POSTER #1086

Virologic Effectiveness of Abacavir/Lamivudine with Darunavir versus Other Protease Inhibitors in Treatment-Experienced HIV-Infected Patients in Clinical Practice

Phil Lackey[1], Anthony Mills[2], Felix Carpio[3], Ricky Hsu[4], Edwin DeJesus[5], Gerald Pierone[6], Cassidy Henegar[7], Emily Brouwer[8], Jennifer Fusco[7], Gregory Fusco[7], and Michael Wohlfeiler[9]

[1] Carolinas Healthcare System, Charlotte, NC; [2] Anthony Mills MD, Inc., Los Angeles CA; [3] Altamed Health Services, Los Angeles, CA; [4] Ricky Hsu MD PC, New York, NY; [5] Orlando Immunology Center, Orlando, FL; [6] AIDS Research and Treatment Center of the Treasure Coast, Vero Beach, FL; [7] OPERA Database, Epividian Inc., Durham, NC; [8] University of Kentucky, Lexington, KY; [9] AIDS Healthcare Foundation, Los Angeles, CA

OPERA® The Longitudinal Cohort

Contact Information:
4819 Emperor Blvd., Suite 400 Durham, NC 27703
P: 919-825-3457 F: 919-313-4505
Email :Cassidy.Henegar@epividian.com

BACKGROUND

The standard of care for HIV treatment is a three-drug regimen consisting of two nucleoside reverse transcriptase inhibitors (NRTIs) and either a protease inhibitor (PI) or an integrase strand transfer inhibitor (INSTI). Darunavir boosted with ritonavir (DRV/r) is the only preferred PI in the US DHHS HIV Treatment Guidelines for naïve patients, recommended in combination with tenofovir (TDF) /emtricitabine (FTC). [1] ABC/3TC+DRV/r is considered an acceptable regimen alternative for certain ART-naïve patients and for treatment-experienced patients. While ABC/3TC+DRV/r is considered an effective and tolerable regimen according to the guidelines, few studies have evaluated virologic response of patients on DRV/r with a backbone of ABC/3TC.

OBJECTIVE:

To compare the virologic effectiveness of ABC/3TC+DRV/r to ABC/3TC+PIs (non-DRV) in treatment-experienced HIV-positive patients.

METHODS

Study Population/ Design

The study population was selected from the Observational Pharmaco-Epidemiology Research & Analysis (OPERA) cohort, which includes prospectively-captured, routine clinical data from patients at 72 outpatient clinics in the U.S.

Figure 1: Geographic Distribution of the OPERA Cohort

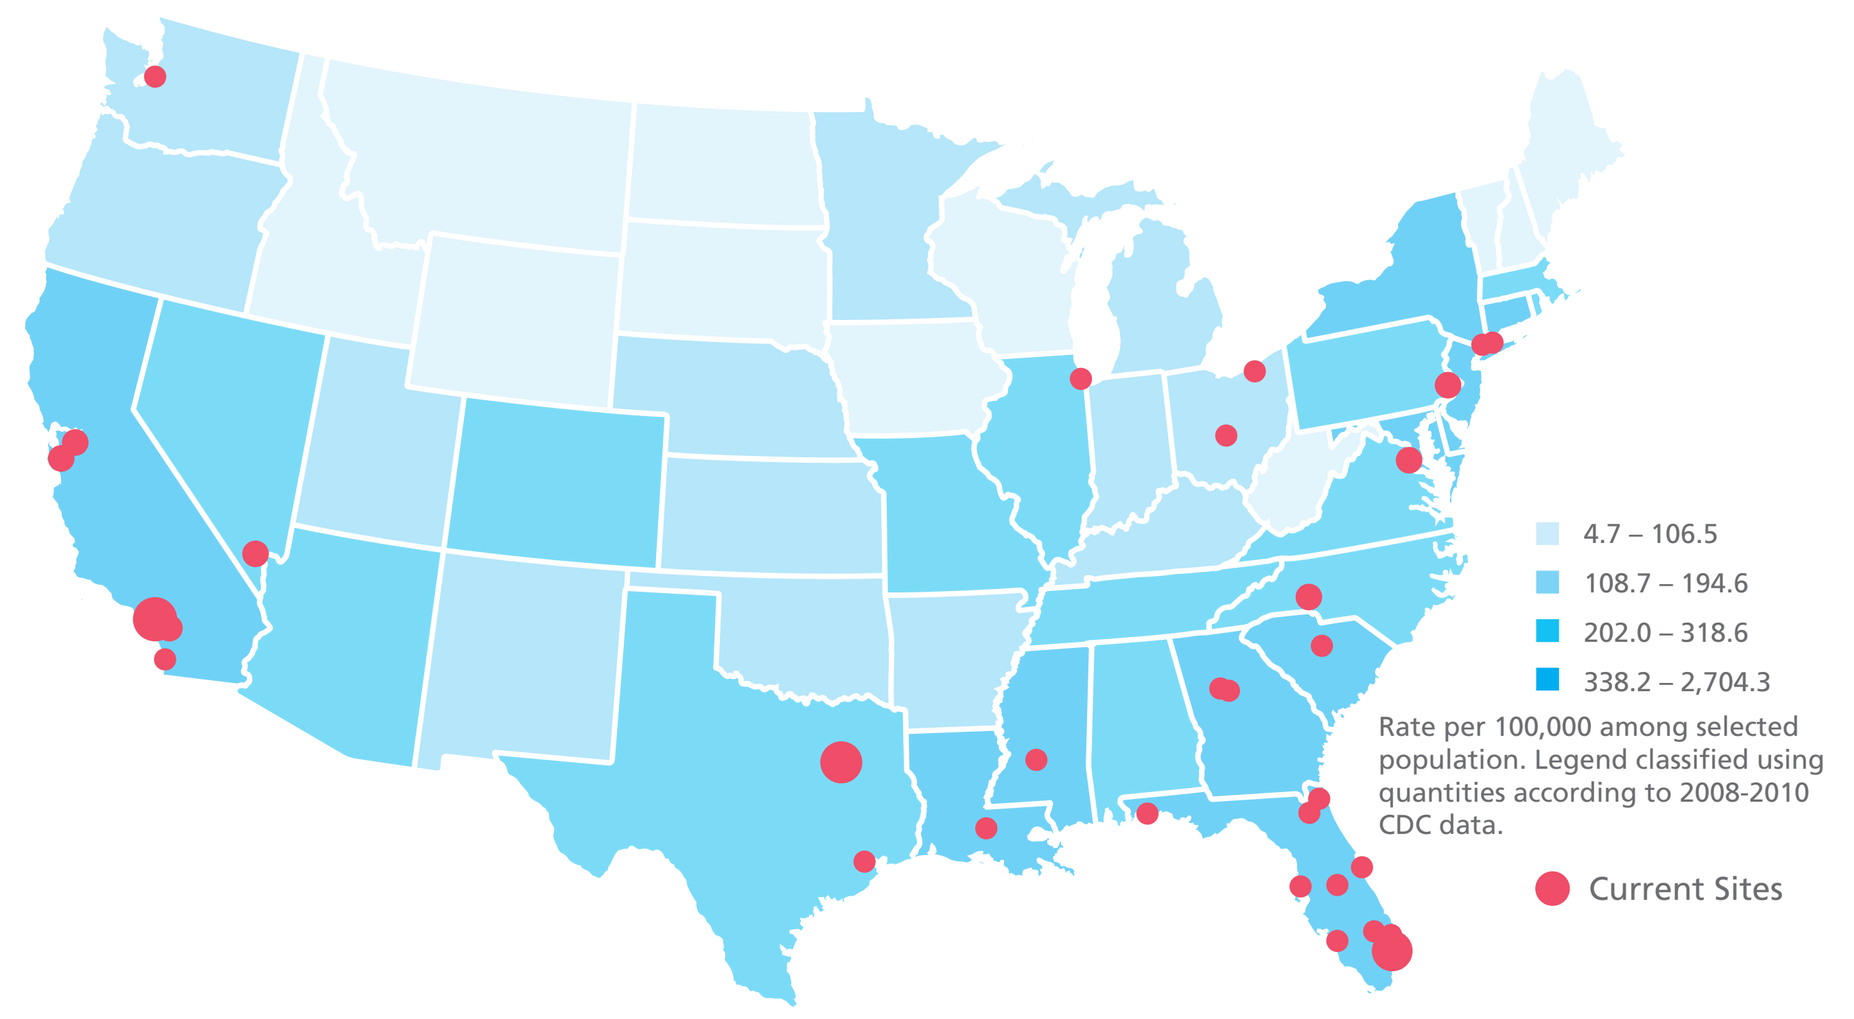

4.7 – 106.5
108.7 – 194.6
202.0 – 318.6
338.2 – 2,704.3
Rate per 100,000 among selected population. Legend classified using quantities according to 2008-2010 CDC data.
Current Sites

Patients meeting the following inclusion criteria were included in the analysis: HIV diagnosis, starting regimen containing ABC/3TC, treatment experienced, and both CD4 and VL results available. Eligible patients were categorized into 2 groups: ABC/3TC+DRV/r or ABC/3TC + another PI. The outcome of interest was an undetectable viral load.

Statistical Analysis

Multivariable logistic regression models were fit to assess the association between regimen exposure and viral load suppression. Sensitivity analyses to balance potential time of follow-up between treatment groups were conducted, including restricting to patients with at least 12 months of follow-up and restricting to patients starting their ABC/3TC regimen in 2009 or later.

RESULTS

Figure 2. OPERA patients eligible for analysis as of May 06, 2015

Total OPERA population at freeze date: 560,990 patients

HIV Diagnosis: 58,707

Received treatment with ARVs: 51,036

Took ABC/3TC: 12,542

ARV-experienced prior to ABC/3TC: 6,173

ABC/3TC started 2005 or later: 4,128

Active in OPERA at ABC/3TC start: 3,770

Had baseline/follow-up results: 2,122

Took ABC/3TC with a PI: 676

ABC/3TC+DRV/r n=151 | ABC/3TC+PI (non-DRV) n=525

Table 1. Baseline characteristics of ART-experienced patients initiating their first regimen of either ABC/3TC+DRV/r or ABC/3TC + PI/r (not DRV)

	ABC/3TC+DRV/r n=151 n (%)*	ABC/3TC+PI/r (non-DRV) n=525 n (%)*	p-value
Clinic Region			
Northeast	0 (0.0)	4 (0.8)	**0.001**
Mid Atlantic	3 (2.1)	15 (2.9)	
South	66 (43.7)	200 (38.1)	
Midwest	0 (0.0)	1 (0.2)	
Southwest	6 (4.0)	1 (0.2)	
West	76 (50.3)	304 (57.9)	
Male Sex	124 (82.1)	415 (79.0)	0.408
Age (Median (IQR)	46.8 (39.6, 53.4)	45.6 (39.1, 52.8)	0.614
African American Race	65 (43.0)	212 (40.4)	0.557
Hispanic Ethnicity	20 (13.2)	67 (12.8)	0.876
AIDS-defining event at or before baseline	16 (11.3)	64 (12.2)	0.593
CD4 count at baseline			
< 250 cells/mm³	51 (33.8)	156 (29.7)	0.340
≥ 250 cells/mm³	100 (66.2)	369 (70.3)	
HIV viral load at baseline			
< 200 copies/mL	89 (58.9)	348 (66.3)	0.096
≥ 200 copies/mL	62 (41.1)	177 (33.7)	
≤ 20,000 copies/mL	130 (86.1)	436 (83.0)	0.372
> 20,0000 copies/ mL	21 (13.9)	89 (17.0)	
Hepatitis B or C co-infection at baseline	4 (2.6)	3 (0.6)	**0.026**

* Reported as n(%) unless otherwise indicated

Figure 3. Proportion of patients initiating ABC/3TC+DRV/r (n=151) and ABC/3TC+PI/r (non-DRV) (n=525) regimens by year of start

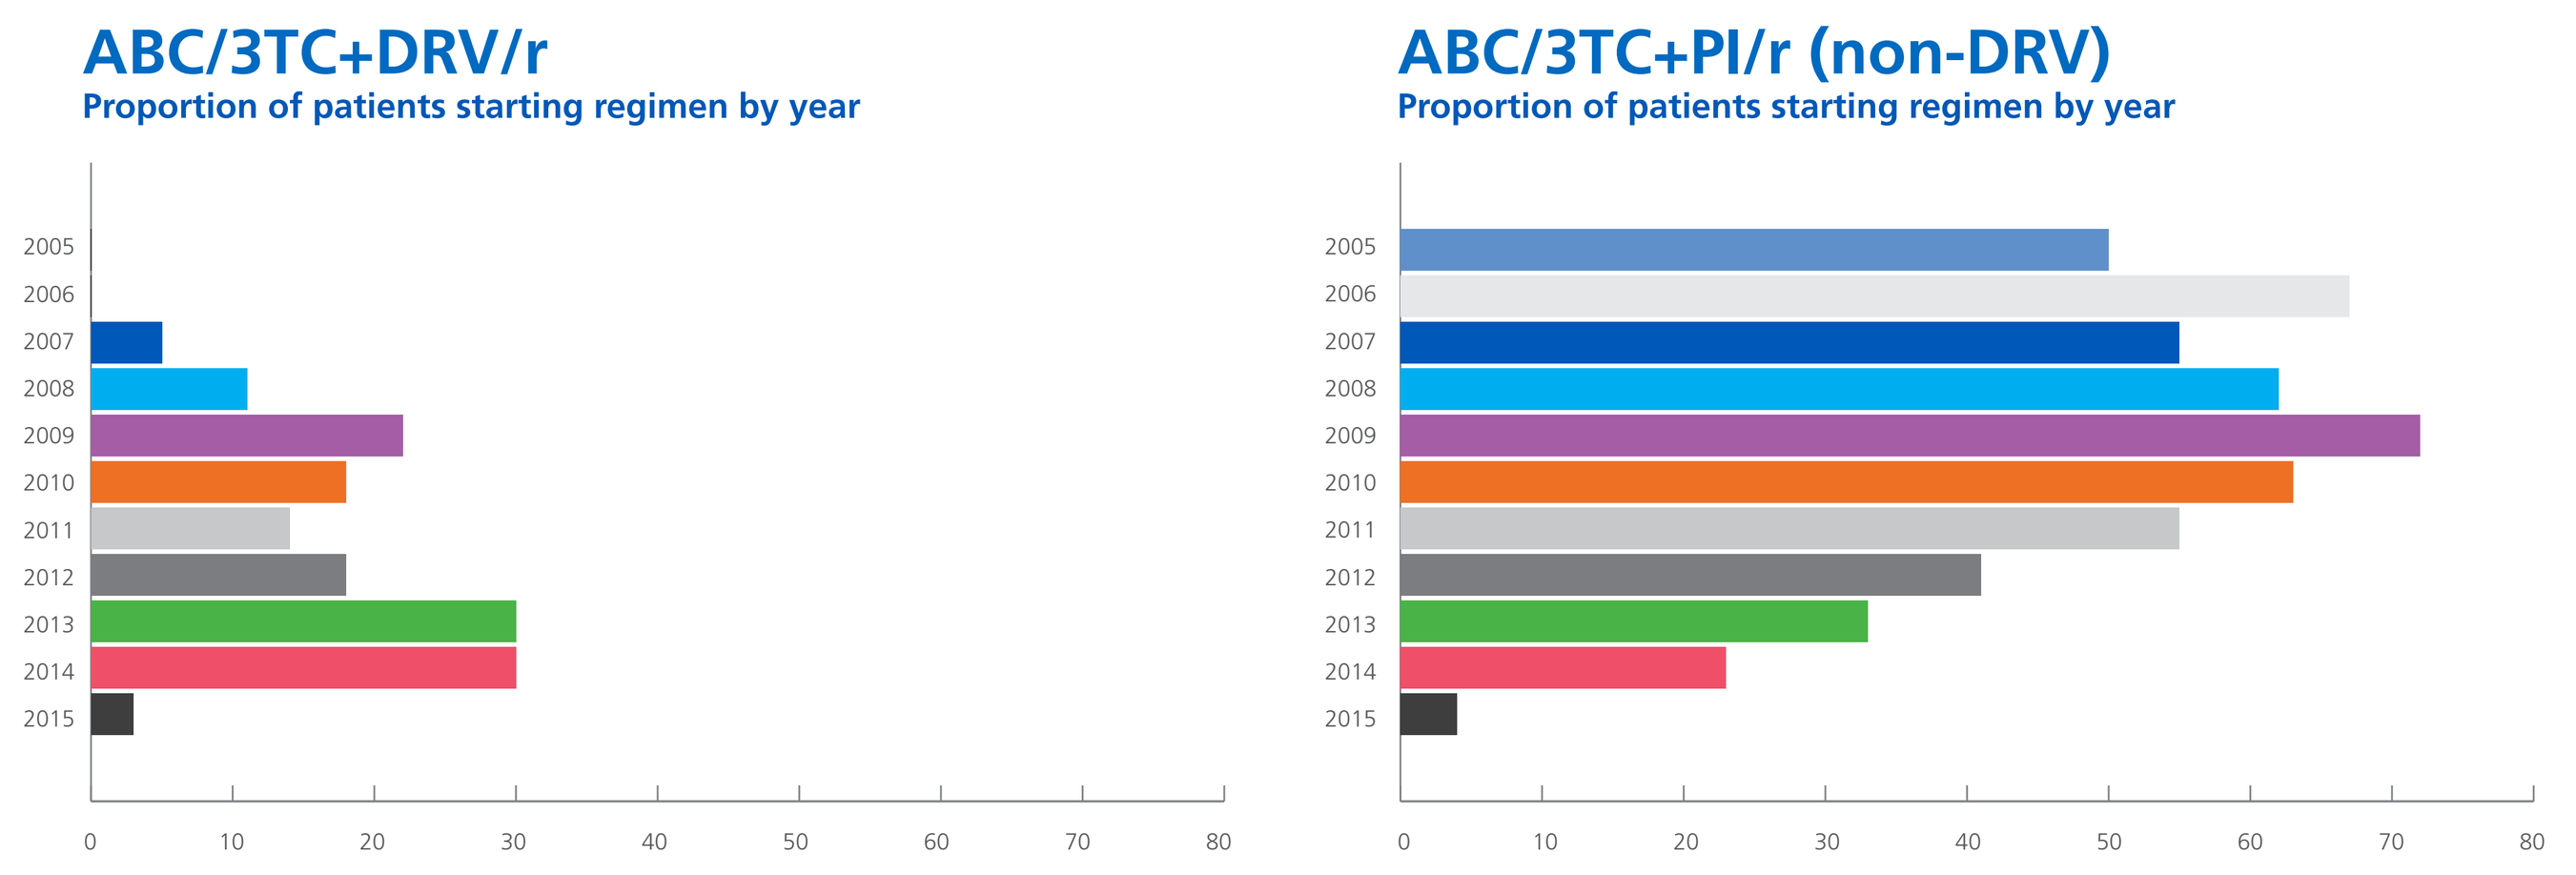

ABC/3TC+DRV/r
Proportion of patients starting regimen by year
ABC/3TC+PI/r (non-DRV)
Proportion of patients starting regimen by year
2005
2006
2007
2008
2009
2010
2011
2012
2013
2014
2015
0
10
20
30
40
50
60
70
80

No patients received ABC/3TC+DRV/r in the first 2 years of the study (2005, 2006). The proportion of patients receiving DRV/r with ABC/3TC increased during the study period, while the proportion of patients receiving ABC/3TC with a different PI showed a decreasing trend.

All patients in the DRV treatment group took DRV boosted with RTV. In the non-DRV treatment group, the majority of patients received atazanavir (n=389, 74.1%), boosted (n=261) or unboosted (n=128) with RTV, or lopinavir + RTV (n=75, 14.3%).

Table 2. Follow-up in as-treated and intent-to-treat analyses*

	ABC/3TC+DRV/r	ABC/3TC+PI/r (non-DRV)	p-value
As-treated follow-up (months)	13.7 (7.6, 22.9)	17.3 (6.9, 34.9)	0.036
Viral load results during follow-up	4 (3, 7)	5 (3, 9)	0.176
CD4 results during follow-up	4 (3, 8)	5 (3, 9)	0.09
Intent-to-treat follow-up (months)	33.1 (17.1, 63.5)	68.1 (43.9, 94.7)	<0.001
Viral load results during follow-up	7 (3, 13)	11 (6, 18)	<0.001
CD4 results during follow-up	7 (3, 14)	12 (6, 20)	<0.001

*Reported as median and interquartile range unless noted

Figure 4.
Crude and adjusted logistic regression results for primary and sensitivity analyses

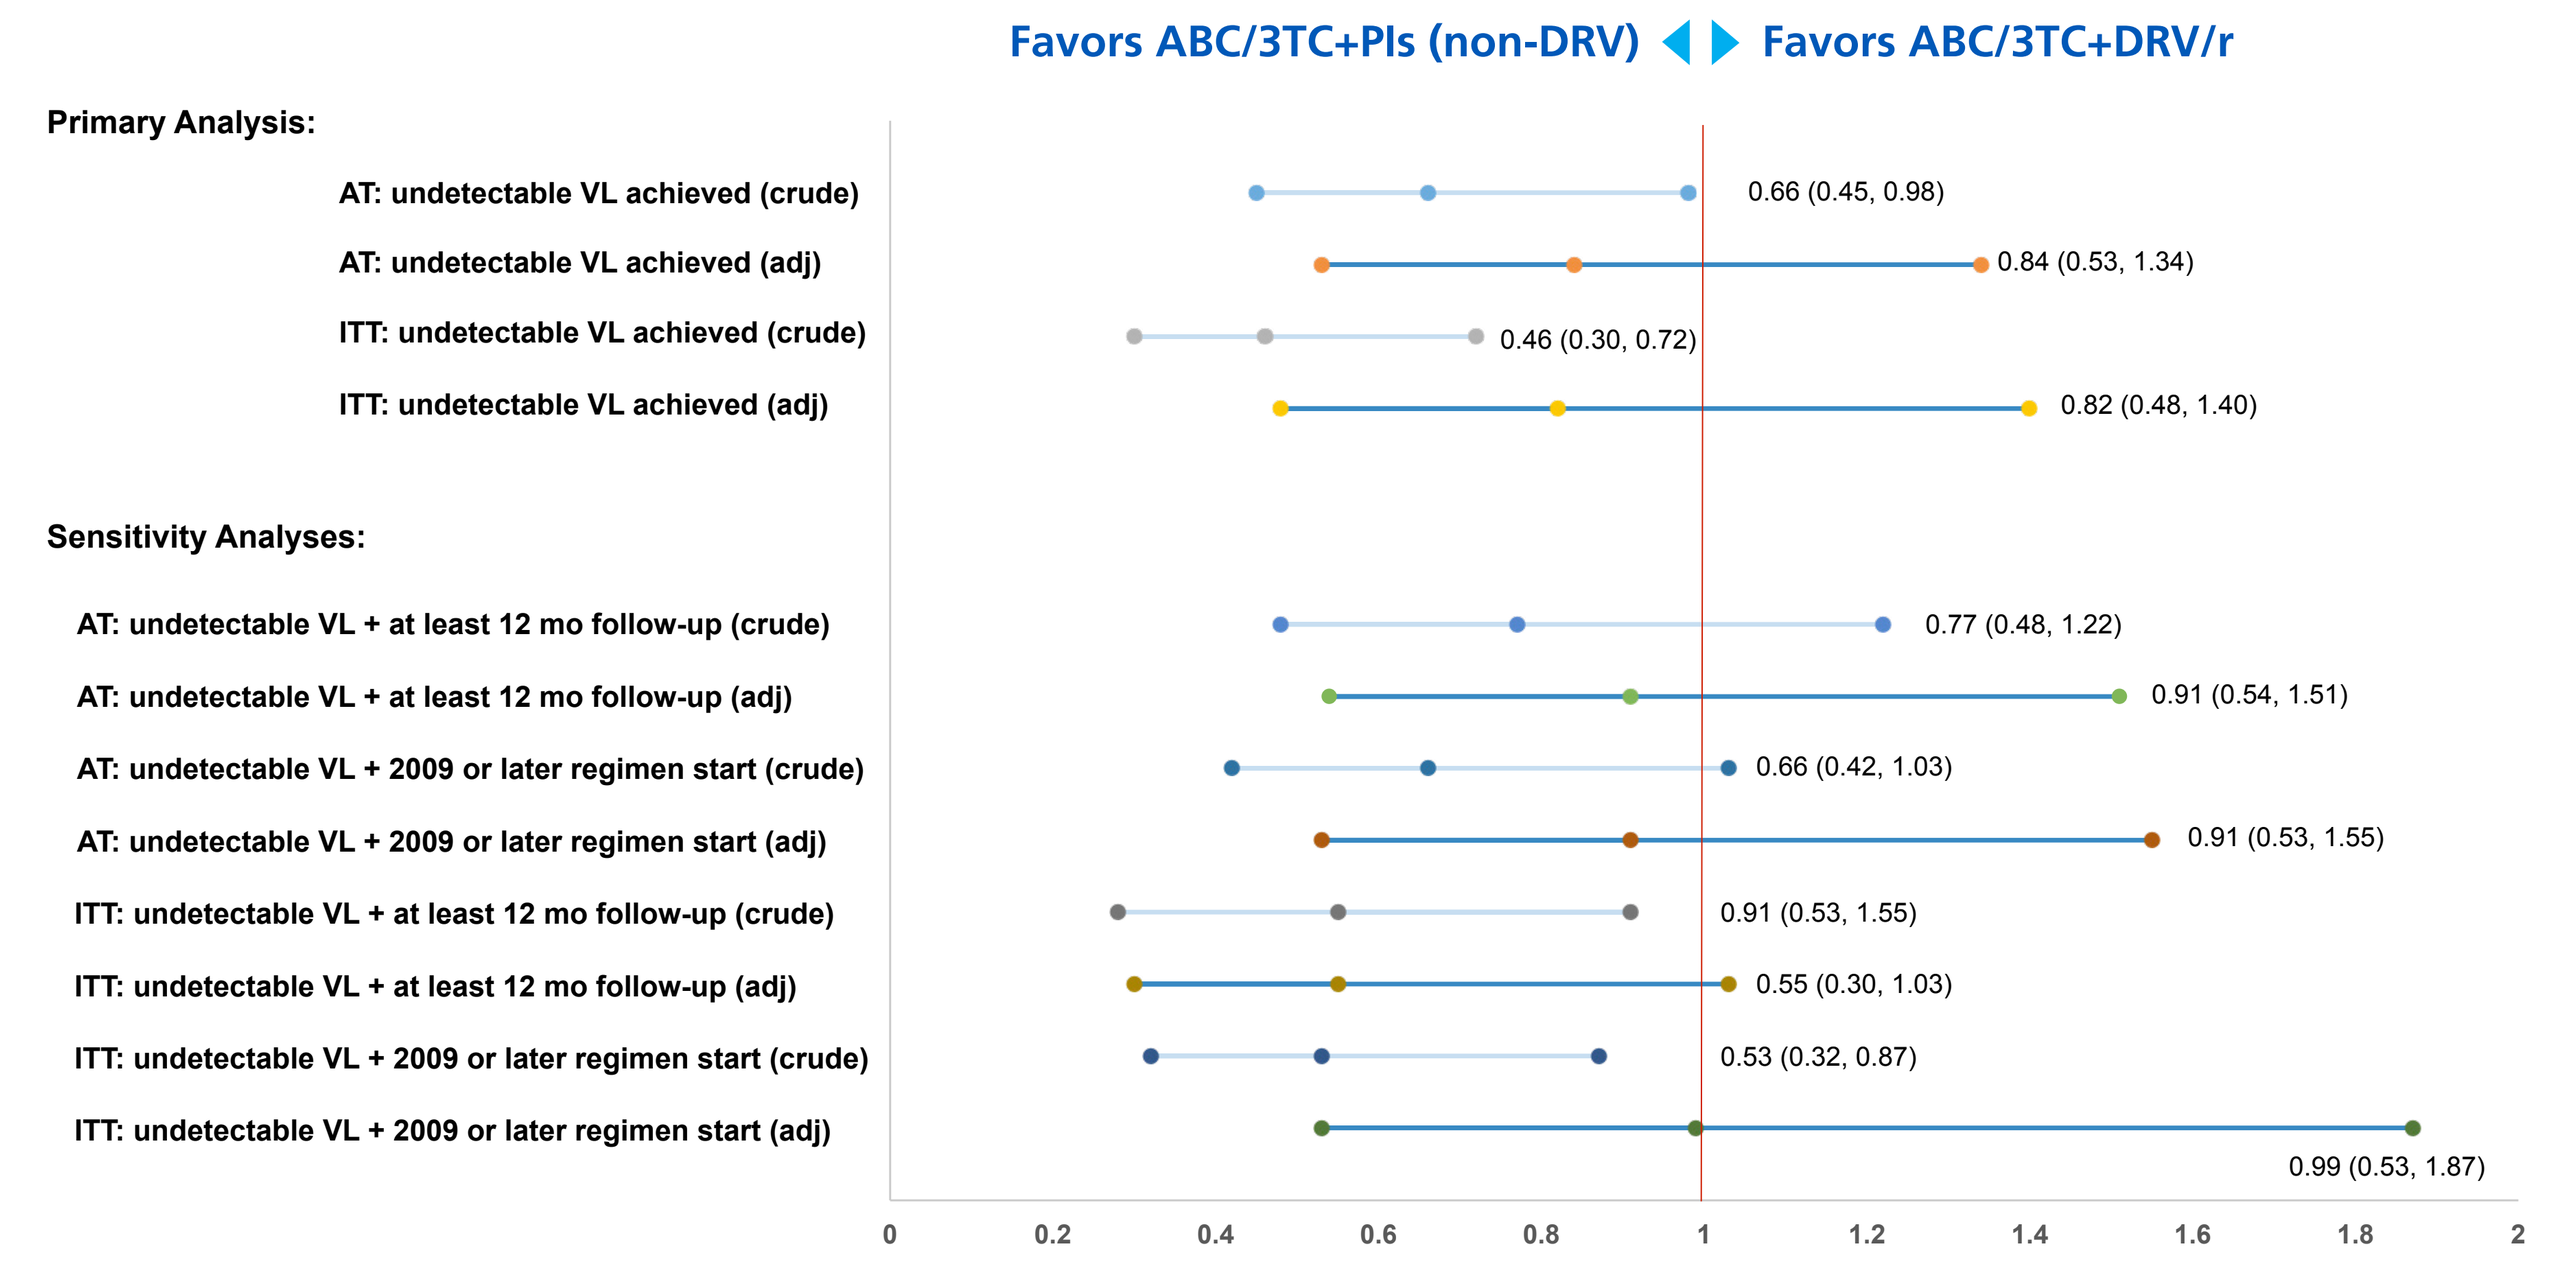

Favors ABC/3TC+PIs (non-DRV)
Favors ABC/3TC+DRV/r
Primary Analysis:
AT: undetectable VL achieved (crude)
0.66 (0.45, 0.98)
AT: undetectable VL achieved (adj)
0.84 (0.53, 1.34)
ITT: undetectable VL achieved (crude)
0.46 (0.30, 0.72)
ITT: undetectable VL achieved (adj)
0.82 (0.48, 1.40)
Sensitivity Analyses:
AT: undetectable VL + at least 12 mo follow-up (crude)
0.77 (0.48, 1.22)
AT: undetectable VL + at least 12 mo follow-up (adj)
0.91 (0.54, 1.51)
AT: undetectable VL + 2009 or later regimen start (crude)
0.66 (0.42, 1.03)
AT: undetectable VL + 2009 or later regimen start (adj)
0.91 (0.53, 1.55)
ITT: undetectable VL + at least 12 mo follow-up (crude)
0.91 (0.53, 1.55)
ITT: undetectable VL + at least 12 mo follow-up (adj)
0.55 (0.30, 1.03)
ITT: undetectable VL + 2009 or later regimen start (crude)
0.53 (0.32, 0.87)
ITT: undetectable VL + 2009 or later regimen start (adj)
0.99 (0.53, 1.87)
0
0.2
0.4
0.6
0.8
1
1.2
1.4
1.6
1.8
2

* Primary analysis comparing odds of achieving undetectable viral load among those taking ABC/3TC+DRV to those taking ABC/3TC+PI (non-DRV)
AT: As-treated analysis, ITT: Intent-to-treat analysis

DISCUSSION

To our knowledge, this is the first observational study comparing the effectiveness of DRV to other PIs when taken in combination specifically with an ABC/3TC NRTI backbone. Prior to adjustment and during the course of the regimen (as-treated analysis), patients taking ABC/3TC+DRV/r appear to be less likely to achieve virologic suppression to an undetectable level compared to patients taking ABC/3TC with other PIs [OR (95% CI): 0.66 (0.44, 0.98)]. After adjusting for factors associated with baseline health (CD4, VL, HBV/HCV co-infection), no significant difference between treatment groups was observed [OR (95% CI): 0.84 (0.53, 1.34)].

While not reaching statistical significance, patients taking ABC/3TC+DRV/r tended to have a lower median CD4 and were less likely to have a viral load <200 copies/mL at baseline. Patients taking DRV/r were also more likely to have hepatitis at the start of the regimen. Patients in both treatment groups seemed to show similar treatment benefit in terms of reductions in viral load (median DRV vs. non-DRV: -23 vs. -23 copies/mL; p=0.72) and gains in CD4 T-cell counts (median DRV vs. non-DRV: 106 vs. 118 cells/mm3; p=0.60] while on their initial ABC/3TC regimens. Even though patients with potentially poorer health indicators at baseline were more likely to initiate ABC/3TC with DRV than with other PIs, they appear to experience similar treatment benefits to patients taking ABC/3TC with other PIs in terms of absolute reductions in viral load.

Unadjusted differences in effectiveness between the two treatment groups were at least partially attributable to differences in follow-up time, particularly for the intent-to-treat analysis. Patients taking ABC/3TC+PI (non-DRV) were more likely to achieve a single undetectable viral load simply because they had a longer opportunity to do so. Two sensitivity analyses attempted to balance the potential length of follow-up between the two treatment groups. The resulting unadjusted and adjusted OR estimates were attenuated toward the null, suggesting that the difference in follow-up time accounted for some of the observed difference in effect.

KEY FINDING:

In an observational study of patients in clinical care in the US, no difference was observed in multivariable logistic regression analysis comparing the use of ABC/3TC+DRV/r versus ABC/3TC+PI (non-DRV).

REFERENCES

1. Panel on Antiretroviral Guidelines for Adults and Adolescents. Guidelines for the use of antiretroviral agents in HIV-1-infected adults and adolescents. Department of Health and Human Services. Available at: http://aidsinfo.nih.gov/contentfiles/lvguidelines/AdultandAdolescentGL.pdf Accessed 25 September, 2015.

ACKNOWLEDGMENTS

This research wouldn't be possible without the participation of HIV caregivers and their patients. Additionally, we are grateful for the following contributions: Robin Beckerman (SAS programming), Rodney Mood (site selection and support), Ted Ising (database architecture and support), and Bernie Stooks (database support).

SUPPORT

This research was funded by ViiV Healthcare.

Epividian®