regimen to RAL QD (n=105)
All ART Experienced Initiating RAL QD (n=306)
Unknown Viral Load
>200 copies/mL
50-200 copies/mL
<50 copies/mL

§Classification based solely on the regimen immediately prior to initiation of RAL QD. Pts classified as other ARV (non-RAL) may have been prescribed RAL BID at a previous point in time.

Table 1: HIV Specific Clinical Characteristics of Raltegravir QD Initiators, by Baseline Treatment Status*

	Pts Switching from RAL BID to RAL QD (*n*=201)§	Pts Switching from Another ARV Regimen to RAL QD (*n*=105)§	All RAL QD Initiators (*n*=322)
Duration Prior ARV Regimen (Days), Median (IQR)	391.0 (171.0-772.0)	271.0 (77.0-489.0)	364.5 (132.0-631.0)
Prior Core Agent Class, n (%)**			
INSTI-based	201 (100.0%)	43 (41.0%)	244 (79.7%)
NNRTI-based	27 (13.4%)	20 (19.0%)	47 (15.4%)
PI-based	57 (28.4%)	25 (23.8%)	82 (26.8%)
Other Core Agent Class	1 (0.5%)	0 (0.0%)	1 (0.3%)
Unknown Core Agent Class	0 (0.0%)	27 (25.7%)	27 (8.8%)
History of AIDS Defining Illness, n (%)			
Any Time Prior to RAL QD Initiation	60 (29.9%)	35 (33.3%)	99 (30.7%)
In the 12-months prior to RAL QD Initiation	4 (2.0%)	11 (10.5%)	19 (5.9%)
CD4 Count (cells/μL)			
Median (IQR)	634 (455-858)	513 (351-774)	590 (376-822)
CD4 Count Category (cells/μL), n (%)			
High (>500)	137 (68.2%)	48 (45.7%)	189 (58.7%)
Moderate (>350 to <=500)	28 (13.9%)	24 (22.9%)	55 (17.1%)
Low (<=350)	31 (15.4%)	24 (22.9%)	64 (19.9%)
VACS†, Median (IQR)	18.0 (11.0-34.0)	18.0 (10.0-31.0)	18.0 (10.0-34.5)

*ART Naive Patients (n=16) not shown separately §Classification based solely on the regimen immediately prior to initiation of RAL QD. Patients classified as other ARV (non-RAL) may have been prescribed RAL BID at a previous point in time. †VACS Mortality Index: Scored by summing pre-assigned points for age, CD4 count, HIV-1 RNA, hemoglobin, platelets, aspartate and alanine transaminase, creatinine, and viral hepatitis C infection. A higher score is associated with a higher risk of 5-year all-cause mortality. **Categories are not mutually exclusive as patients may have exposure to multiple core agents

- 86% of patients had at least one of the comorbidities depicted in Figure 3 with hypertension being the most common (45.7%) followed by hyperlipidemia (41.3%), anemia (26.7%), anxiety disorders (25.2%) and diabetes (19.6%)
- At the time of RAL QD initiation, almost two-thirds of the study population had ≥2 comorbidities, 28.9% had ≥4 comorbidities and 7.5% had ≥6 of the comorbidities detailed in Figure 3. The distribution of comorbidity frequency in the study population is illustrated in Figure 4.

Figure 3: Baseline Comorbidity In RAL QD Initiators

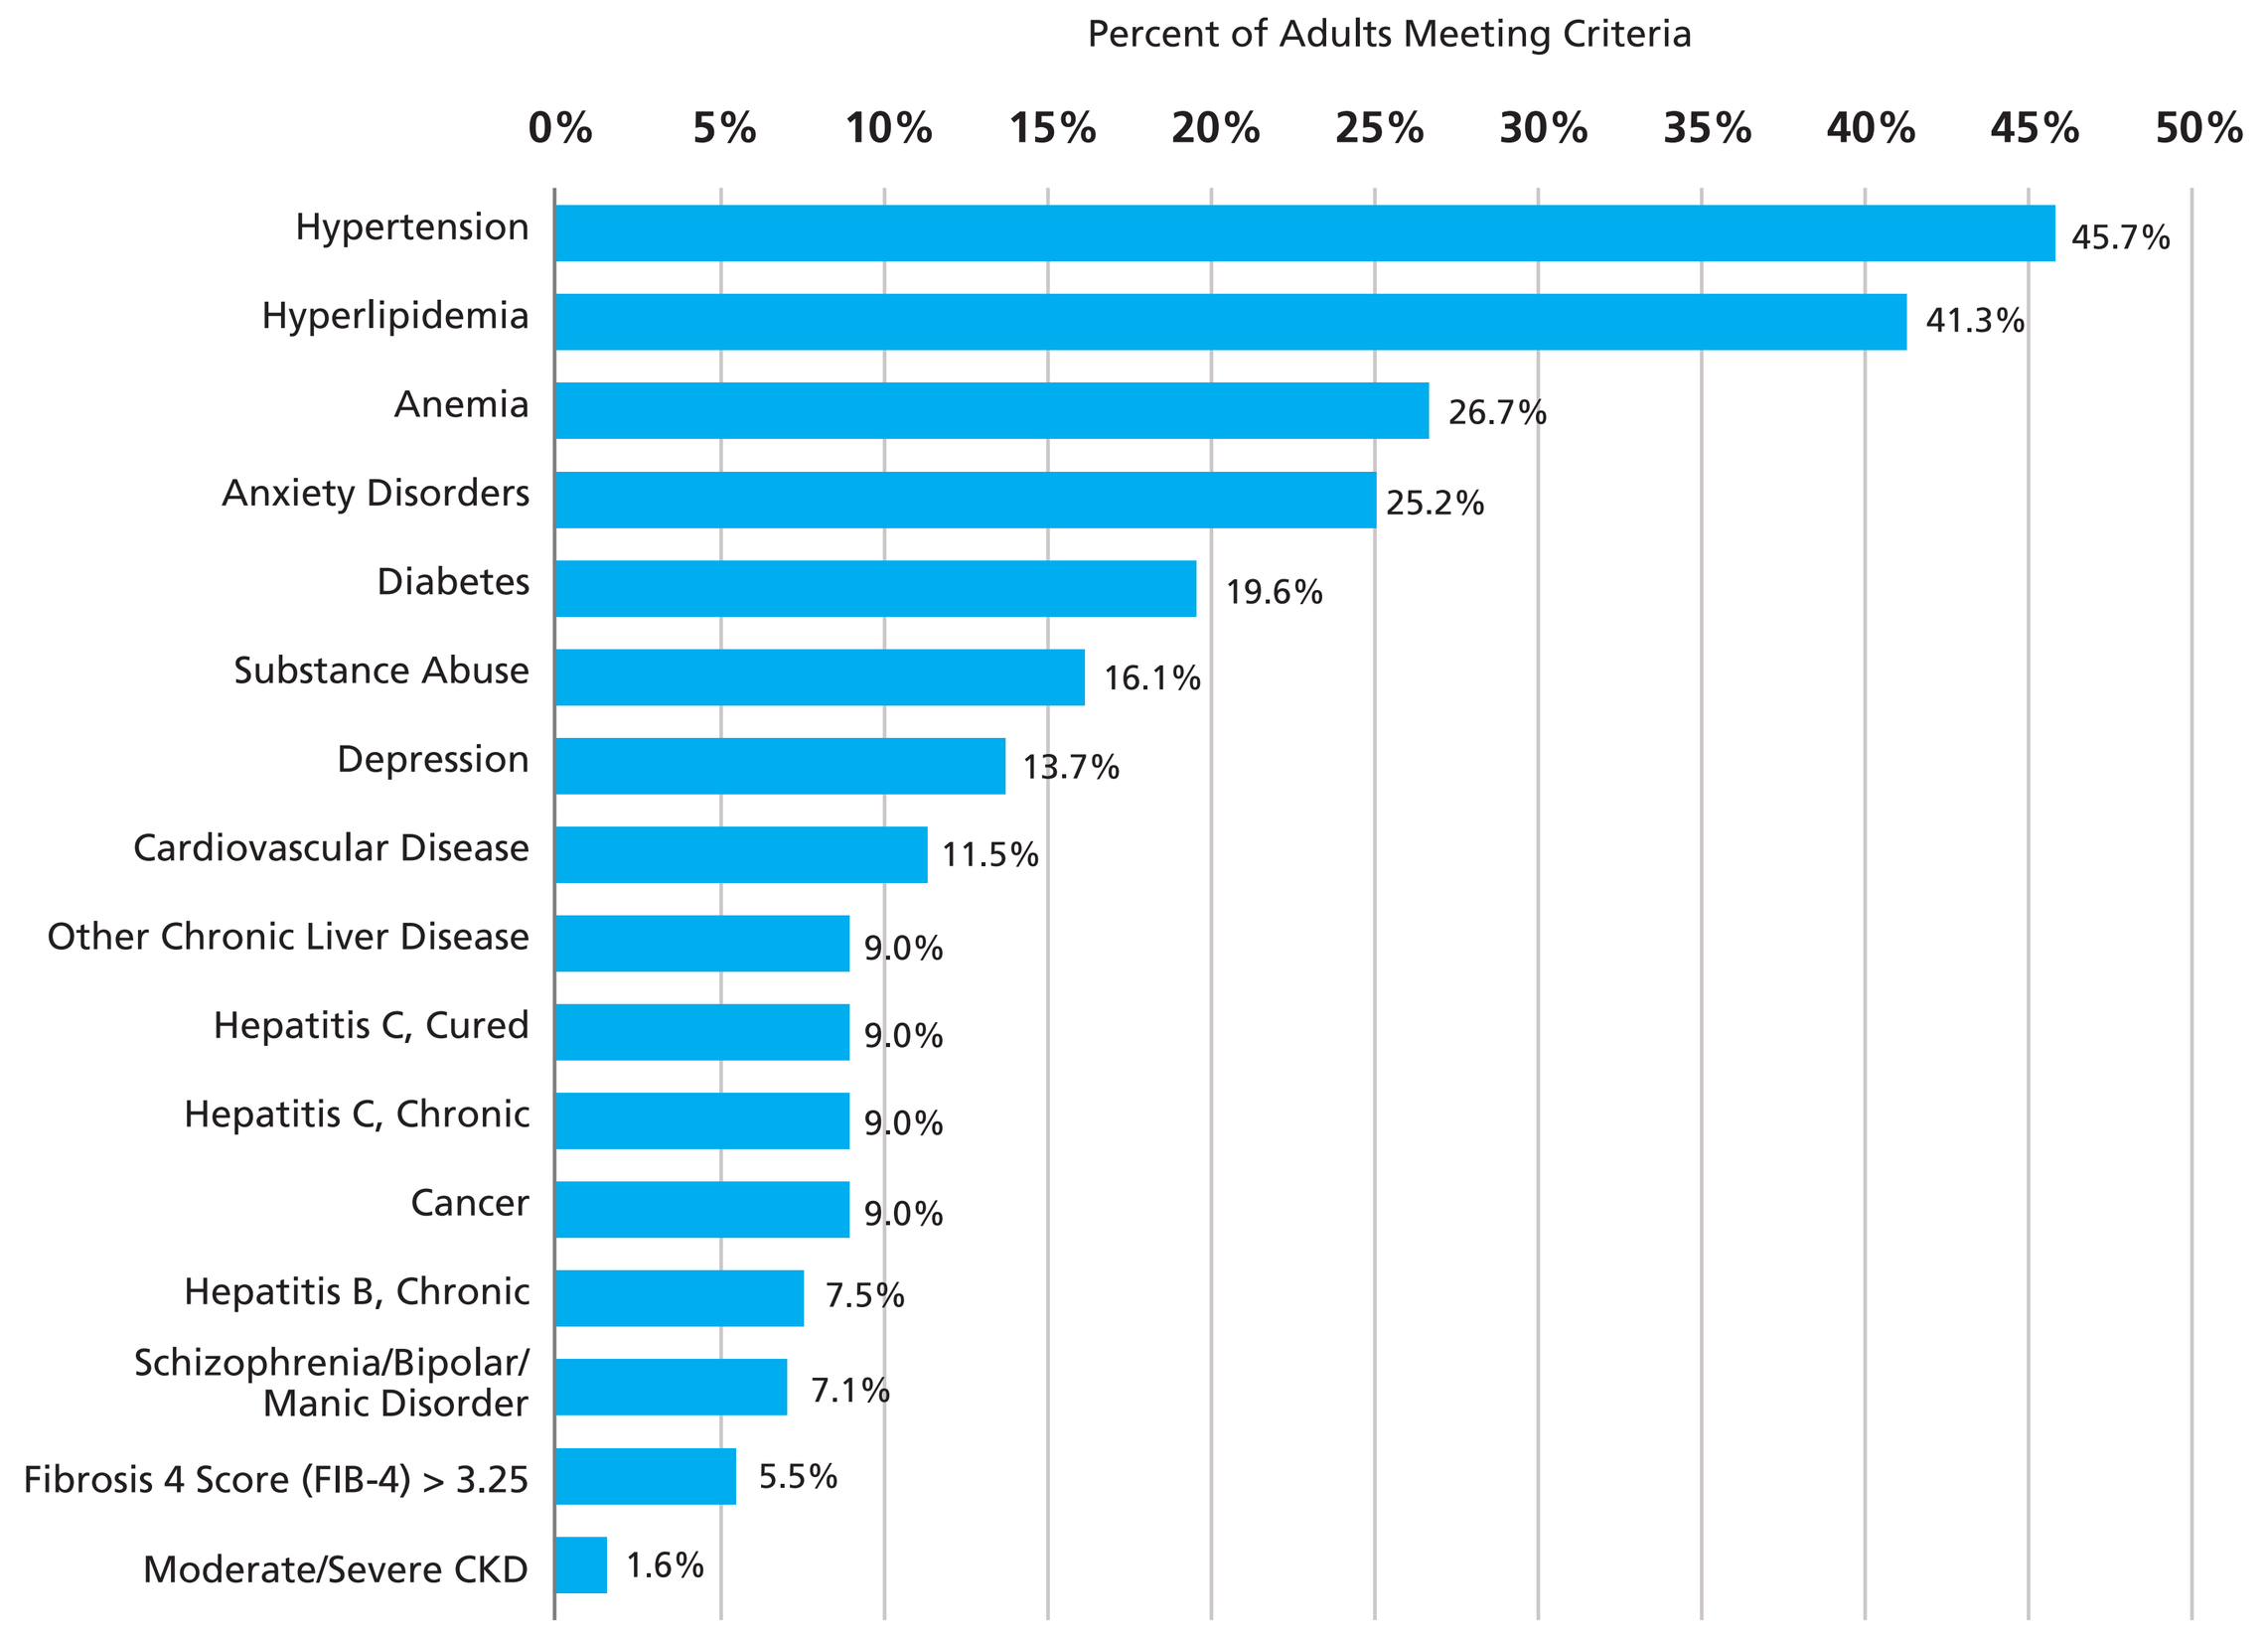

Percent of Adults Meeting Criteria
0% 5% 10% 15% 20% 25% 30% 35% 40% 45% 50%
Hypertension 45.7%
Hyperlipidemia 41.3%
Anemia 26.7%
Anxiety Disorders 25.2%
Diabetes 19.6%
Substance Abuse 16.1%
Depression 13.7%
Cardiovascular Disease 11.5%
Other Chronic Liver Disease 9.0%
Hepatitis C, Cured 9.0%
Hepatitis C, Chronic 9.0%
Cancer 9.0%
Hepatitis B, Chronic 7.5%
Schizophrenia/Bipolar/ Manic Disorder 7.1%
Fibrosis 4 Score (FIB-4) > 3.25 5.5%
Moderate/Severe CKD 1.6%

Figure 4: Count of Baseline Comorbidities Among Raltegravir QD Initiators, by Baseline Treatment Status*

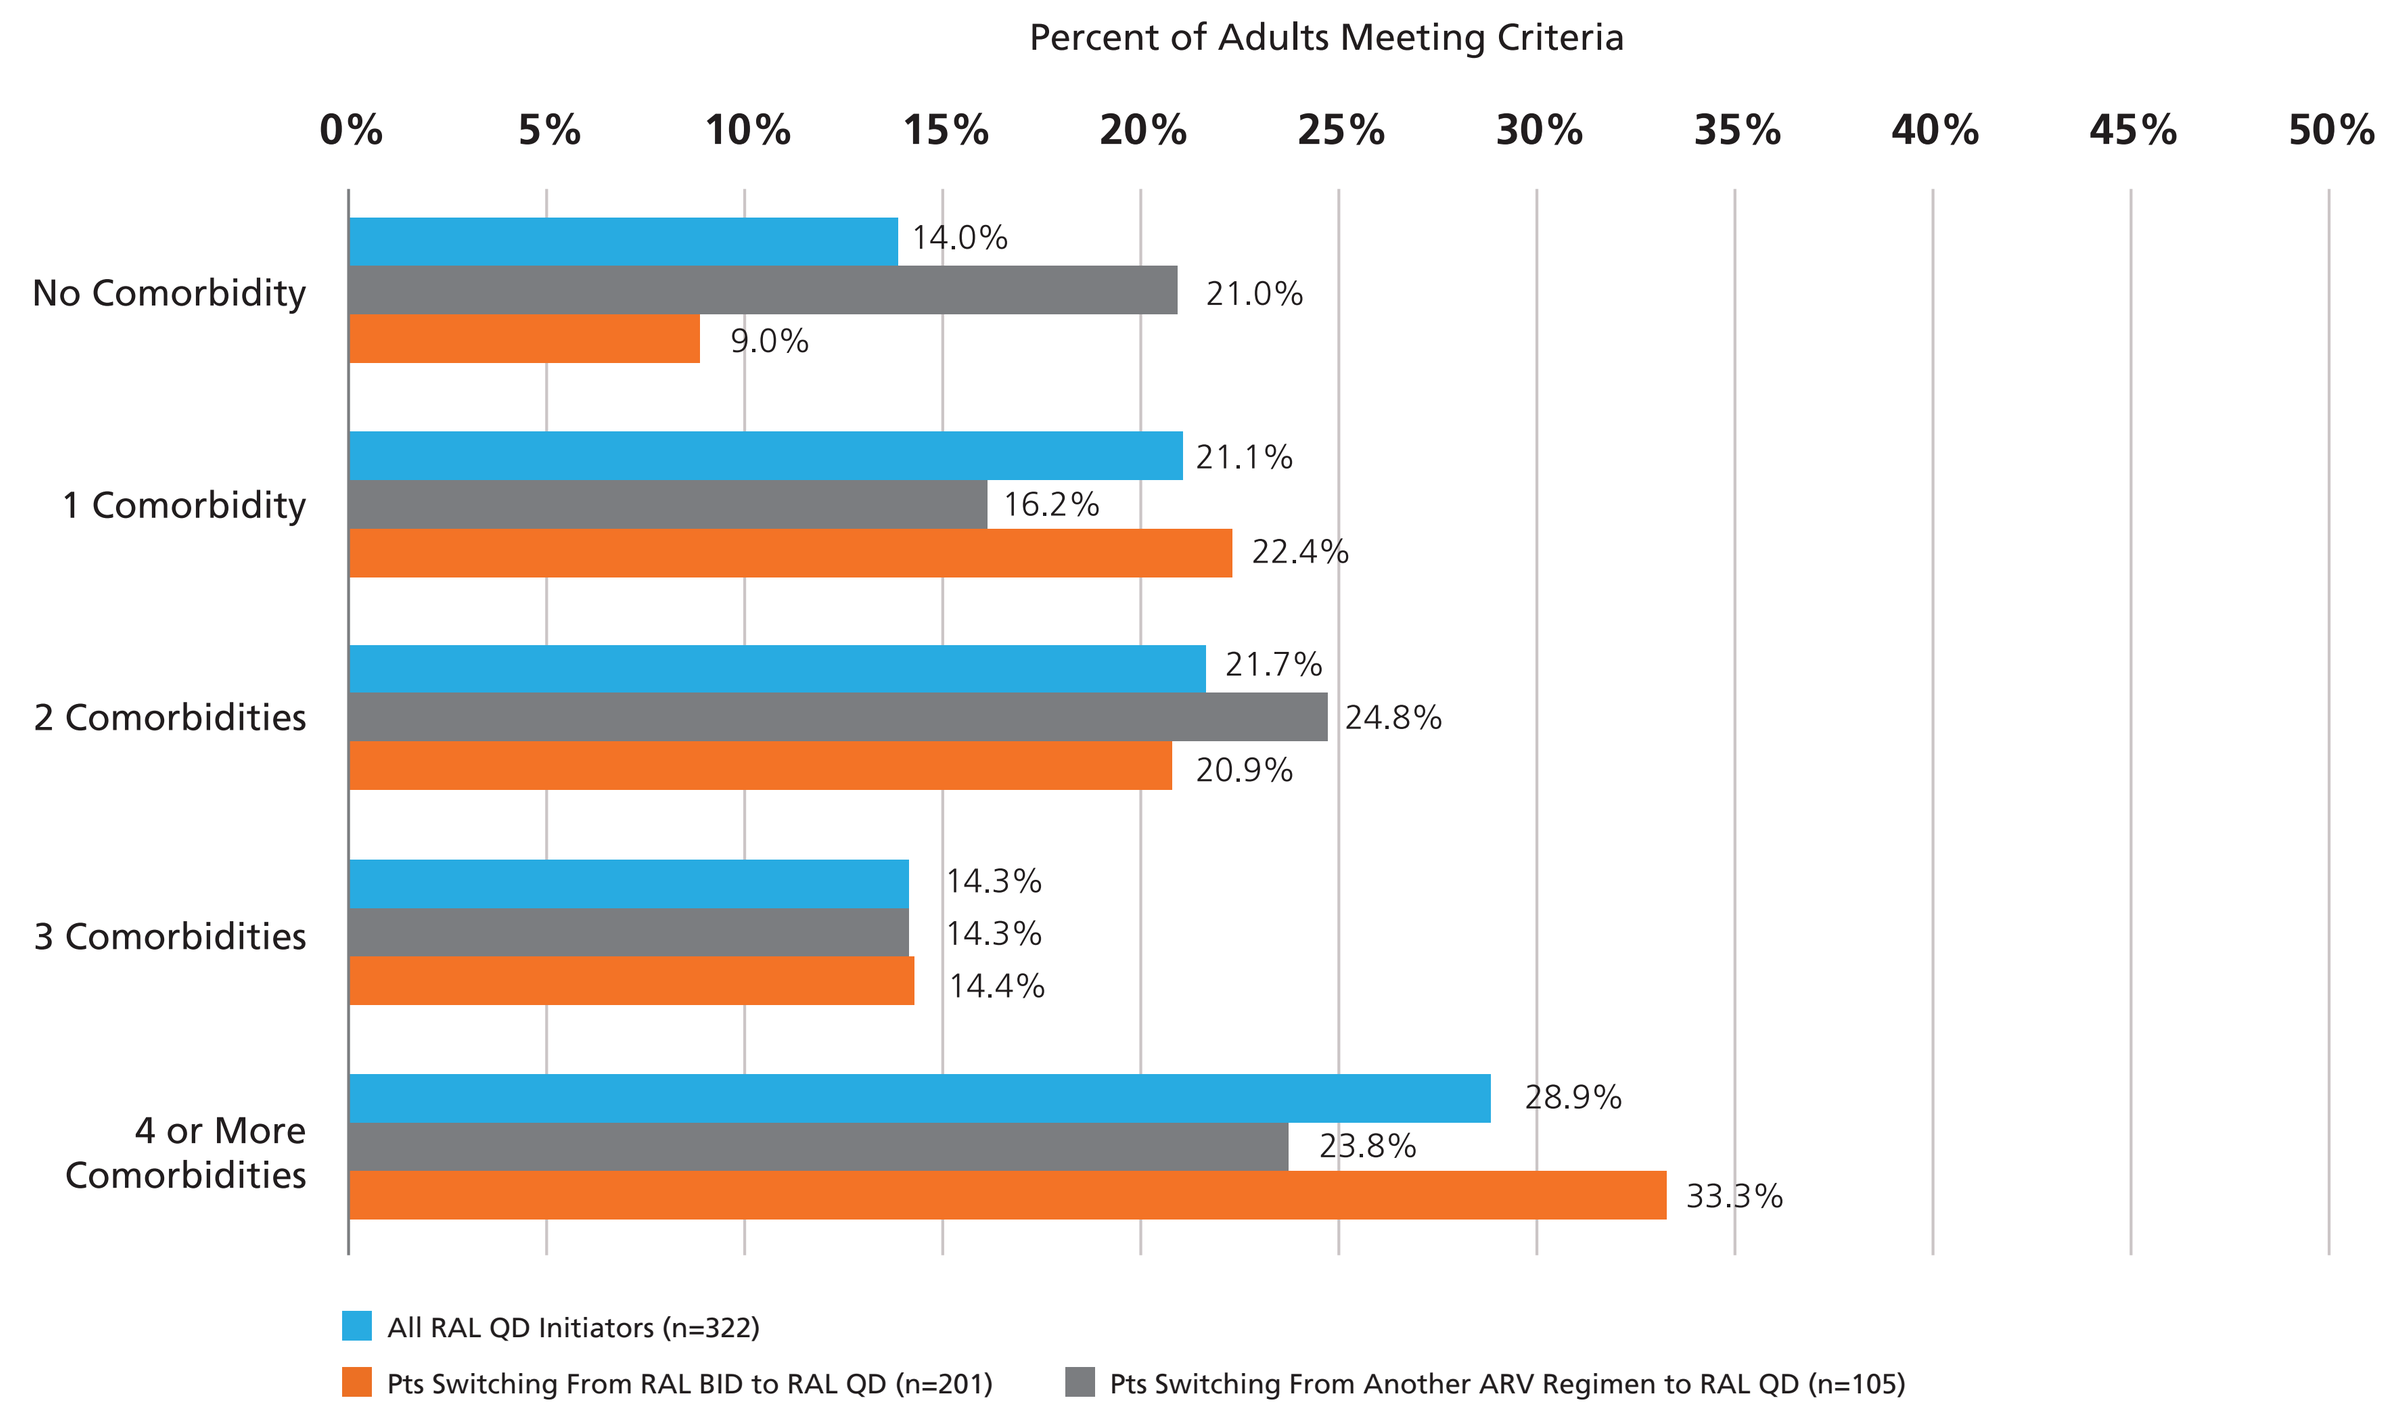

Percent of Adults Meeting Criteria
0% 5% 10% 15% 20% 25% 30% 35% 40% 45% 50%
No Comorbidity 14.0% 21.0% 9.0%
1 Comorbidity 21.1% 16.2% 22.4%
2 Comorbidities 21.7% 24.8% 20.9%
3 Comorbidities 14.3% 14.3% 14.4%
4 or More Comorbidities 28.9% 23.8% 33.3%
All RAL QD Initiators (n=322)
Pts Switching From RAL BID to RAL QD (n=201)
Pts Switching From Another ARV Regimen to RAL QD (n=105)

*ART Naive Patients (n=16) not shown separately §Classification based solely on the regimen immediately prior to initiation of RAL QD. Patients classified as other ARV (non-RAL) may have been prescribed RAL BID at a previous point in time.

- The median number of prescriptions for concomitant medications prescribed with RAL QD regimens was 5.5 (IQR: 4-8). The most frequent concurrently prescribed drug classes were lipid lowering agents (28.3%), anti-depressant agents (23.0%), anti-diabetic agents (14.3%), antibiotics (10.6%) and immunomodulators (11.2%). Figure 5 depicts the distribution of co-medications by drug class.

Figure 5: Concurrent Prescription In RAL QD Initiators

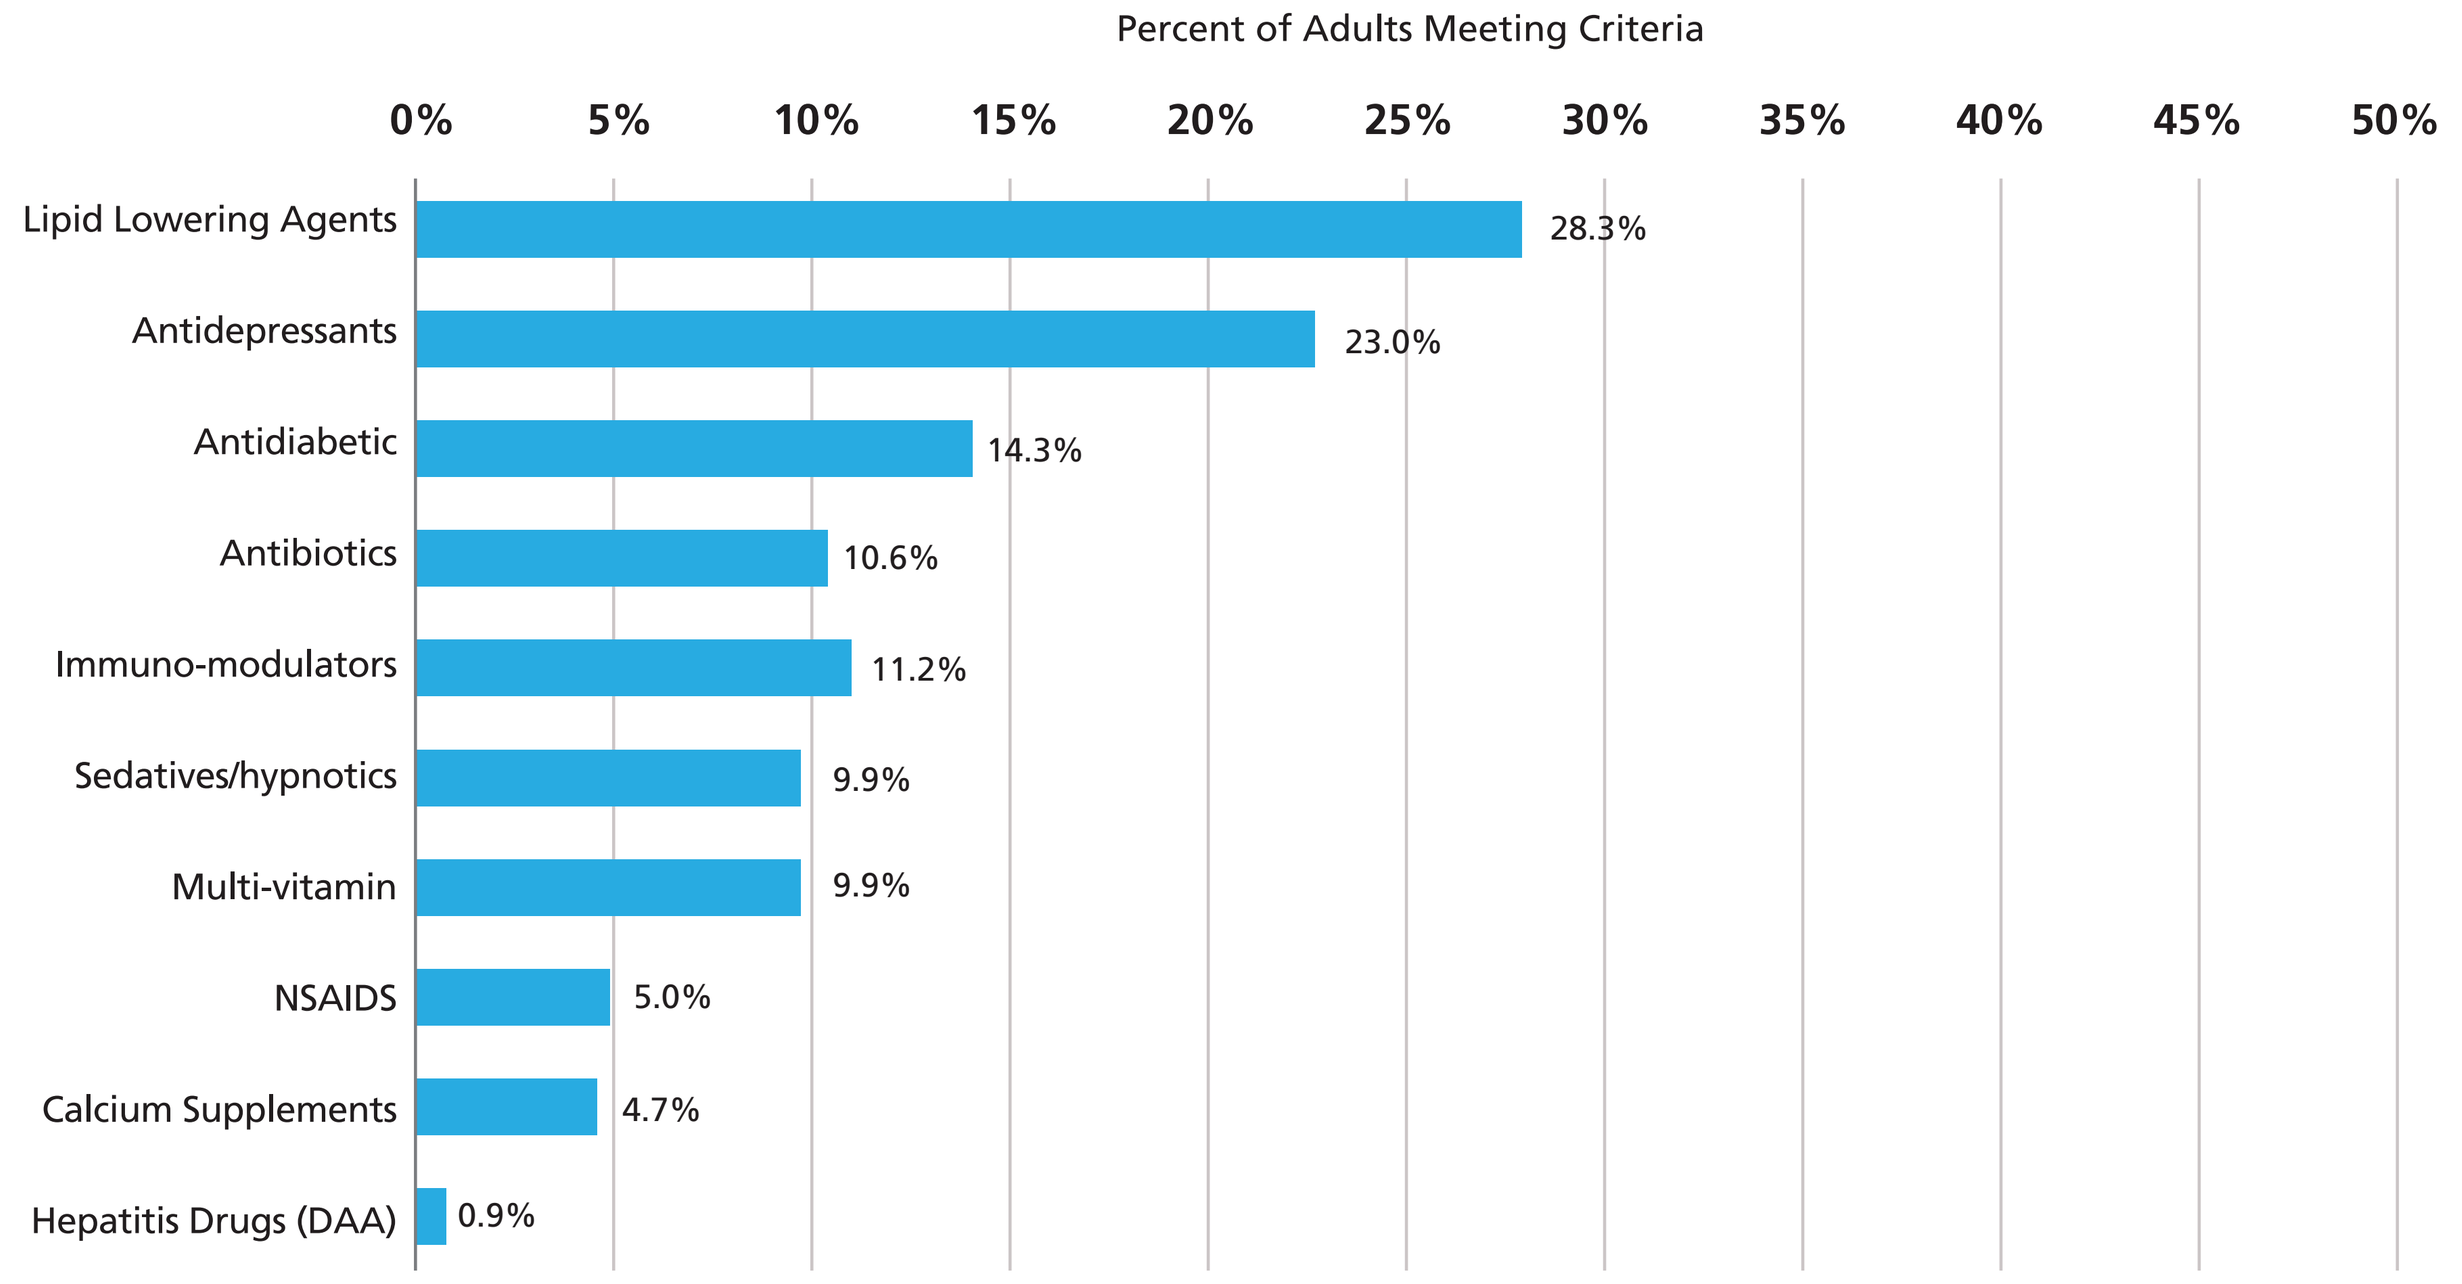

Percent of Adults Meeting Criteria
0% 5% 10% 15% 20% 25% 30% 35% 40% 45% 50%
Lipid Lowering Agents 28.3%
Antidepressants 23.0%
Antidiabetic 14.3%
Antibiotics 10.6%
Immuno-modulators 11.2%
Sedatives/hypnotics 9.9%
Multi-vitamin 9.9%
NSAIDS 5.0%
Calcium Supplements 4.7%
Hepatitis Drugs (DAA) 0.9%

LIMITATIONS

- Sample size in the ART naive subset was too small to report
- Issues confronting population-level assessments include such aspects as differential medical care by practice size and specialty, academic and research orientation of the health care practitioner, ethnic-based and gender-based attitudes and geographic regional health care practices. Accordingly, the results observed in this study may not generalize to the larger U.S. population.

CONCLUSIONS

- The majority of HIV-1 infected patients who received RAL QD are
 - treatment-experienced individuals previously suppressed (with viral load of <200 copies/mL) either on RAL BID or another ARV regimen
 - older than 50 years of age
 - diagnosed with multiple comorbidities
 - prescribed numerous non-ART medications to treat these comorbidities

KEY FINDINGS

In patients with HIV-1 infection and other concomitant chronic illnesses that require multiple medications, RAL QD provides an important option for simplifying their HIV regimen

REFERENCES

1. Rockstroh JK et al. J Acquir Immune Defic Syndr. 2013 May 1;63(1):77-85.
2. Eron JJ et al. Lancet Infect Dis. 2013 Jul;13(7):587-96.
3. US DHSS Guidelines for the use of antiretroviral agents in HIV-1-infected adults and adolescents. AIDSinfo. 2018
4. Cahn et al. J Acquir Immune Defic Syndr. 2018 Aug 15;78(5):589-598.

ACKNOWLEDGMENT

This research would not be possible without the generosity of the OPERA HIV caregivers and their patients. Additionally, we are grateful for the following individuals: Laurie Costa and Lito Torres (SAS programming), Jeff Briney (QA), Ted Ising (Database Arch & Mgmt), Bernie Stooks (Database Mgmt), Judy Johnson (Med Terminology Classification), Rodney Mood (Site Support & Data Analyst), Laurence Brunet and Terra Fatukasi (Figure Preparation & Proofreading)

SUPPORT

Funding for this research was provided by Merck Sharp & Dohme Corp., a subsidiary of Merck & Co., Inc., Kenilworth, NJ, USA

MERCK INVENTING FOR LIFE

Epividian®

https://bit.ly/2N878Yx