POSTER
#664

Psychiatric Disorders Observed in HIV+ Patients Using 6 Common 3rd Agents in OPERA

Ricky Hsu[1,2], Jennifer Fusco[3], Cassidy Henegar[3], Felix Carpio[4], Karam Mounzer[5], Michael Wohlfeiler[6], Vani Vannappagari[7], Michael Aboud[8], Lloyd Curtis[9], Gregory Fusco[3]

[1] New York University Langone Medical Center, New York, NY, USA; [2] AIDS Healthcare Foundation, New York, NY, [3] Epividian, Inc., Durham, NC, USA; [4] Altamed Health Services, Los Angeles, CA, USA; [5] Philadelphia FIGHT, Philadelphia, PA, USA; [6] AIDS Healthcare Foundation, Los Angeles, CA, USA; [7] ViiV Healthcare, Research Triangle Park, NC USA; [8] ViiV Healthcare, London, UK; [9] GlaxoSmithKline, London, UK

OPERA®
The Longitudinal Cohort

Contact Information:
4819 Emperor Blvd., Suite 400 Durham, NC 27703
P: 919-825-3457 F: 919-313-4505
Email: jennifer.fusco@epividian.com

BACKGROUND

- Symptoms of psychiatric disorders (PD) are substantially more frequent in people living with HIV compared to the general population.[1]
- Higher rates of PD in HIV patients are partially attributable to treatment with antiretroviral (ARV) drugs, which are associated with a variety of adverse effects on the central nervous system (CNS).[2]
- A recent study observed higher than expected rates of PD symptoms in a clinical setting of patients receiving dolutegravir (DTG) as compared to what was seen in clinical trials.[3,4]

OBJECTIVE:

To evaluate psychiatric disorders in patients receiving dolutegravir (DTG)-containing regimens and five other widely prescribed anchor agent ARVs in clinical practice.

METHODS

Study Population

The study population was selected from the OPERA cohort, which includes prospectively-captured, routine clinical data from patients at 79 outpatient clinics in 15 states in the United States.

Figure 1. Individual electronic medical record databases are anonymized and aggregated to create OPERA

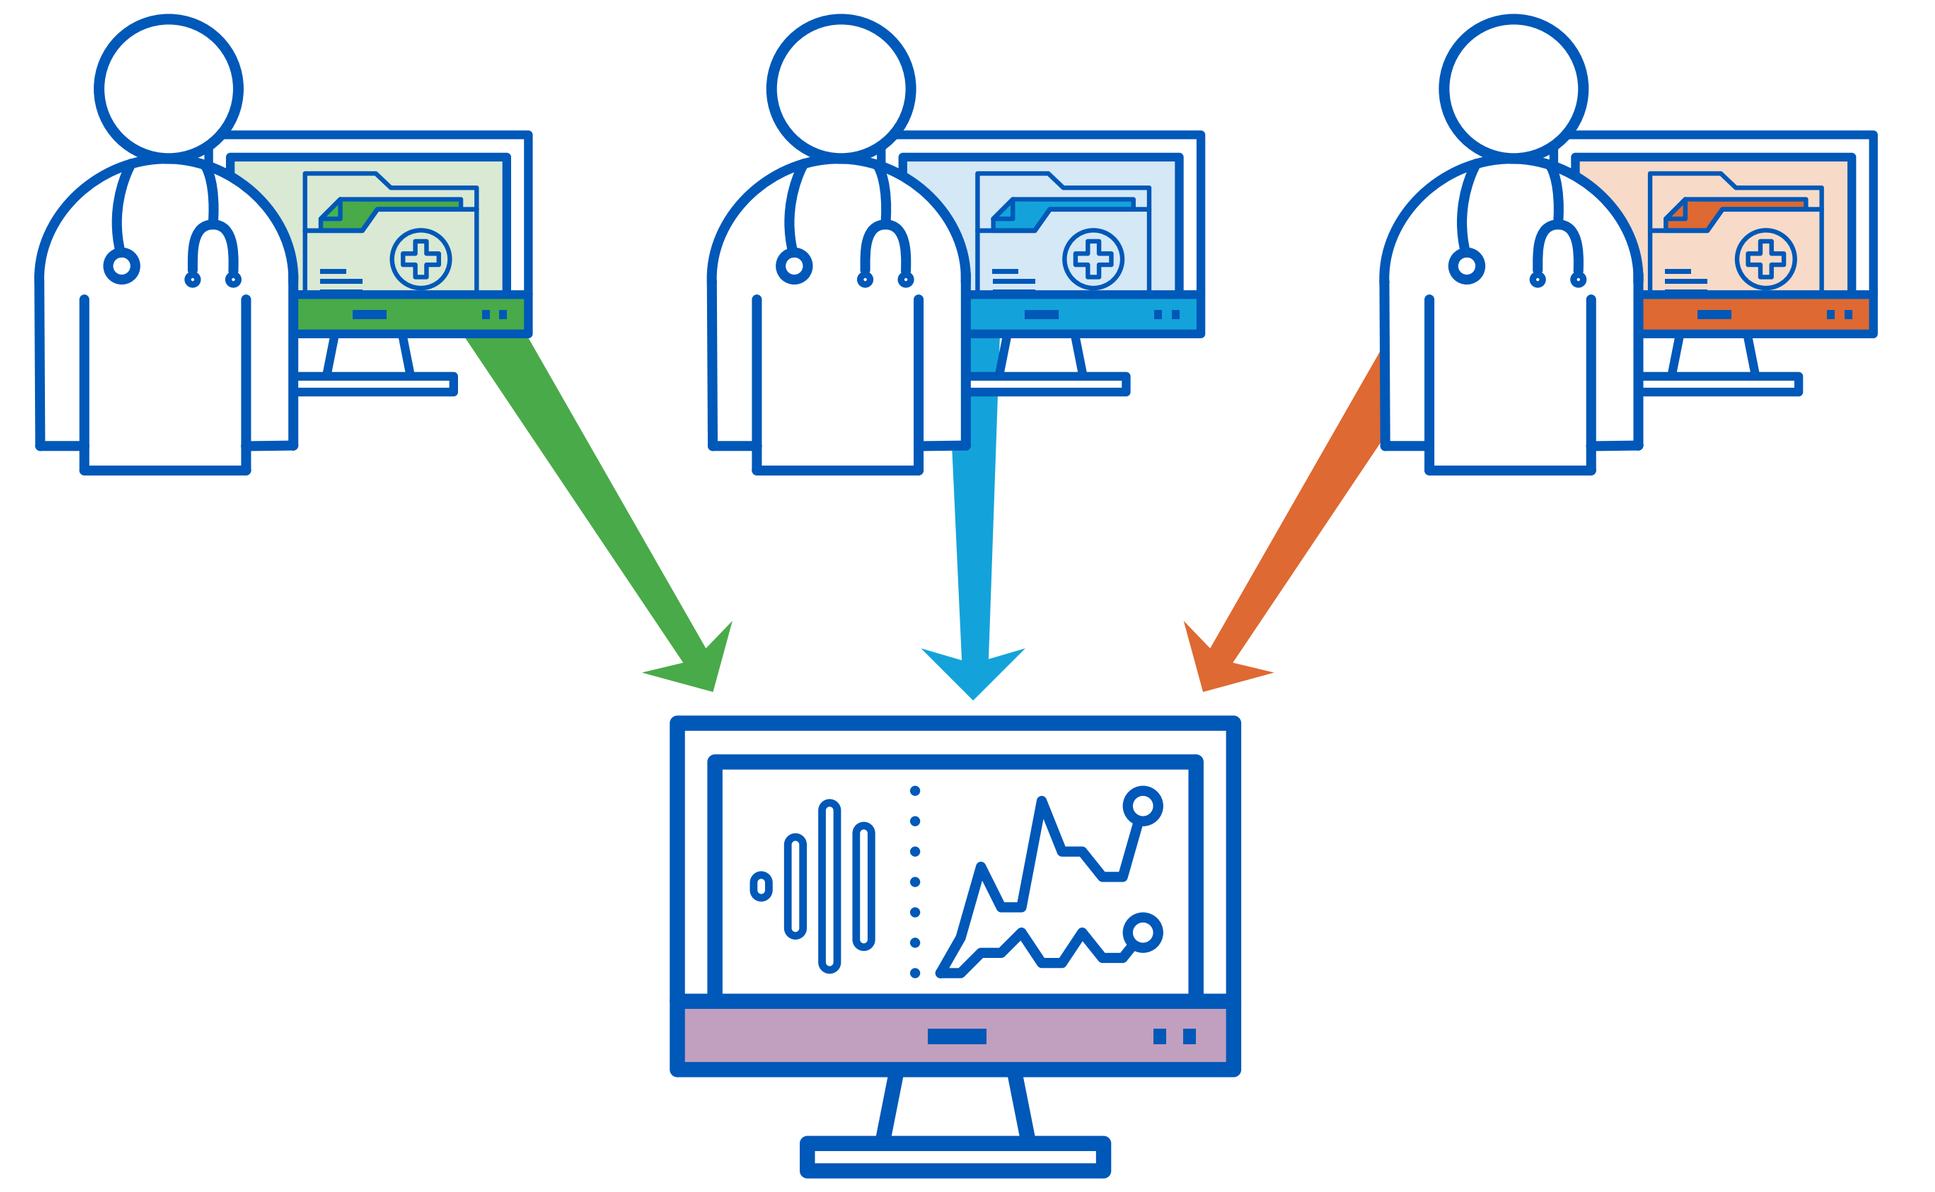

Study Design and Analysis

- Patients prescribed a regimen containing DTG, efavirenz (EFV), raltegravir (RAL), darunavir (DRV), rilpivirine (RPV), or elvitegravir (EVG) for the first time in the OPERA database between January 1, 2013 and August 15, 2015 were analyzed. Patients with exposure to any of the anchor drugs prior to the observation period or two or more anchor drugs at the same time were excluded.
- Patients were observed from the regimen start date until regimen discontinuation, loss to follow-up, death, or data freeze (August 15, 2016) .
- PD events included diagnoses of the following psychiatric conditions during the observation period: anxiety, depression, insomnia, and suicidality. Discontinuations within 14 days of a PD event were also analyzed.
 - Prevalent diagnoses: all diagnoses that occur for that condition regardless of whether the patient had the same diagnosis prior to baseline.
 - Incident diagnoses: only diagnoses for which the patient has no history prior to the observation period.
- Time to PD diagnosis and PD diagnosis with discontinuation within 14 days were evaluated using Kaplan-Meier curves.

RESULTS

Table 1. Baseline demographic and clinical characteristics by anchor agent

	DTG N=2180	EFV N=1622	RAL N=917	DRV N=1759	RPV N=1758	EVG N=3303
Age, yrs[1]	43 (31, 52)	**42 (30, 50)**	**48 (39, 55)**	**45 (34, 52)**	**33 (26, 45)**	**35 (27, 46)**
Male	1859 (85.3%)	**1419 (87.6%)**	**749 (81.8%)**	**1395 (79.4%)**	**1415 (80.6%)**	**2861 (86.7%)**
African American	805 (36.9%)	656 (40.4%)	**273 (29.8%)**	**737 (41.9%)**	**870 (49.5%)**	1301 (39.4%)
ART-naive	1247 (57.2%)	**1519 (93.6%)**	**601 (65.5%)**	**1119 (63.6%)**	**1466 (83.4%)**	**2672 (80.9%)**
Viral Load Log10 c/mL[1]	3.9 (1.3, 4.8)	4.0 (1.3, 4.9)	**1.8 (1.3, 4.4)**	**4.1 (1.6, 5.0)**	4.0 (2.5, 4.5)	**4.5 (3.4,5.0)**
CD4 cells/ µL[1]	452 (274, 650)	459 (294, 674)	481 (266, 680)	**364 (152, 587)**	**504 (360, 677)**	**414 (259, 590)**

[1] Median (Interquartile range)
[2] Bold text indicates a statistically significant difference (p<0.01) when compared to DTG

- Out of 70,106 HIV-positive patients in the OPERA database, 11,539 initiated their first regimen containing one of the ARVs of interest in the study period.
- At baseline patients receiving DTG were less likely to be treatment-naive compared to patients initiating each of the other anchor drugs (p<0.0001). (Table 1)

Figure 2. History of psychiatric diagnoses at baseline by anchor agent

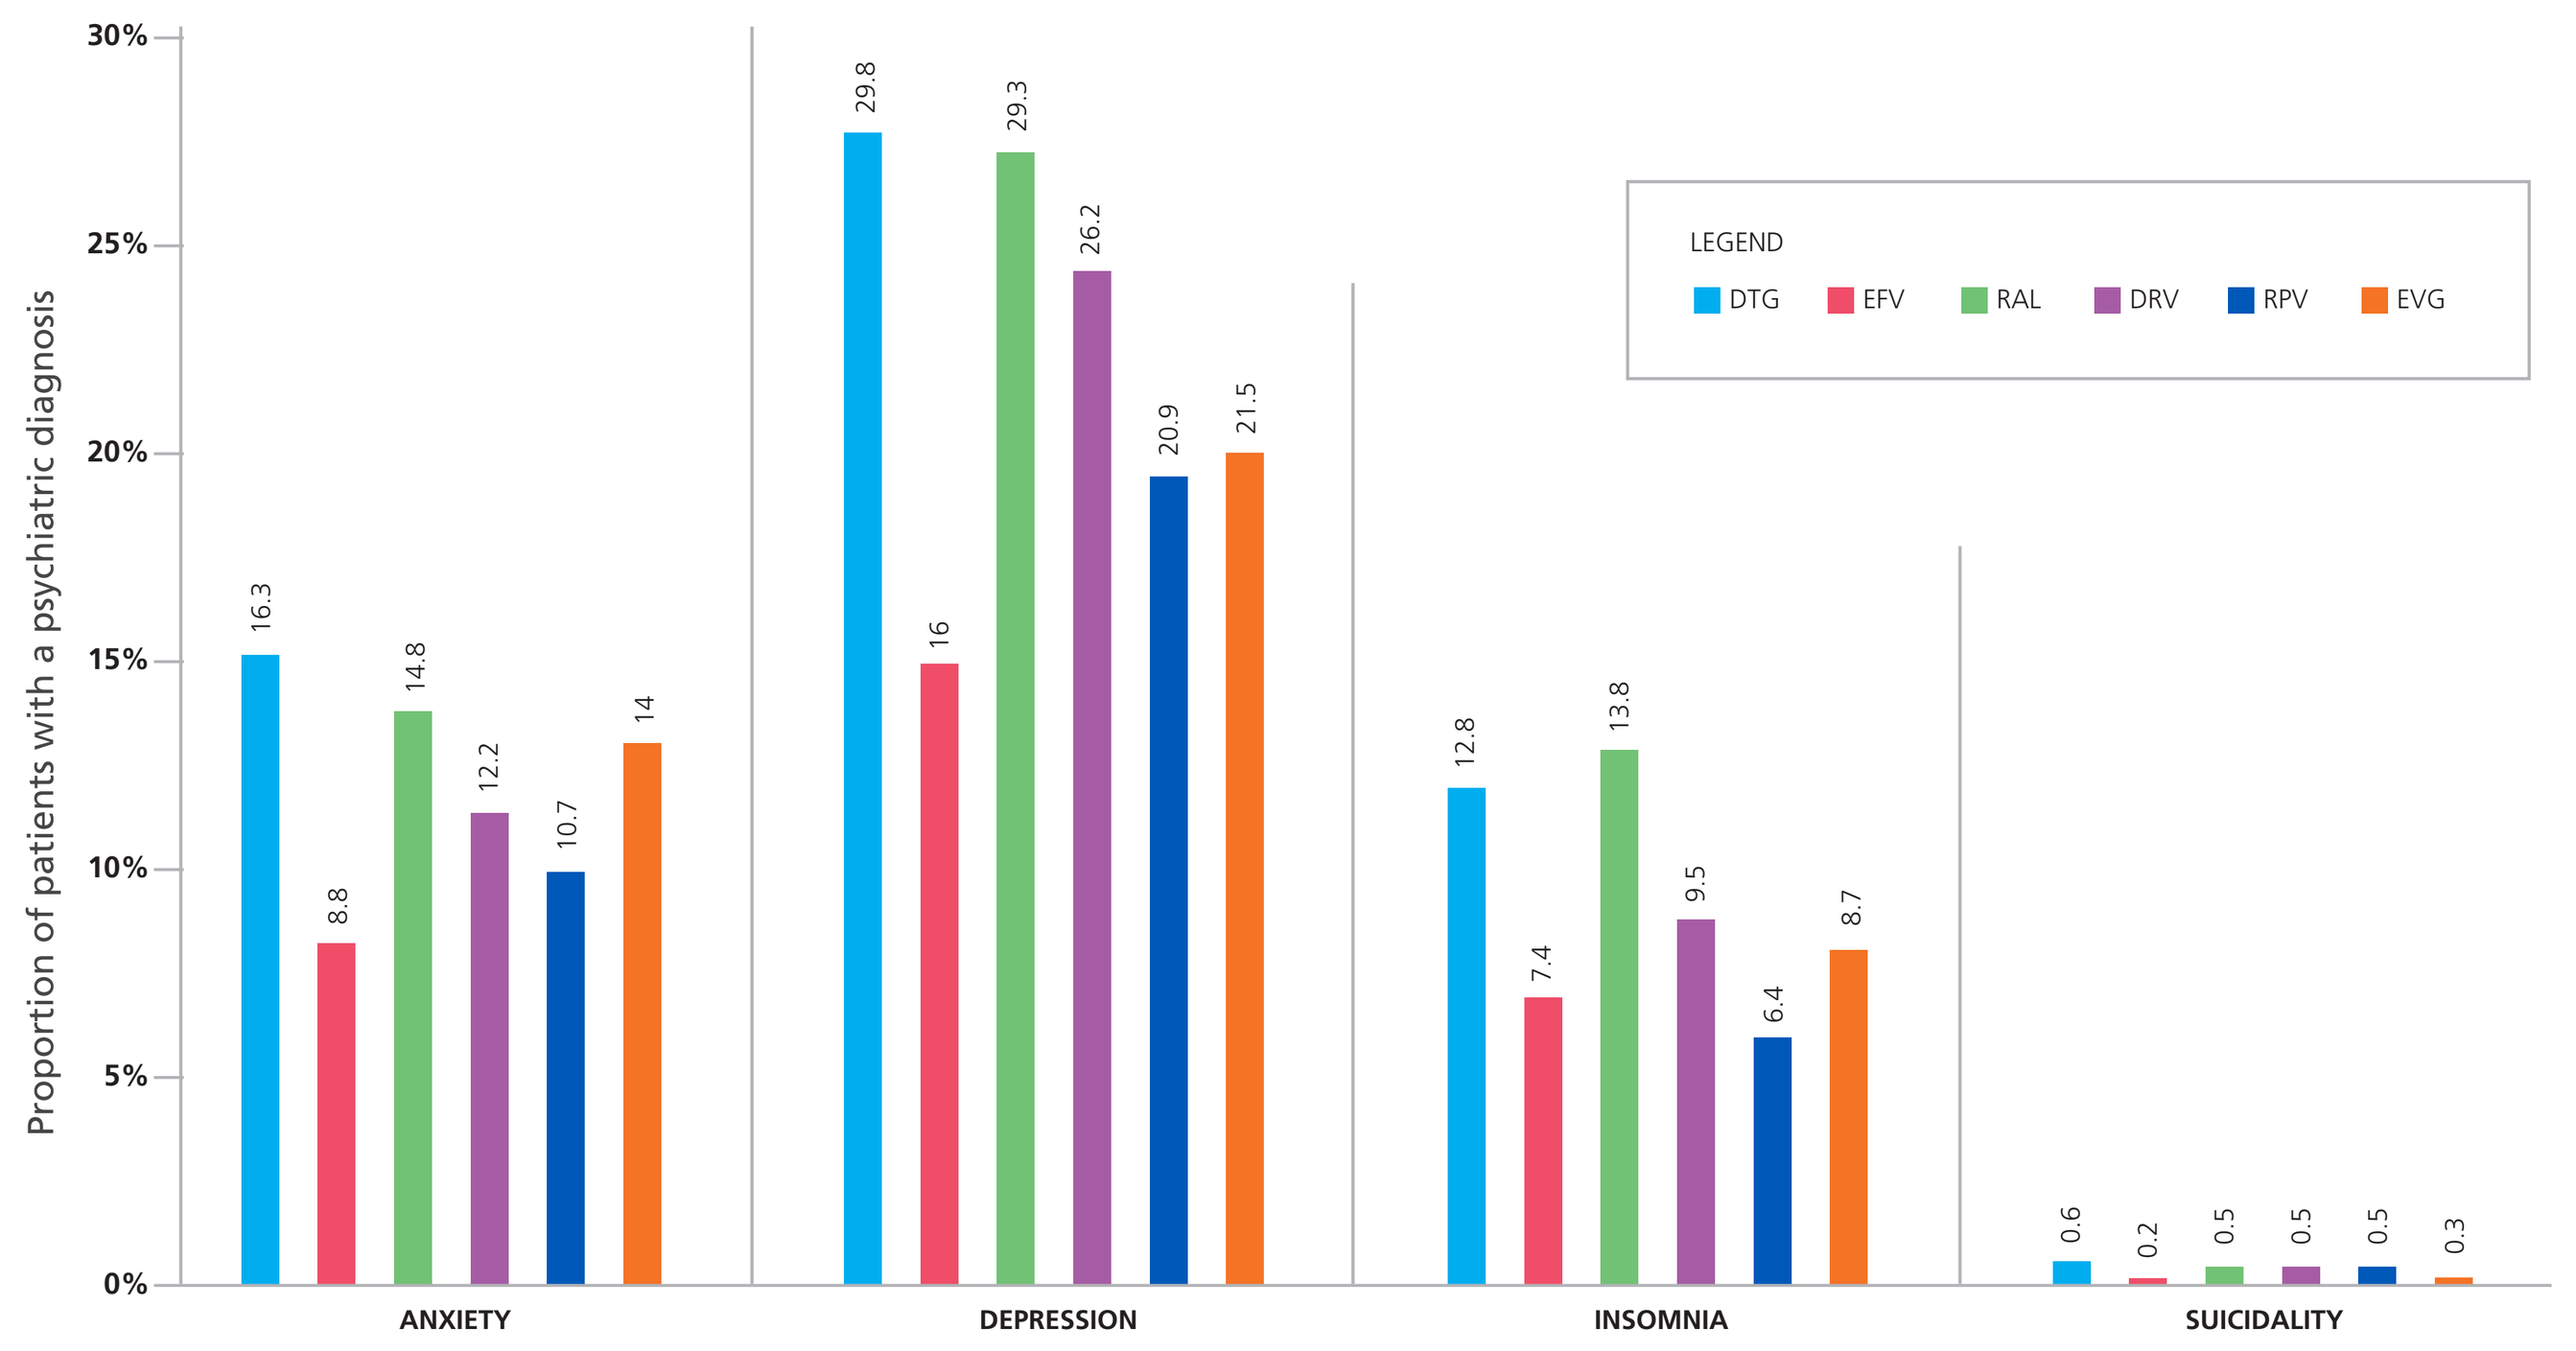

Proportion of patients with a psychiatric diagnosis
30%
25%
20%
15%
10%
5%
0%
16.3
8.8
14.8
12.2
10.7
14
29.8
16
29.3
26.2
20.9
21.5
12.8
7.4
13.8
9.5
6.4
8.7
0.6
0.2
0.5
0.5
0.5
0.3
ANXIETY
DEPRESSION
INSOMNIA
SUICIDALITY
LEGEND
DTG
EFV
RAL
DRV
RPV
EVG

- Patients receiving DTG were more likely to have a history of anxiety at baseline than patients receiving EFV, DRV, or RPV (all p<0.001). History of depression was more common in patients receiving DTG than in patients receiving EFV, RPV, or EVG (all p<0.0001). Patients taking DTG were also more likely to have a history of insomnia than patients taking EFV, DRV, RPV, or EVG (all p<0.0001). (Figure 2)
- When considering PD diagnoses, depression was the most common for all drugs. Patients prescribed RAL-containing regimens experienced more prevalent depression (p=0.006) and anxiety diagnoses (p=0.01) than patients taking DTG-containing regimens. Patients taking EVG-containing regimens also experienced more prevalent anxiety diagnoses (p=0.006) than patients taking DTG. (Figure 3)
- The incidence of new PD diagnoses (excluding those with a history of the condition) resulted in fewer events for all conditions except suicidality which was rare even without considering past history. There were no significant differences in incident PD diagnoses between patients receiving DTG and patients receiving any of the other anchor drugs. (Figure 3)
- Patients receiving DTG were the least likely to experience a discontinuation within 14 days of a prevalent PD diagnosis (DTG: 0.8%; EFV: 3.0%, p<0.0001; RAL: 3.7%, p<0.0001; DRV: 2.0%, p=0.0009; RPV: 1.5%, p=0.04; EVG: 1.2%, p=0.1), or an incident diagnosis of PD (DTG: 0.3%; EFV: 2.2%, p<0.0001; RAL: 1.7%, p<0.0001; DRV: 1.0%, p=0.006; RPV: 1.0%, p=0.006; EVG: 0.8%, p=0.04).

Figure 3. Prevalent and Incident psychiatric diagnoses during follow-up by anchor agent

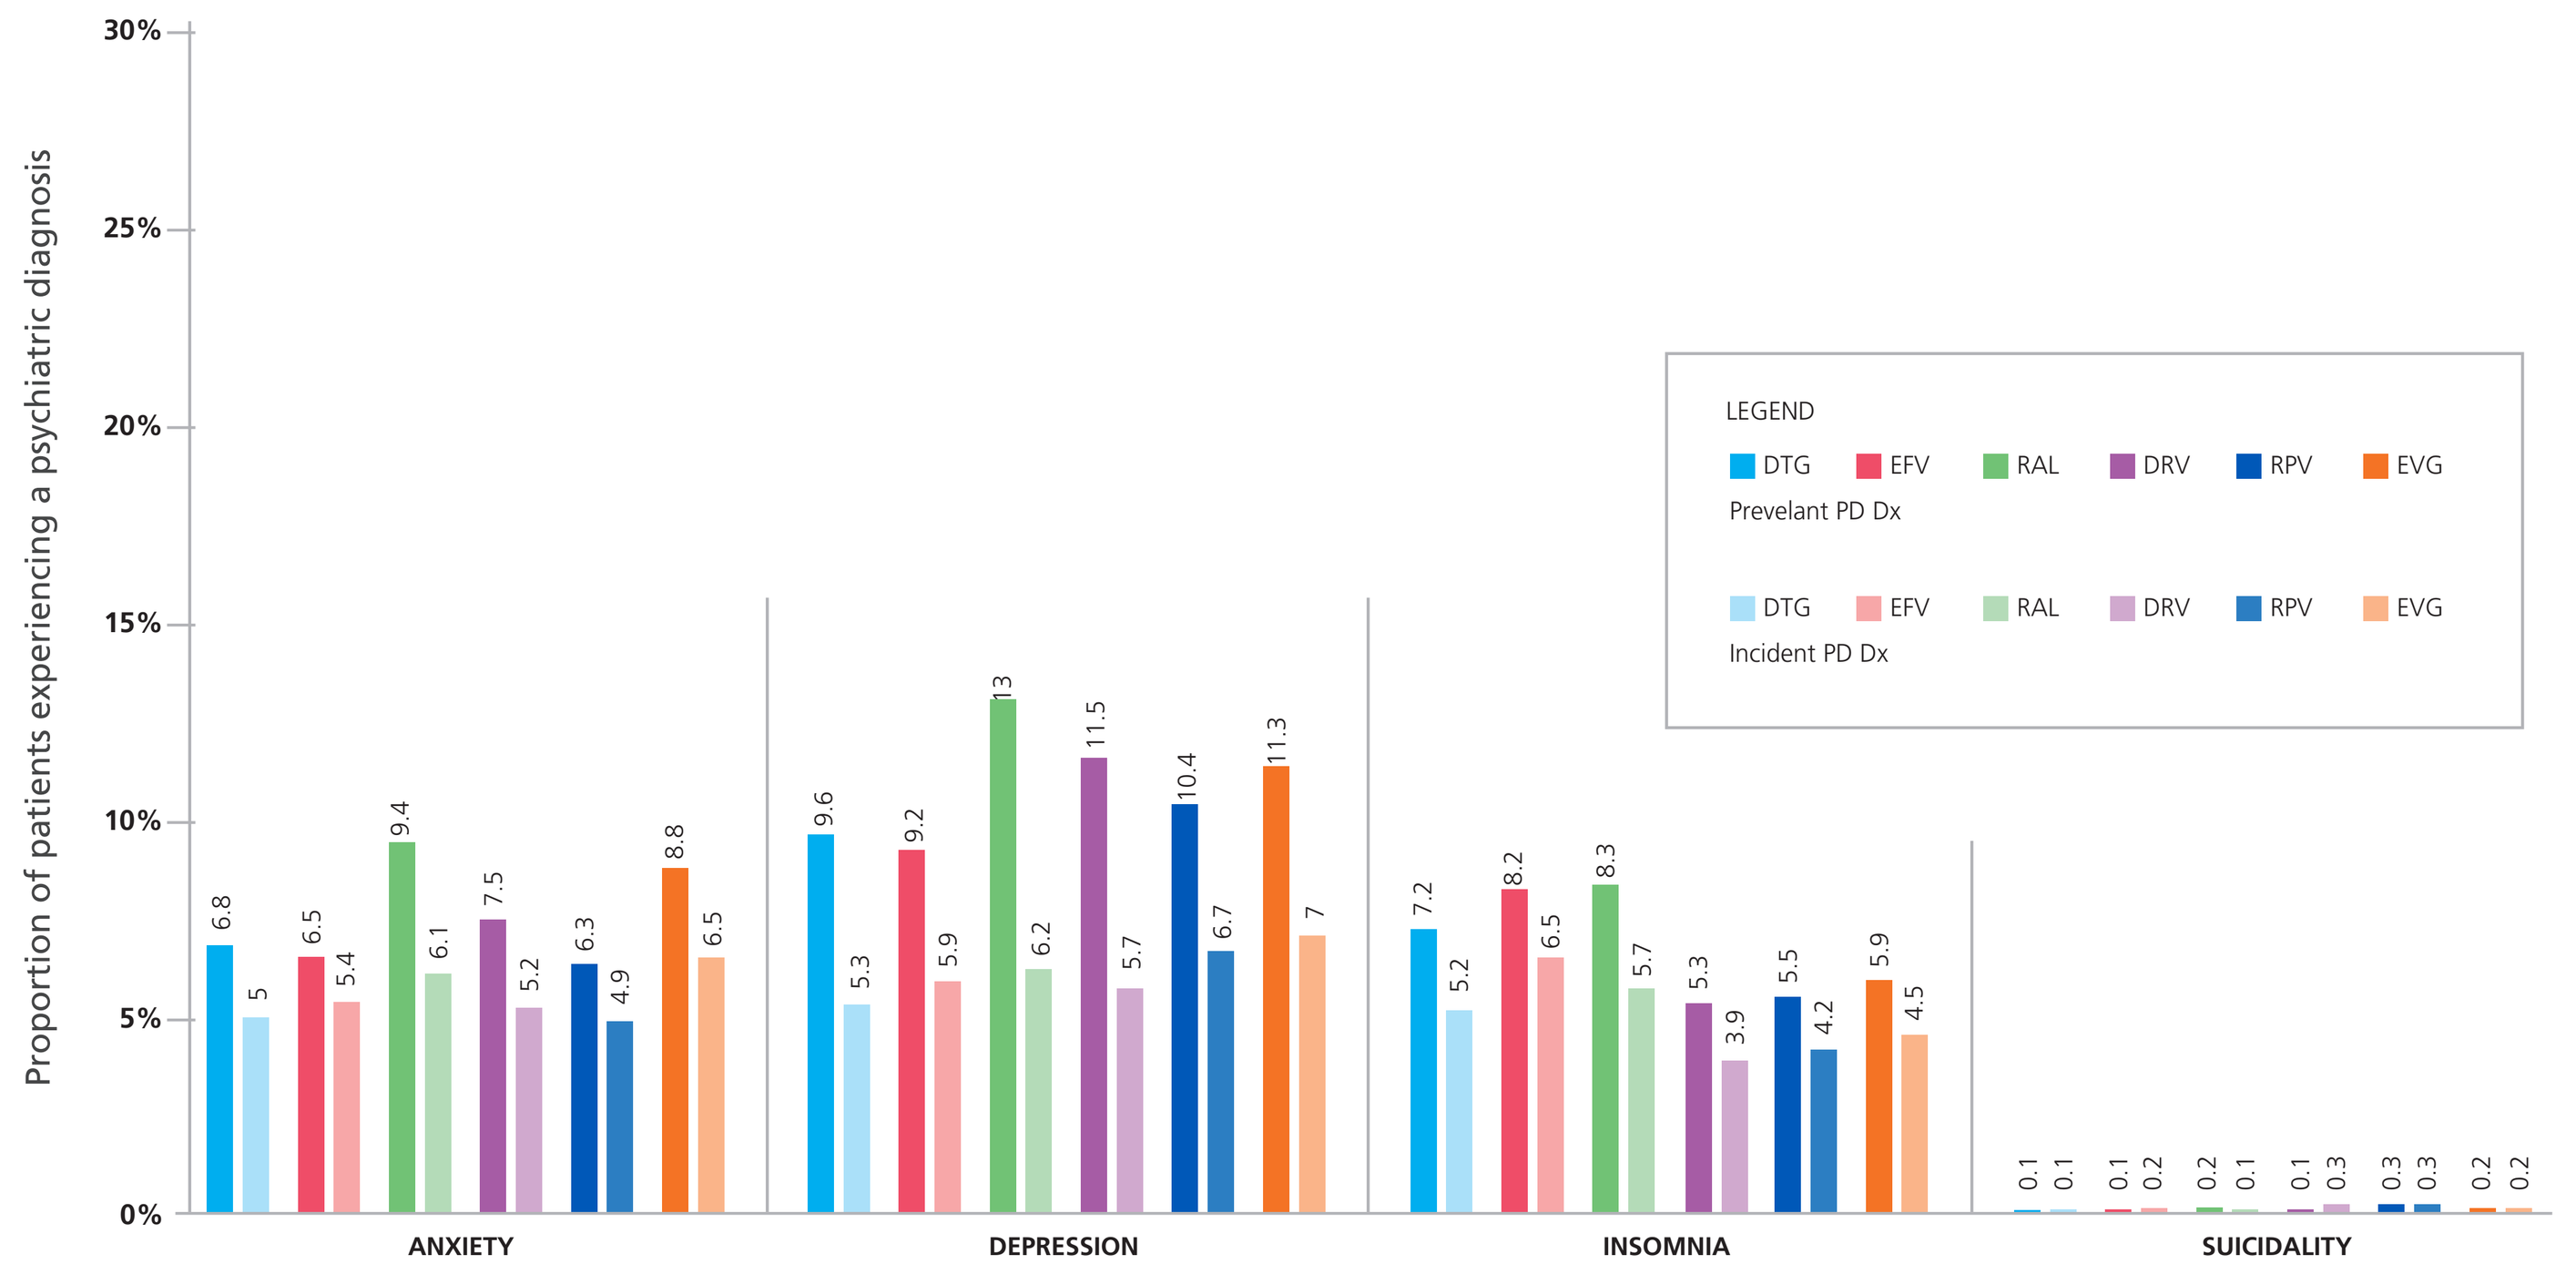

Proportion of patients experiencing a psychiatric diagnosis
30%
25%
20%
15%
10%
5%
0%
6.8
5
6.5
5.4
9.4
6.1
7.5
5.2
6.3
4.9
8.8
6.5
9.6
5.3
9.2
5.9
13
6.2
11.5
5.7
10.4
6.7
11.3
7
7.2
5.2
8.2
6.5
8.3
5.7
5.3
3.9
5.5
4.2
5.9
4.5
0.1
0.1
0.1
0.2
0.2
0.1
0.1
0.3
0.3
0.3
0.2
0.2
ANXIETY
DEPRESSION
INSOMNIA
SUICIDALITY
LEGEND
DTG EFV RAL DRV RPV EVG
Prevelant PD Dx
DTG EFV RAL DRV RPV EVG
Incident PD Dx

Figure 4. Kaplan-Meier estimates of time to first psychiatric disorder with discontinuation with number of subjects at risk

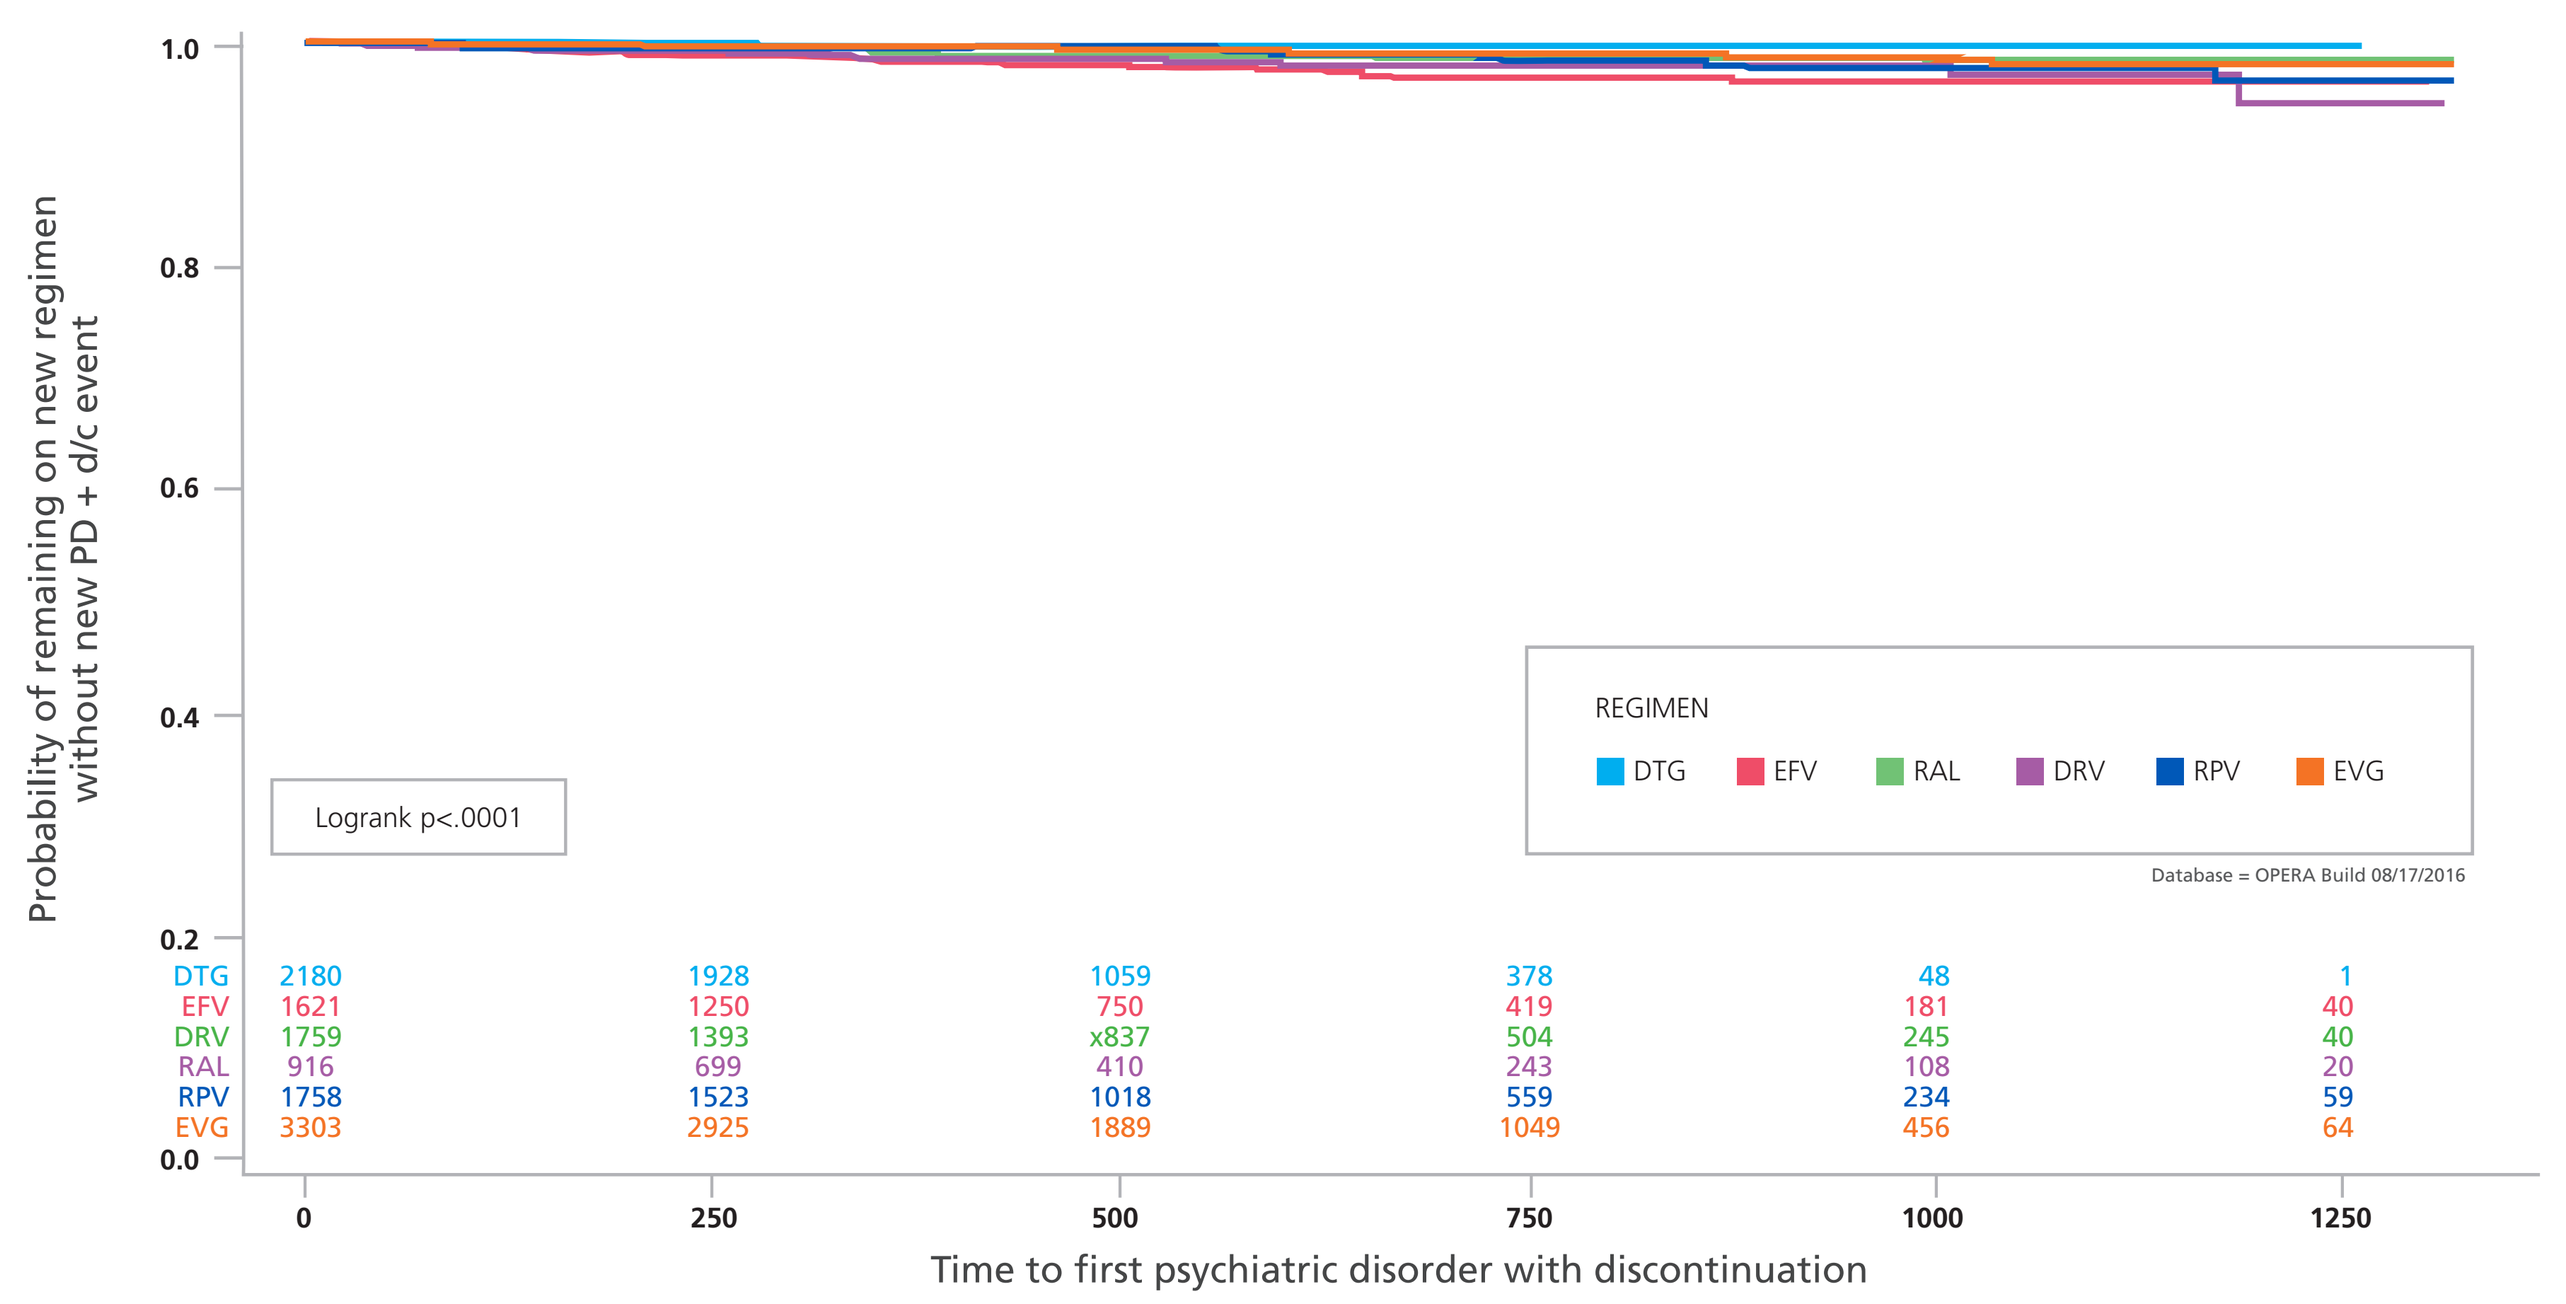

Probability of remaining on new regimen without new PD + d/c event
1.0
0.8
0.6
0.4
0.2
0.0
Logrank p<.0001
REGIMEN
DTG
EFV
RAL
DRV
RPV
EVG
Database = OPERA Build 08/17/2016
DTG 2180 1928 1059 378 48 1
EFV 1621 1250 750 419 181 40
DRV 1759 1393 x837 504 245 40
RAL 916 699 410 243 108 20
RPV 1758 1523 1018 559 234 59
EVG 3303 2925 1889 1049 456 64
0
250
500
750
1000
1250
Time to first psychiatric disorder with discontinuation

DISCUSSION

- In a large cohort of HIV+ patients in care, a history of PD diagnoses at baseline were common. Patients receiving DTG were more likely to have a history of PD diagnosis at baseline than patients taking EFV, DRV, RPV, or EVG. Baseline history of PD was similar between patients taking DTG and patients taking RAL.
- Patients taking DTG were not more likely than patients taking any of the other anchor drugs to experience a new diagnosis of PD after baseline (incident diagnosis).
- Patients receiving DTG-containing regimens were less likely to experience a prevalent or incident PD diagnosis followed by discontinuation of ART within 14 days of diagnosis compared to the other anchor drugs.
- Patient characteristics were unbalanced across the ARVs of interest (age, race, sex, AIDS, VL and CD4 at baseline). Analyses did not control for these differences.

KEY FINDING:

DTG use was not associated with an increased risk of psychiatric events or drug discontinuation due to psychiatric events, despite more patients with a history of psychiatric disorders being prescribed DTG treatment.

REFERENCES

1. Dube B, Benton T, Cruess DG, Evans DL. Neuropsychiatric manifestations of HIV infections and AIDS. *J Psychiatry and Neurosci* 2005; 30:237-246.
2. Abers MS, Shandera WX, Kass JS. Neurological and psychiatric adverse effects of antiretroviral drugs. *CNS Drugs* 2015; 28: 131-145.
3. van den Berk G, Oryszczyn J, Blok W, van der Meche N, Regez R, Ait Moha D, Brinkman K. Unexpectedly High Rate of Intolerance for Dolutegravir in Real-Life Setting. Poster #948. Presented at: Conference on Retroviruses and Opportunistic Infections. Boston, MA. February 22-25, 2016.
4. Fettiplace A, Stainsby C, Winston A, et al. Psychiatric symptoms in patients receiving dolutegravir. *JAIDS* 2016; DOI:10.1097/QAI.0000000000001269.

ACKNOWLEDGMENTS

This research wouldn't be possible without the participation of HIV caregivers and their patients. Additionally, we are grateful for the following contributions: Robin Beckerman (SAS programming), Jeff Briney (QA/QC), Rodney Mood (site selection and support), Ted Ising (database architecture and support), Bernie Stooks (database support) and Judy Johnson (medical terminology classification).

SUPPORT

This research was funded by ViiV Healthcare.

ViiV Healthcare

Epividian®