Clinical Effectiveness of Guideline-Recommended Antiretroviral Therapy Core Agents in HIV/HCV Co-infected Patients in the OPERA Observational Database

Douglas Dieterich[1], Laurence Brunet[2], Jennifer Fusco[2], Ricky Hsu[3,4], Vani Vannappagari[5], Lloyd Curtis[5], Maria Claudia Nascimento[5], and Gregory Fusco[2]

[1]Mt. Sinai Healthcare System, New York, NY; [2]Epividian, Durham, NC; [3]AIDS Healthcare Foundation, New York, NY; [4]NYU Langone Medical Center, New York, NY; [5]ViiV Healthcare, Research Triangle Park, NC

OPERA® The Longitudinal Cohort

Contact Information:
Laurence Brunet
4505 Emperor Blvd, Suite 220, Durham, NC
P: 919-827-0010 Email: laurence.brunet@epividian.com

BACKGROUND

- HIV infection affects the natural history of HCV, resulting in faster progression of HCV in HIV/HCV co-infected patients
- Effective antiretroviral therapy (ART) can slow HCV progression to a rate comparable to HCV mono-infection
- INSTI trials included very few HIV/HCV co-infected patients, who had a higher incidence of liver biochemistry increases compared to HIV mono-infected patients, although these increases were similar across core agent comparators

OBJECTIVE:

To compare the effectiveness of the commonly used core agents dolutegravir (DTG), elvitegravir (EVG), raltegravir (RAL) and darunavir (DRV) in patients with HIV/HCV co-infection in the U.S.

METHODS

Study Population

- Observational Pharmaco-Epidemiology Research & Analysis (OPERA®) observational database: prospective electronic health record data from 79 HIV out-patient clinics in 15 U.S. states following 79,883 people living with HIV
- Inclusion criteria: HIV/HCV co-infected patients ≥13 years of age initiating DTG, EVG, RAL or DRV with an HIV viral load >200 copies/ml between August 12, 2013 and June 30, 2016
- Baseline: date of DTG, EVG, RAL or DRV initiation
- Censoring events: 1) discontinuation of the core agent (gap ≥45 days), 2) cessation of continuous clinical activity (≥1 clinic visit or telephone contact), 3) death, or 4) study end (June 30, 2017)

Exposure

- Core agents of interest (DTG, EVG, RAL or DRV), excluding regimens containing >1 core agent of interest

Outcomes

- Grade 3-4 liver enzyme elevation (LEE): alanine aminotransferase (ALT), aspartate aminotransferase (AST) or alkaline phosphatase (ALK) >5.0 X upper limit of normal (ULN), or bilirubin >2.5 X ULN
- Viral suppression: viral load <50 copies/mL within 12 months of core agent initiation

Stratification

- ART-naïve: no history of ART prior to baseline and baseline viral load ≥1,000 copies/mL
- ART-experienced: record of any ART treatment prior to baseline or baseline viral load ≥200 and <1,000 copies/mL

Statistical Analyses

- Incidence of grade 3-4 liver enzyme elevation (LEE): among patients with normal baseline liver enzyme levels (AST, ALT, ALK and bilirubin ≤1 X ULN) and who remained HCV treatment naïve throughout follow-up
- 12-month suppression probability: Kaplan-Meier
- Time to viral suppression: multivariate Cox Proportional Hazards models adjusted for age, sex, race, HIV RNA, CD4 cell count, history of AIDS and VACS score at baseline

RESULTS

Patient Characteristics (Table 1)

Table 1. Baseline demographic and clinical characteristics of ART-naïve patients

	ART-naïve N=527				ART-experienced N=592			
	DTG, n(%)	EVG, n(%)	RAL, n(%)	DRV, n(%)	DTG, n(%)	EVG, n(%)	RAL, n(%)	DRV, n(%)
N (%)	**140 (27)**	**164 (31)**	**65 (12)**	**158 (30)**	**138 (23)**	**101 (17)**	**135 (23)**	**218 (37)**
Age ≥50 years	46 (33)	56 (34)	27 (42)	54 (34)	59 (43)	44 (44)	52 (38)	67 (31)
Female sex	33 (24)	41 (25)	16 (25)	32 (20)	19 (14)	23 (23)	34 (25)	49 (22)
African American	43 (31)	61 (37)	27 (42)	67 (42)	52 (38)	39 (39)	49 (36)	84 (38)
CD4 cell count ≤200 cells/µl	38 (27)	33 (20)	25 (39)	70 (44)	30 (22)	24 (24)	45 (33)	81 (37)
HIV RNA (copies/ml)								
≥200 to <1,000	NA	NA	NA	NA	50 (36)	30 (30)	37 (27)	62 (28)
≥1,000 to <10,000	22 (16)	29 (18)	14 (22)	25 (16)	23 (17)	25 (25)	36 (27)	40 (18)
≥10,000 to <100,000	78 (56)	90 (55)	36 (55)	73 (46)	44 (32)	33 (33)	38 (28)	73 (34)
≥100,000	40 (29)	45 (27)	15 (23)	60 (38)	21 (15)	13 (13)	24 (18)	43 (20)
History of AIDS-defining illness	6 (4)	12 (7)	8 (12)	23 (15)	26 (19)	16 (16)	53 (39)	74 (34)
VACS* ≥45	45 (32)	48 (29)	29 (45)	59 (37)	45 (33)	30 (30)	54 (40)	84 (38)
HCV treatment-naïve	137 (98)	162 (99)	63 (97)	157 (99)	125 (91)	99 (98)	115 (85)	196 (90)

* VACS Mortality Index: Scored by summing pre-assigned points for age, CD4 count, HIV-1 RNA, hemoglobin, platelets, aspartate and alanine transaminase, creatinine, and viral hepatitis C infection. A higher score is associated with a higher risk of 5-year all-cause mortality

Liver Enzyme Elevation During Follow-Up

- Grade 3-4 LEE was rare among ART-naïve (Figure 1A) and ART-experienced (Figure 1B) patients who remained HCV-treatment naïve throughout follow-up and had normal baseline liver enzyme levels, with no statistically significant difference in incidence across core agent used

Figure 1. Incidence of grade 3-4 liver enzyme elevation in (A) ART-naïve patients and (B) ART-experienced patients*

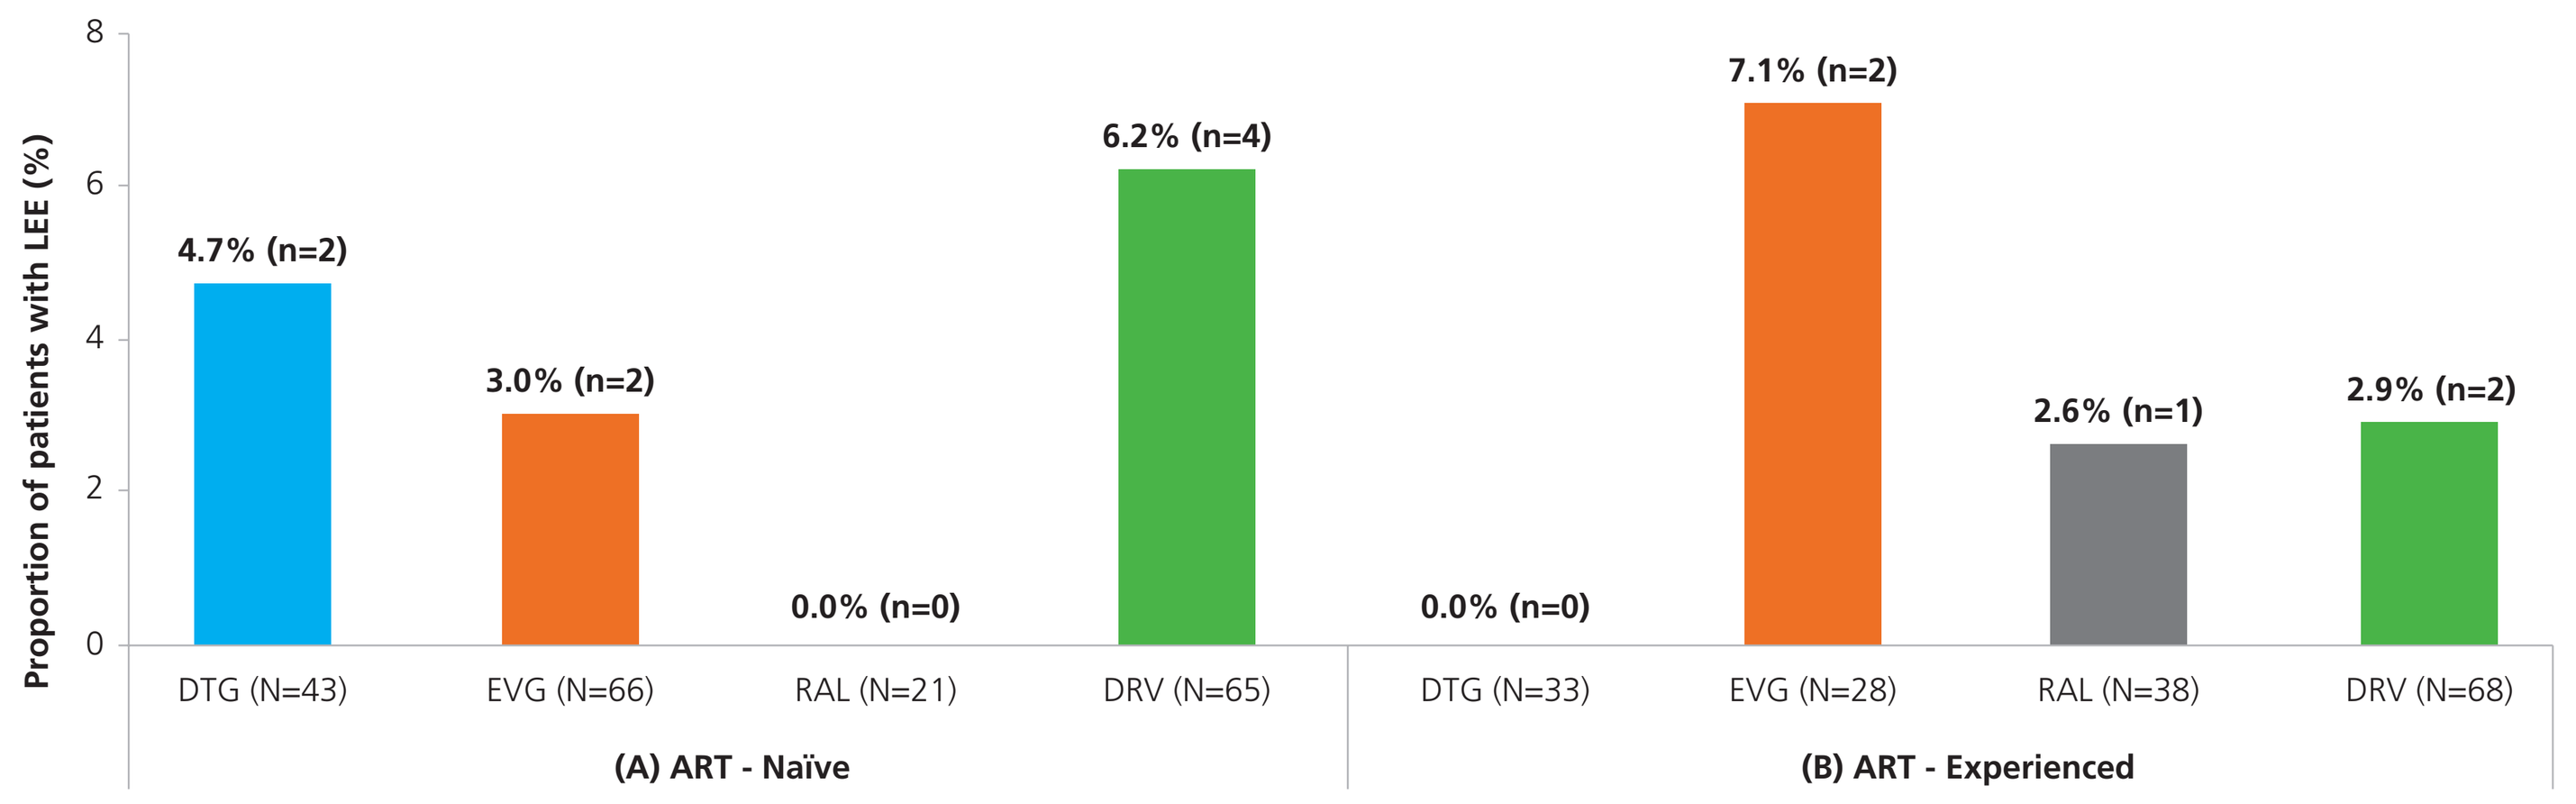
Proportion of patients with LEE (%)
8
6
4
2
0
4.7% (n=2)
3.0% (n=2)
0.0% (n=0)
6.2% (n=4)
0.0% (n=0)
7.1% (n=2)
2.6% (n=1)
2.9% (n=2)
DTG (N=43)
EVG (N=66)
RAL (N=21)
DRV (N=65)
DTG (N=33)
EVG (N=28)
RAL (N=38)
DRV (N=68)
(A) ART - Naïve
(B) ART - Experienced

*Population restricted to patients with normal baseline liver enzyme levels (AST, ALT, ALK and bilirubin ≤1 X ULN) and who remained HCV treatment naïve throughout follow-up

Cumulative Probability of Viral Suppression

- DRV users had the lowest cumulative probability of viral suppression by 12 months of ART, in both ART-naïve and ART-experienced patients (Figure 2)
- Among ART-naïve patients, viral suppression probability was not statistically significantly different between DTG, EVG and RAL (Figure 2A)
- Among ART-experienced patients, viral suppression probability was not statistically significantly different between any of the core agents (Figure 2B)

Figure 2. Cumulative probability of HIV viral suppression in (A) ART-naïve and (B) ART-experienced patients

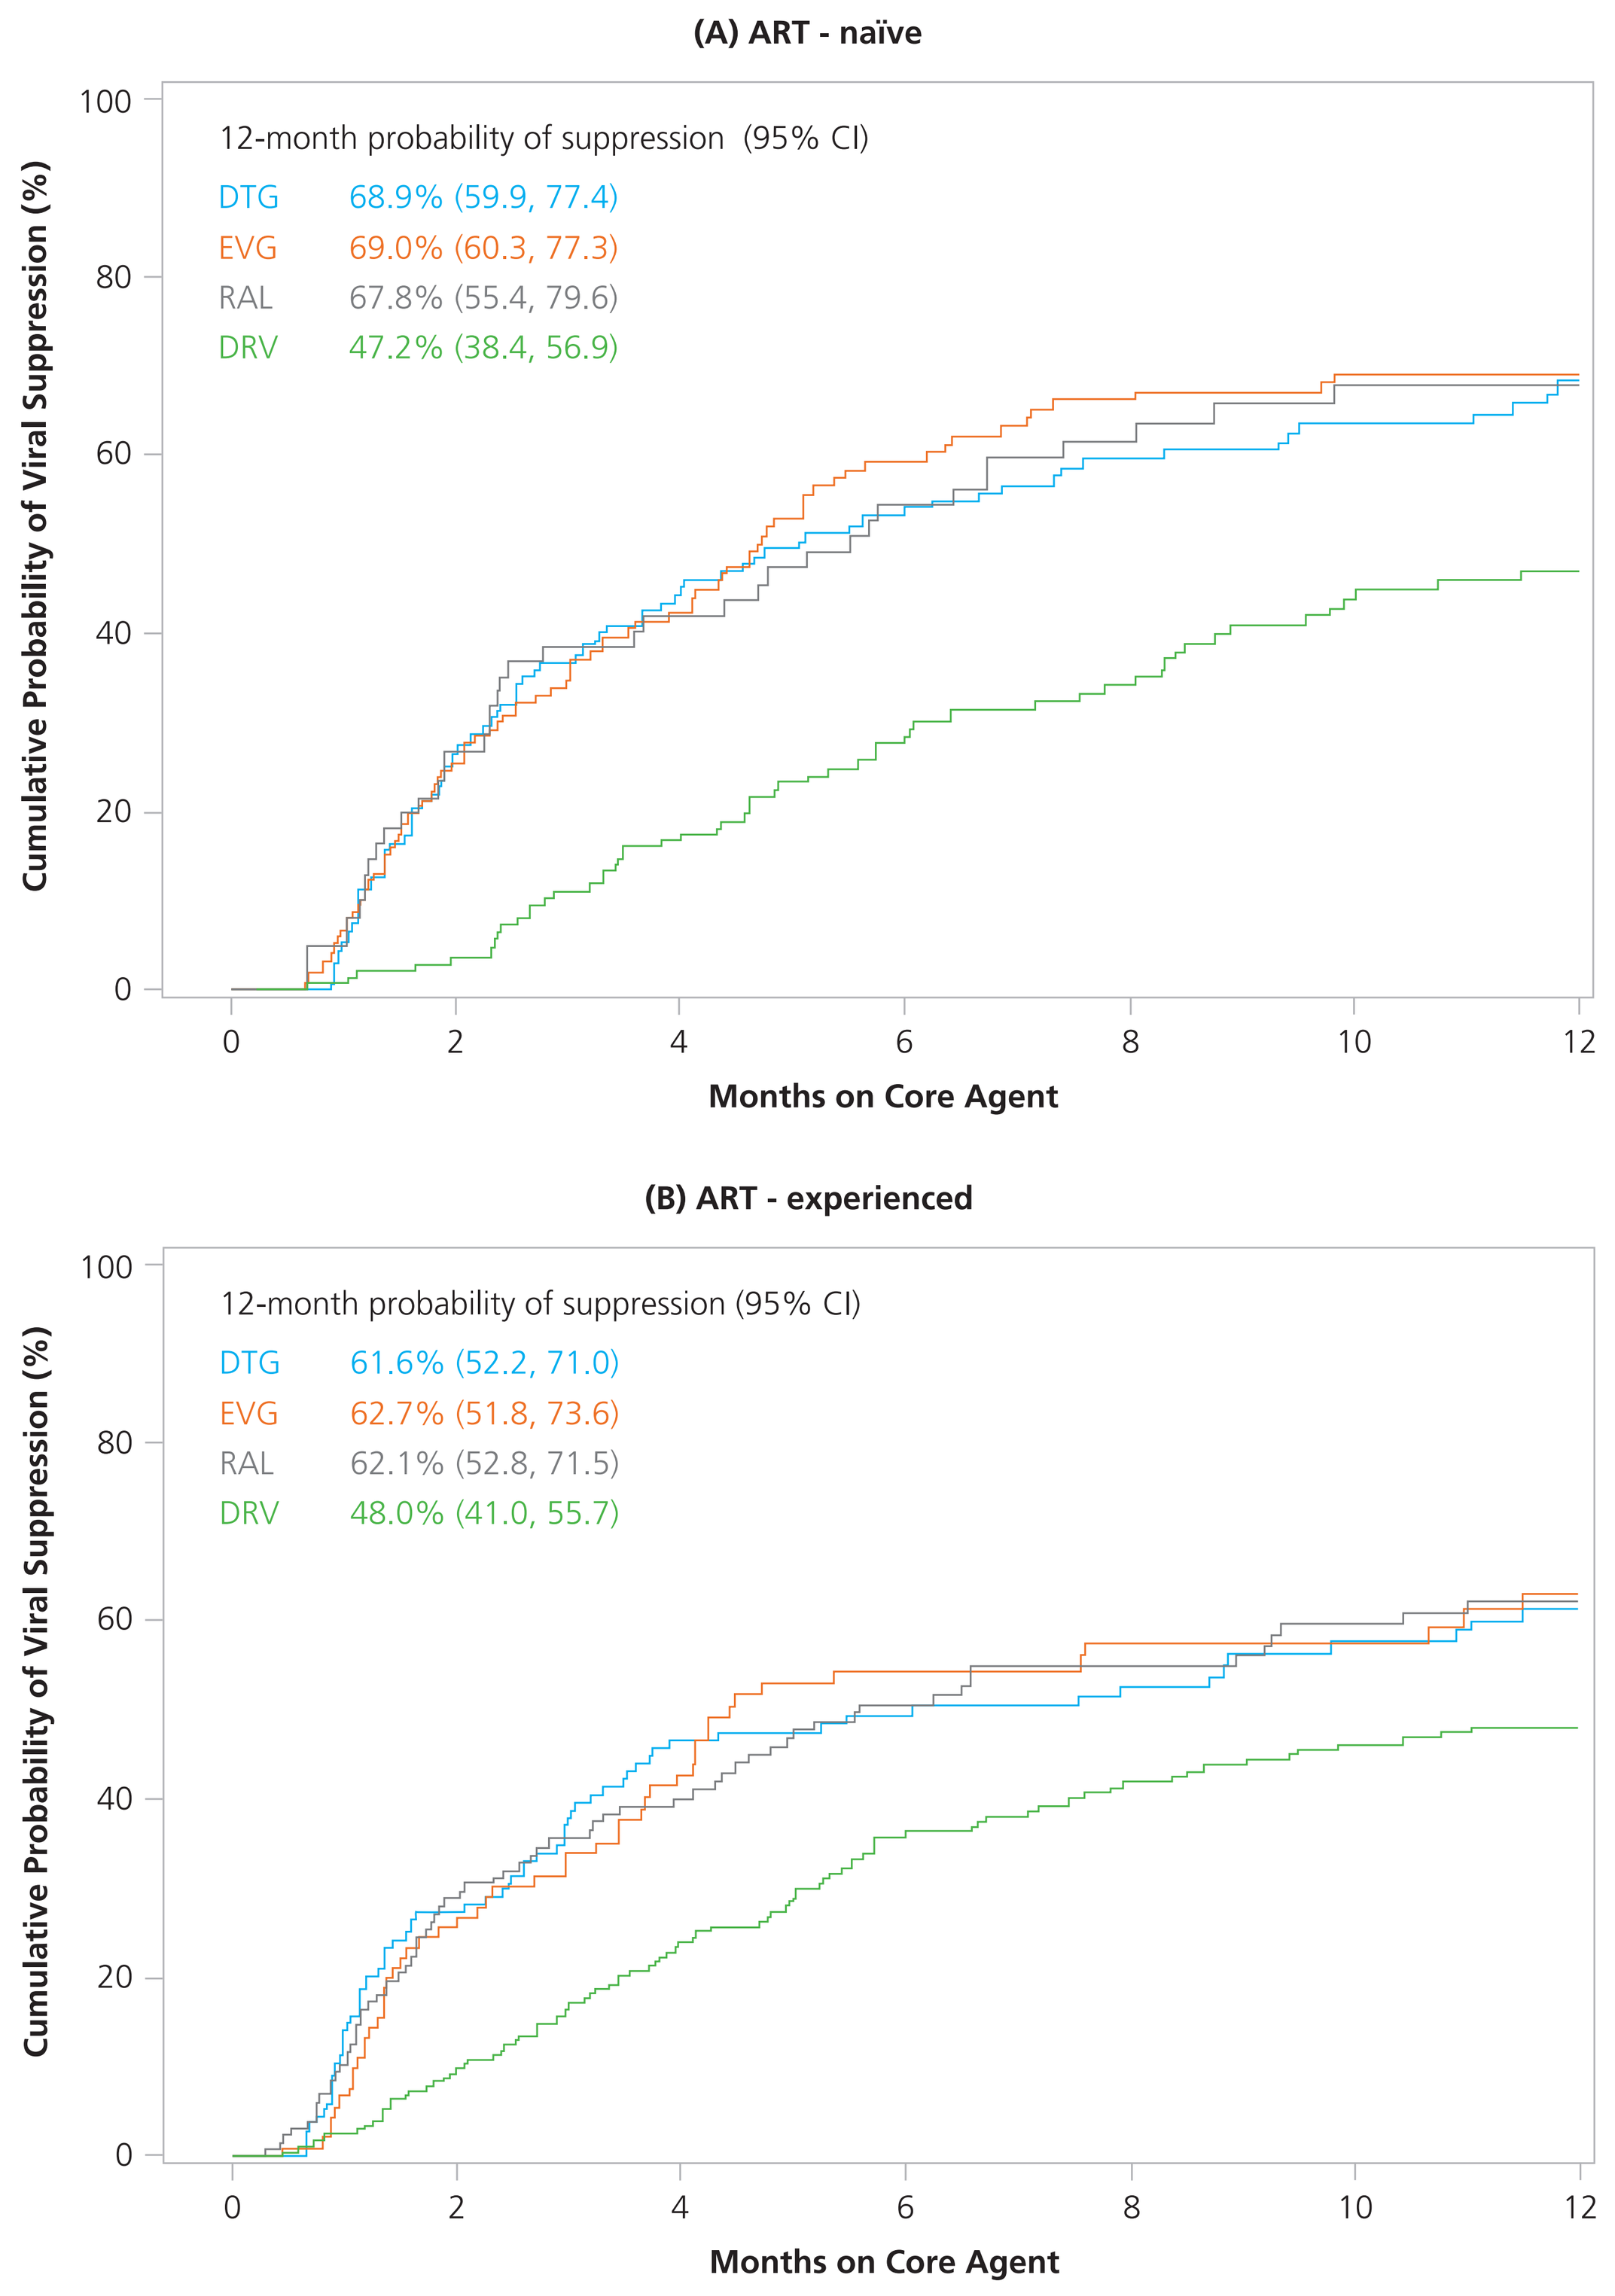
(A) ART - naïve
12-month probability of suppression (95% CI)
DTG 68.9% (59.9, 77.4)
EVG 69.0% (60.3, 77.3)
RAL 67.8% (55.4, 79.6)
DRV 47.2% (38.4, 56.9)
Cumulative Probability of Viral Suppression (%)
100
80
60
40
20
0
0
2
4
6
8
10
12
Months on Core Agent
(B) ART - experienced
12-month probability of suppression (95% CI)
DTG 61.6% (52.2, 71.0)
EVG 62.7% (51.8, 73.6)
RAL 62.1% (52.8, 71.5)
DRV 48.0% (41.0, 55.7)
Cumulative Probability of Viral Suppression (%)
100
80
60
40
20
0
0
2
4
6
8
10
12
Months on Core Agent

Time to Viral Suppression

- Among ART-naïve patients, only DRV users had a slower time to viral suppression compared to DTG users; adjusted hazard ratio (aHR): 0.47 (95% CI: 0.33, 0.66) (Figure 3A)
- Among ART-experienced patients, only DRV users had a slower time to viral suppression compared to DTG users; aHR: 0.67 (95% CI: 0.48, 0.94) (Figure 3B)

Figure 3. Association between core agents and time to viral suppression in (A) ART-naïve and (B) ART-experienced patients

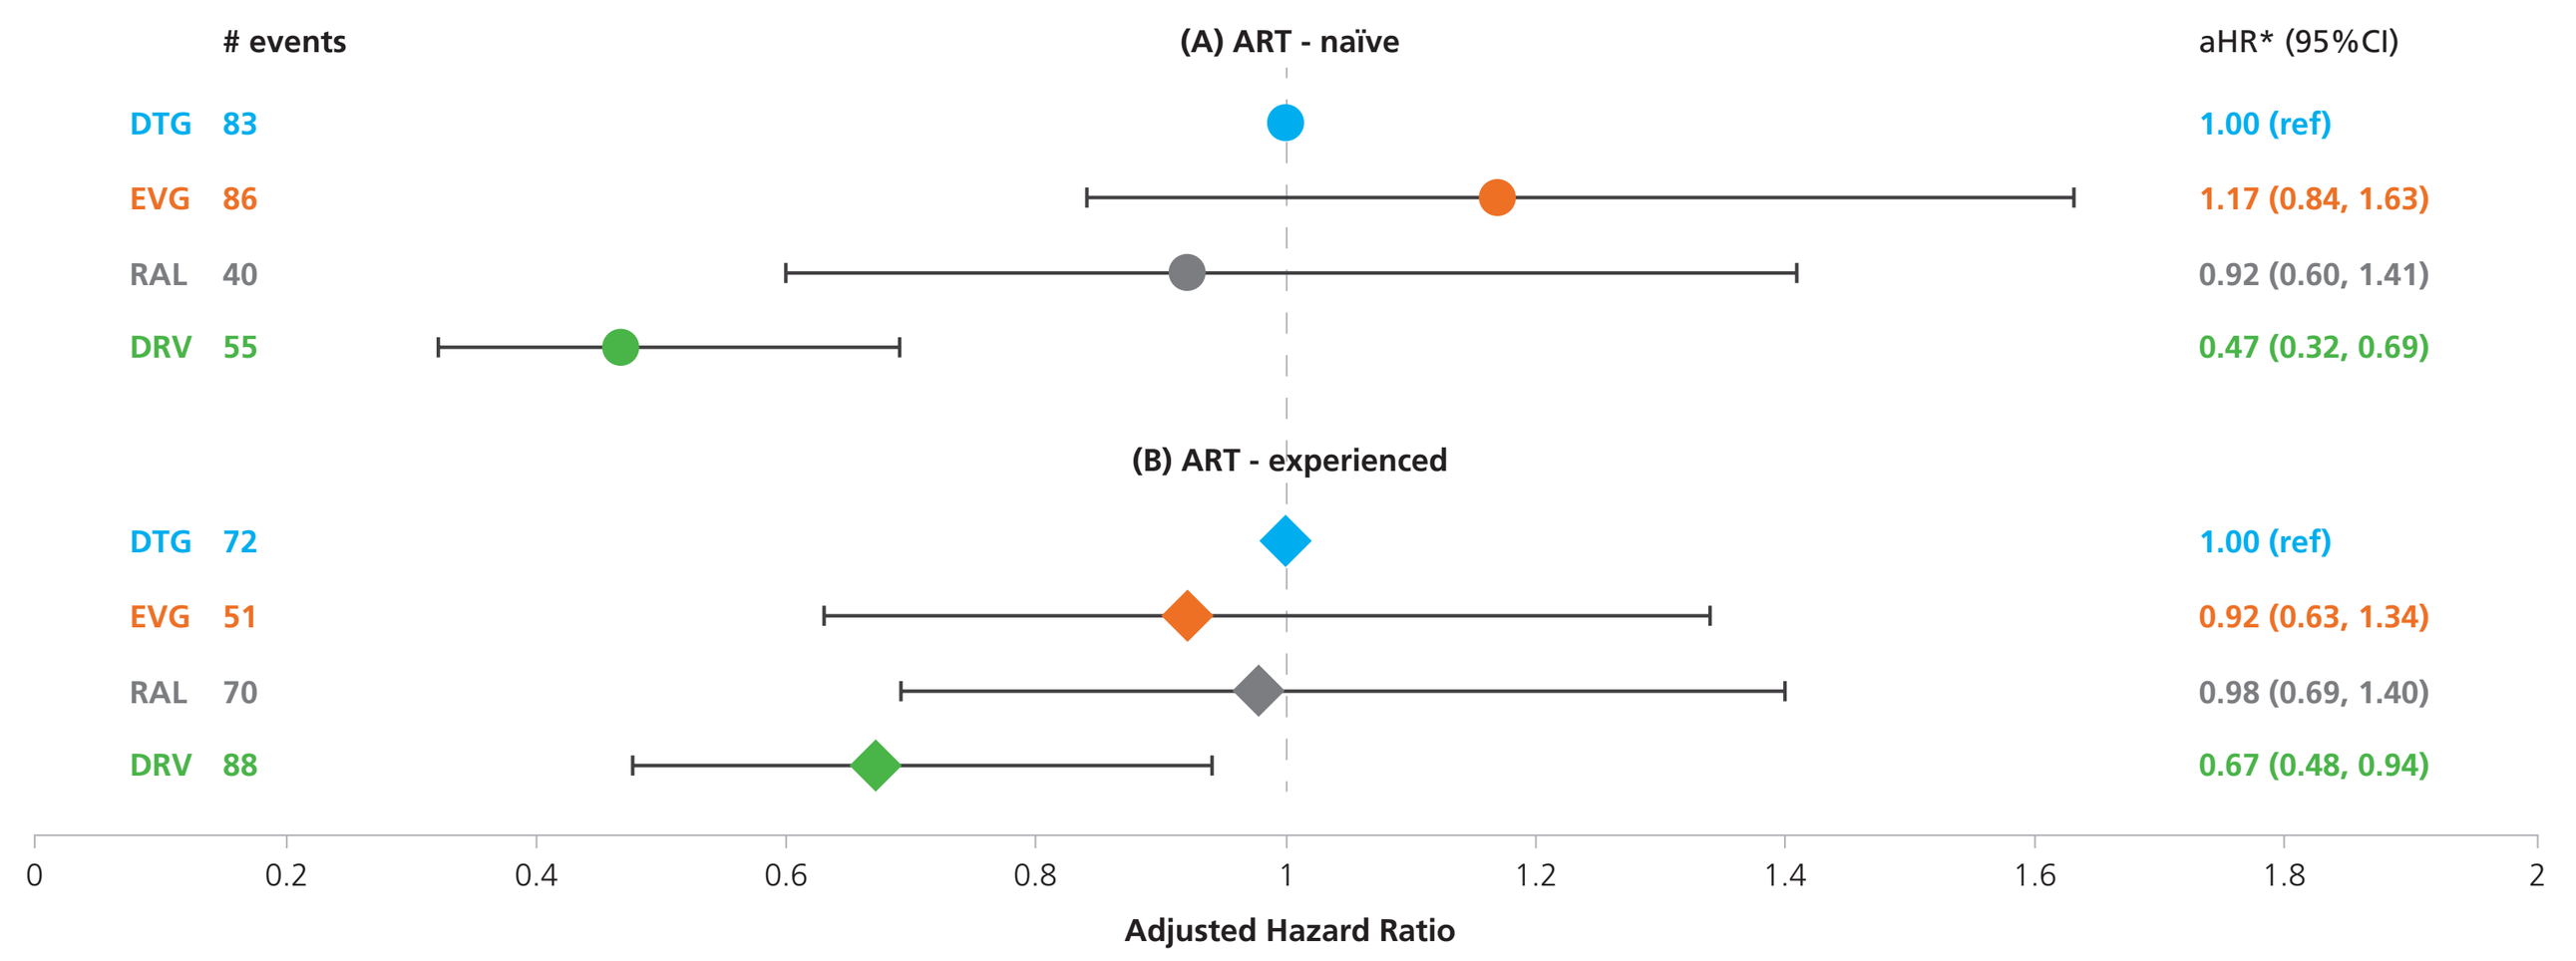
events
(A) ART - naïve
aHR* (95%CI)
DTG 83
1.00 (ref)
EVG 86
1.17 (0.84, 1.63)
RAL 40
0.92 (0.60, 1.41)
DRV 55
0.47 (0.32, 0.69)
(B) ART - experienced
DTG 72
1.00 (ref)
EVG 51
0.92 (0.63, 1.34)
RAL 70
0.98 (0.69, 1.40)
DRV 88
0.67 (0.48, 0.94)
0
0.2
0.4
0.6
0.8
1
1.2
1.4
1.6
1.8
2
Adjusted Hazard Ratio

* Adjusted for age ≥50, sex, race, HIV RNA (cubic splines with knots at 10,000 and 100,000 copies/mL for ART-naïve and knots at 1,000 and 10,000 copies/mL for ART-experienced); CD4 cell count ≤200 cells/µL, history of AIDS and VACS score (15-29, 30-44, ≥45 vs. <15) at baseline

DISCUSSION

- LEE were rare among ART-naïve and ART-experienced patients with normal baseline liver enzyme levels without HCV treatment and all core agents were comparable, although the small number of events was a limiting factor (Figure 1)
- Patterns of 12-month probability of viral suppression and time to suppression were comparable for ART-naïve and ART-experienced patients
- Compared to DTG, 12-month probability of viral suppression was significantly lower with DRV, but not with RAL or EVG in ART-naïve patients (Figure 2A)
- 12-month probability of viral suppression did not differ significantly between any of the core agents in ART-experienced patients (Figure 2B)
- Only DRV was associated with a slower time to suppression compared to DTG in ART-naïve and ART-experienced patients, after adjustment for confounding (Figure 3)
- Both DRV and DTG are recommended for patients with known or suspected poor adherence

KEY FINDINGS

Among HIV/HCV co-infected patients, all INSTIs (DTG, EVG, RAL) performed as well in terms of viral suppression, while DRV use resulted in poorer virologic outcomes. Comparable results were obtained among ART-naïve patients and ART-experienced patients who switched with a viral load ≥200 copies/ml.

ACKNOWLEDGMENTS

This research would not be possible without the generosity of the OPERA HIV caregivers and their patients. Additionally, we are grateful for the following individuals: Robin Beckerman (SAS programming), Jeff Briney (QA), Ted Ising (Database Arch & Mgmt), Bernie Stooks (Database Mgmt), Judy Johnson (Med Terminology Classification), Rodney Mood (Site Support & Data Analyst).

SPONSORSHIP

This research was funded by ViiV Healthcare.

ViiV Healthcare

Epividian®

Presented at the 22nd International AIDS Conference – Amsterdam, the Netherlands

#AIDS2018 | @AIDS_conference | www.aids2018.org

AIDS 2018